THE WEE1 INHIBITOR DEBIO 0123 ENHANCES THE EFFICACY OF STANDARD OF CARE DNA DAMAGING AGENTS IN LUNG CANCER MODELS

Luke Piggott, Anne Vaslin-Chessex, Noémie Luong, Benjamin Tschumi, Grégoire Vuagniaux

Debiopharm International SA, Chemin Messidor 5-7, CH-1002 Lausanne, Switzerland.

Debiopharm™ WE DEVELOP FOR PATIENTS

ABSTRACT #4894

SUMMARY

Debio 0123 is an investigational, orally bioavailable, highly selective, adenosine triphosphate (ATP)-competitive inhibitor of the WEE1 tyrosine kinase. WEE1 is a key regulator of cell cycle progression that influences entry into mitosis by modulating activity of cyclin-dependent kinase 1 (CDK1, also referred to as cell division cycle 2 [CDC2]). Inhibition of WEE1 presents an opportunity as a therapeutic target in cancer therapy, either in cells relying on cell cycle checkpoints regulated by WEE1 or to potentiate chemotherapy and radiation therapy[1]. The proposed mechanism of action of Debio 0123 involves promoting entry into uncontrolled mitosis for cells with accumulated DNA damage and, ultimately, cell death via mitotic catastrophe.

The nonclinical data suggest Debio 0123 to be a good candidate for clinical development with the potential to improve therapy outcomes of patients with cancer, as monotherapy or when administered in combination with modalities that induce DNA damage, for example chemotherapies and radiotherapy. Small cell lung cancer (SCLC) is an aggressive disease with poor clinical outcomes that carries a high mutational burden and genomic instability. Here we investigated the ability of Debio 0123 to enhance SCLC response to standard of care (SOC) DNA damaging agents carboplatin and etoposide *in vitro* and *in vivo.*

BACKGROUND

Debio 0123 is a selective and orally available ATP-competitive inhibitor of WEE1 kinase

A

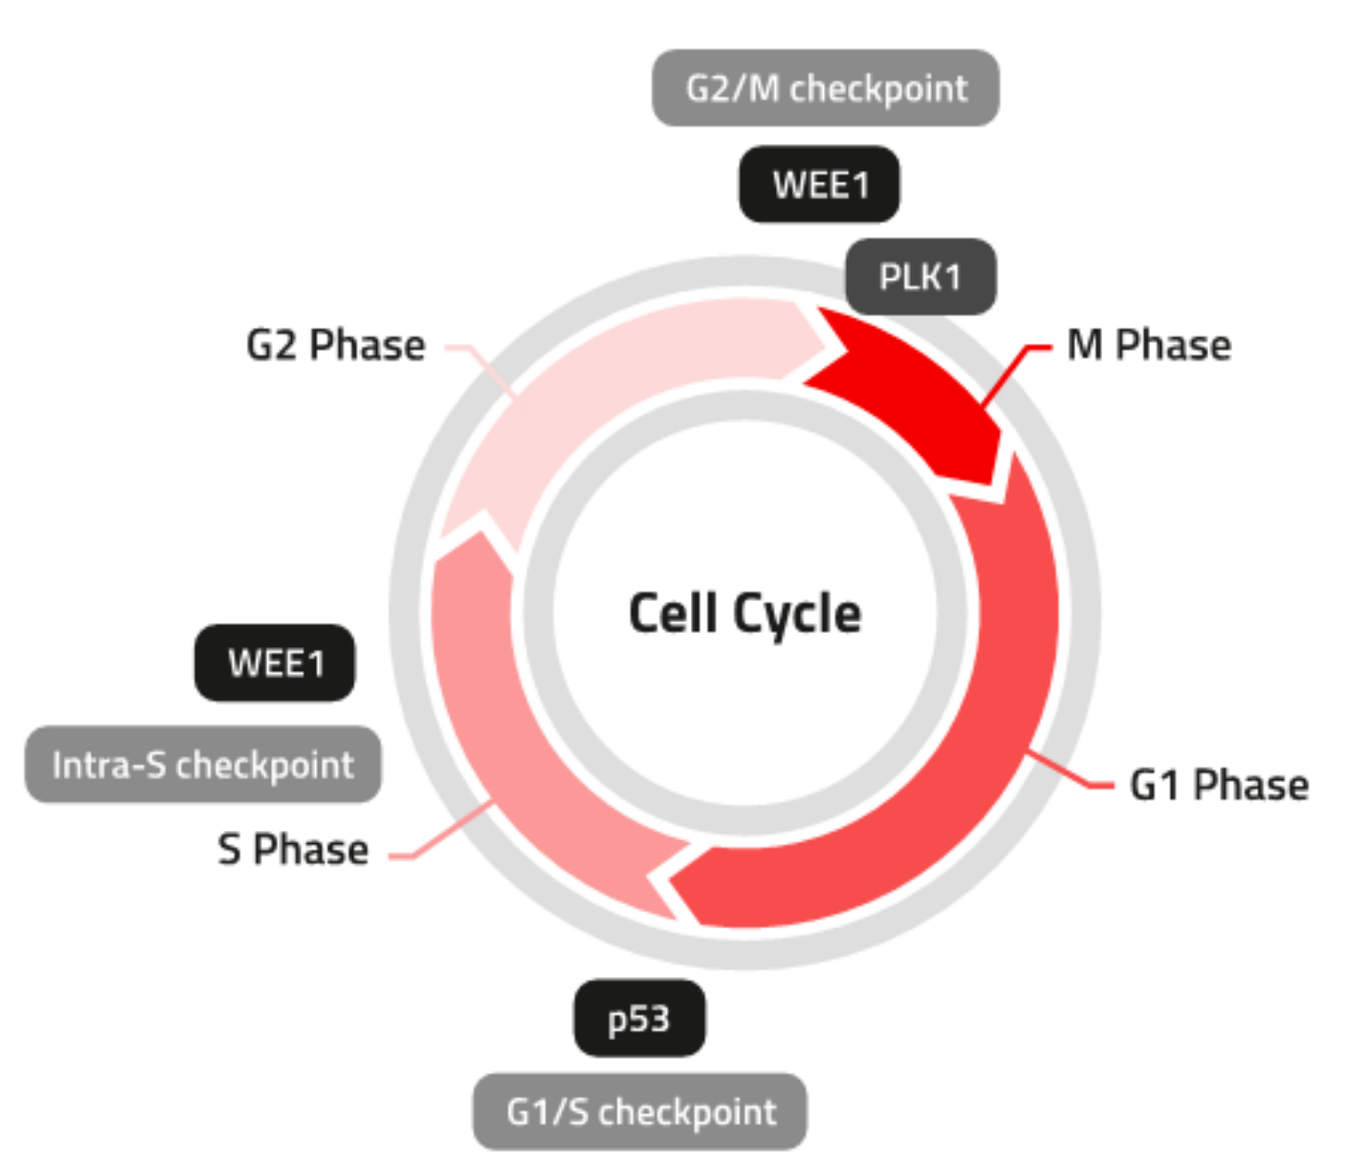
G2/M checkpoint
WEE1
PLK1
G2 Phase
M Phase
Cell Cycle
WEE1
Intra-S checkpoint
S Phase
G1 Phase
p53
G1/S checkpoint

B

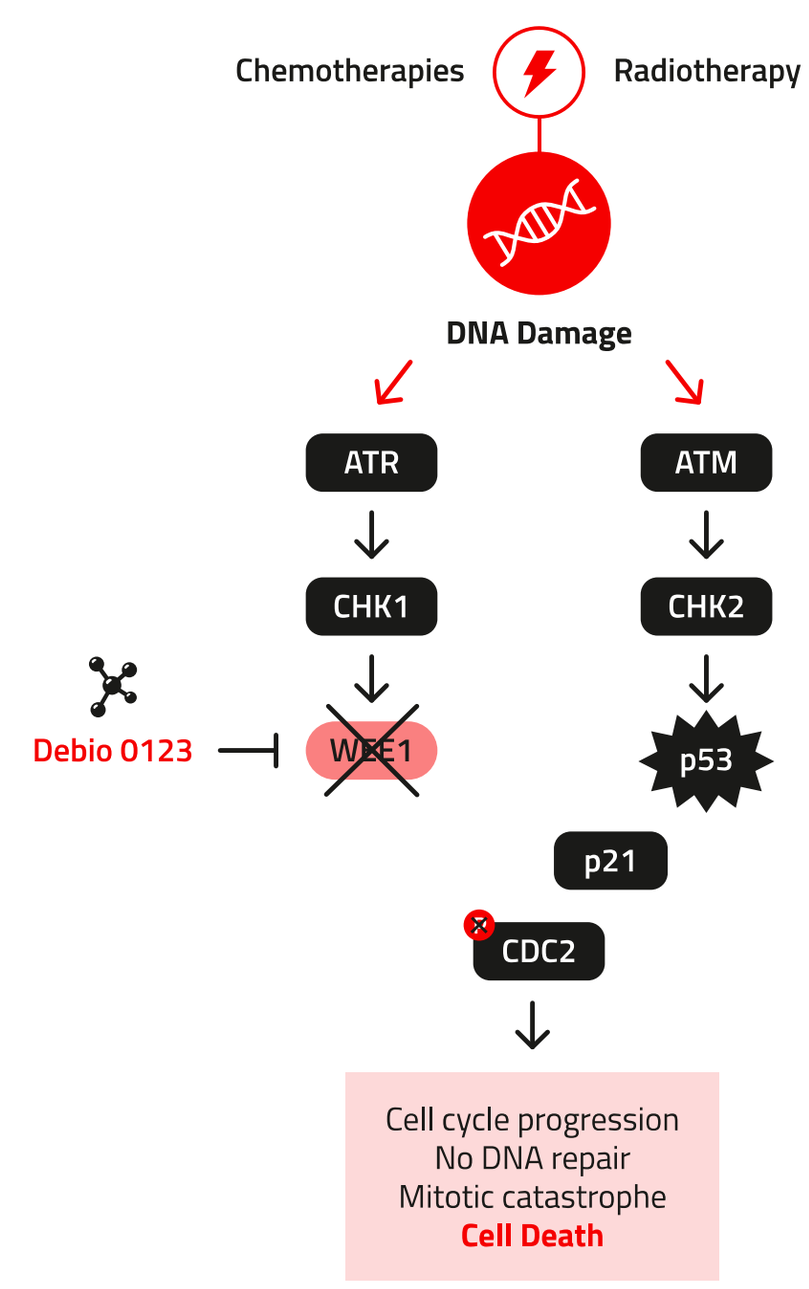
Chemotherapies
Radiotherapy
DNA Damage
ATR
ATM
CHK1
CHK2
Debio 0123
WEE1
p53
p21
CDC2
Cell cycle progression
No DNA repair
Mitotic catastrophe
Cell Death

Figure 1. (A) Cell cycle checkpoints. In cancer cells, DDR pathways are often upregulated due to genomic instability. Wee1 is a key regulator of the G2/M and S-phase checkpoints where it leads to cell cycle arrest allowing DNA damage repair. **(B) WEE1 inhibition.** Inhibition of WEE1 reduces the phosphorylation of CDC2 (CDK1) permitting cells to proceed through the cell cycle with an accumulation of DNA damage leading to mitotic catastrophe and ultimately cell death.

WEE1 acts at both the G2/M and S-phase checkpoints making WEE1 inhibition amenable to combination with multiple chemotherapies with different mechanism of actions. Debio 0123 is a highly selective and potent WEE1 inhibitor[2]. Compared to AZD1775, Debio 0123 does not inhibit PLK1 or PLK2[3].

High potency and selectivity

IC_{50} on WEE1: 0.8nM
Selective against 450 kinases (500nM)

More selective than competition

Target	AZD1775 IC_{50} (nM)	Debio 0123 IC_{50} (nM)
WEE1	43	41
PLK1	79	> 10 000
PLK2	79	> 10 000

Studies conducted using versions of AZD1775 synthesized by third-party contract research chemists, using publicly available information

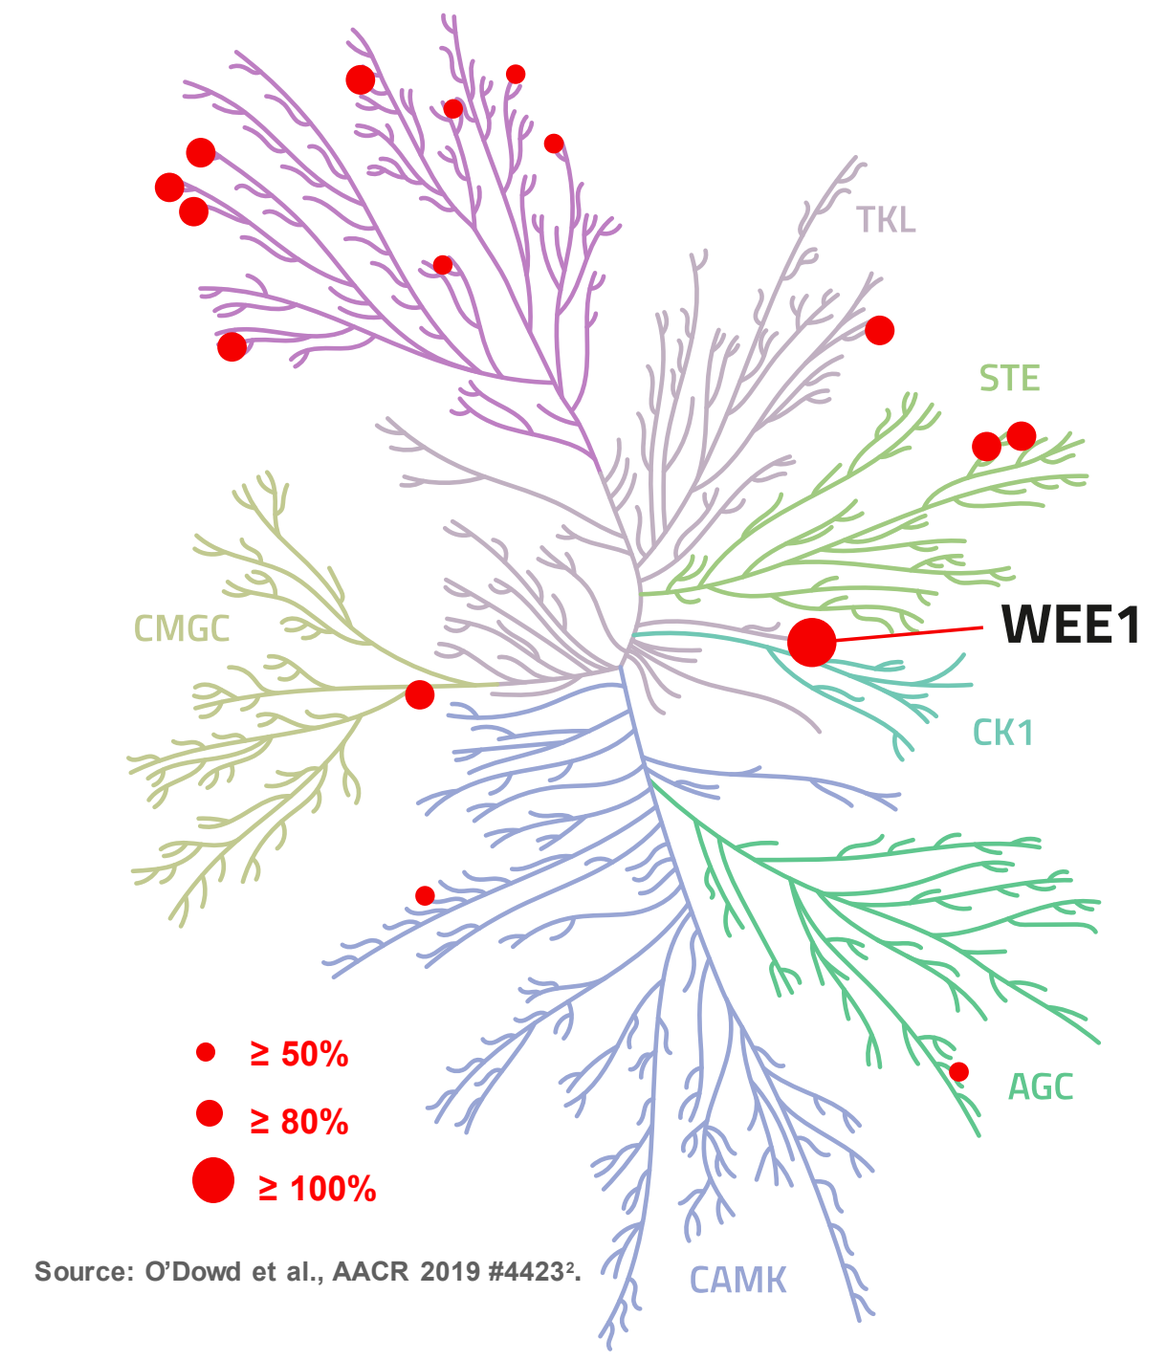
TKL
STE
WEE1
CMGC
CK1
AGC
CAMK
≥ 50%
≥ 80%
≥ 100%

Source: O'Dowd et al., AACR 2019 #4423[2].

Figure 2. Cell free potency and selectivity profile screening at fixed concentration of 500nM (>100-fold IC_{50} WEE1)

METHODS

These studies were conducted in accordance with institutional guidelines and NCRI Guidelines for the welfare and use of animals in cancer research[4].

***In vitro* cytotoxicity**: NCI-H446 tumor cells were plated at a density of 5000 cells/90ul in 96 well plates. Cells were treated with Debio 0123 and carboplatin, either alone or in combination at 9 different doses (3-fold serial dilutions starting at 10µM for Debio 0123 and 300µM for carboplatin) for 72 hours. Cell viability was then assessed by CellTiter-Glo viability assay and synergy of drug combination at each dose calculated using Bliss synergy and Loewe additivity methods.

Mouse xenograft models: Female Balb/c nude mice were inoculated subcutaneously with 5 x 10^6 NCI-H1048 SCLC tumor cells. Tumor size was measured using a caliper twice per week and tumor volume (TV) was calculated using the following formula: TV = $ab^2/2$, where a is the length of the tumor, and b is the width. Debio 0123 was orally administered once a day for up to 28 consecutive days (30mg/kg QD), carboplatin was administered once a week for up to 3 weeks (50mg/kg QW) and etoposide was administered once a week for up to 3 weeks (25mg/kg or 12.5mgkg QW) in mice with established tumors (mean TV = 100-150mm^3).

PDX mouse model: Female athymic nude mice were inoculated subcutaneously with tumor fragments from SC6 PDX tumors. Tumor size was measured using a caliper twice per week and tumor volume (TV) was calculated using the following formula: TV = $ab^2/2$, where a is the length of the tumor, and b is the width. Debio 0123 was orally administered once a day for up to 28 consecutive days (30mg/kg QD) and etoposide was administered twice a week for up to 3 weeks (6mg/kg 3 days on 4 days off) in mice with established tumors (mean TV = 100-150mm^3).

RESULTS

Debio 0123 demonstrates synergy with carboplatin *in vitro* at a broad range of doses

Treatment of NCI-H446 monolayer cultures with Debio 0123 and carboplatin resulted in IC_{50} values of 2.5µM and 50.9µM respectively. Increased sensitivity to combination treatment was observed with an IC_{50} at a dose of 1µM Debio 0123 + 10µM Carboplatin. Bliss synergy and Loewe additivity analysis demonstrated that synergy was observed at a broad range of Debio 0123 and carboplatin doses. Similar synergy was also observed in vitro with etoposide.

A

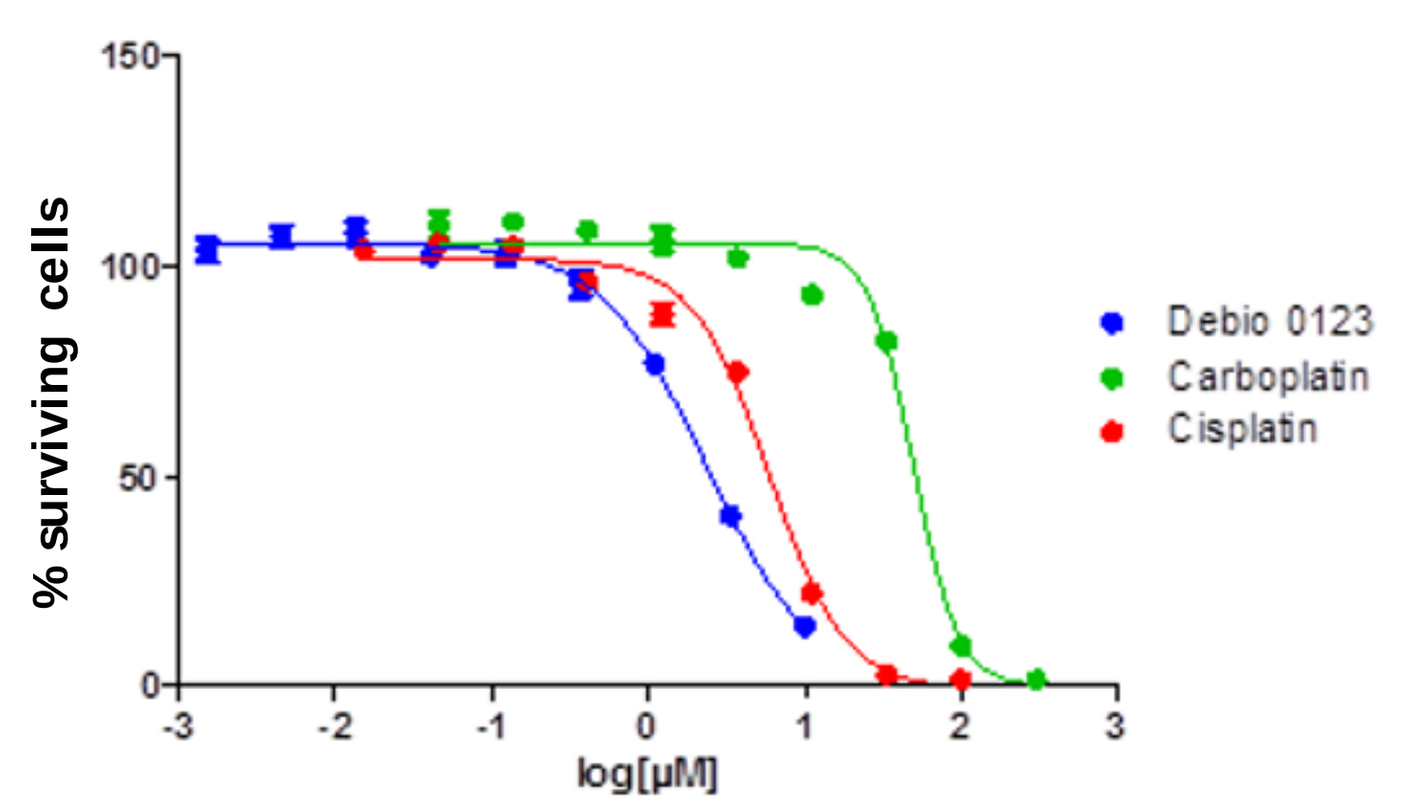
% surviving cells
log[µM]
Debio 0123
Carboplatin
Cisplatin

Absolute IC50 (µM)	
Debio 0123	2.537
Carboplatin	50.905
Cisplatin	5.967

B

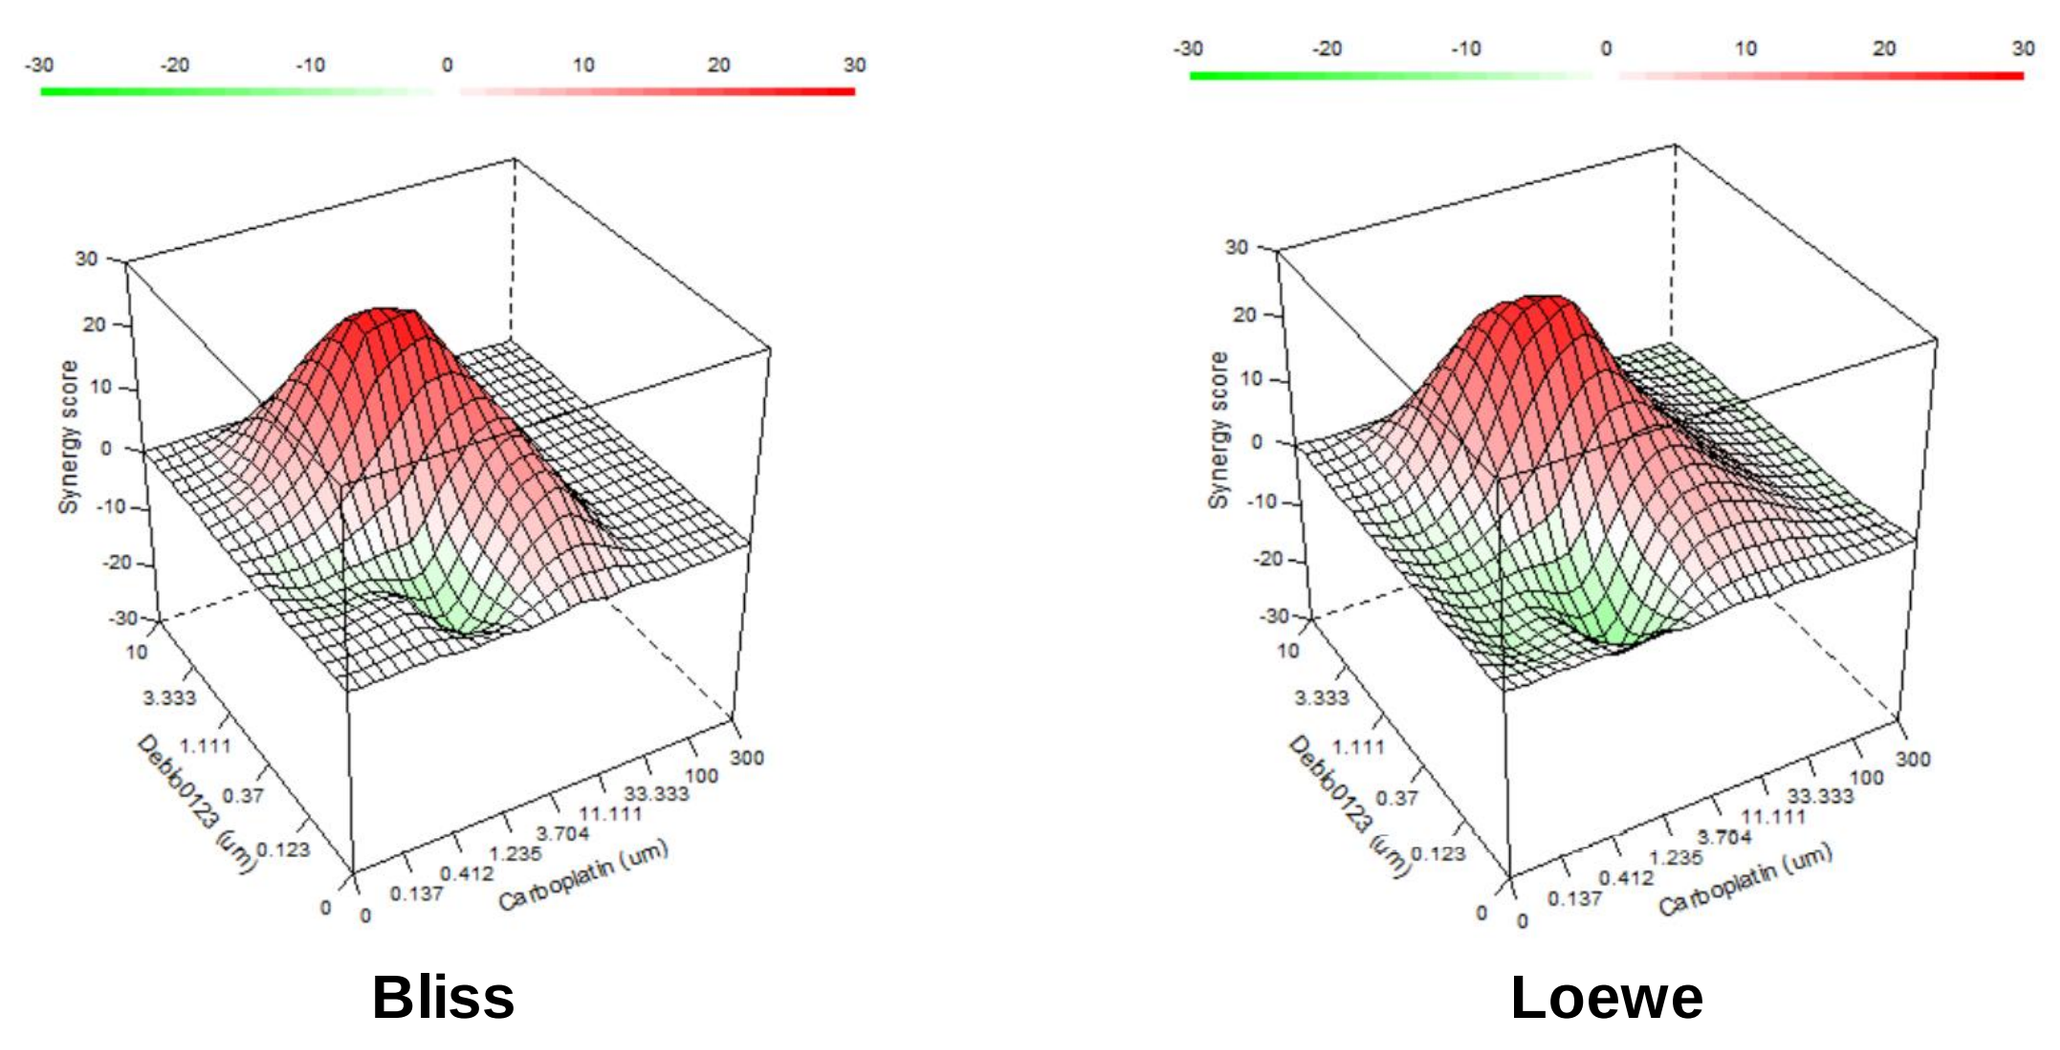
Synergy score
Debio0123 (um)
Carboplatin (um)
Bliss
Loewe

Figure 3. Debio 0123 demonstrates synergy with carboplatin in SCLC cell line. (A) Absolute IC_{50}s for Debio 0123 and carboplatin monotherapy in NCI-H446 SCLC cell line. **(B)** Bliss synergy and Loewe additivity assessment of Debio 0123 in combination with carboplatin in NCI-H446 SCLC tumor cell line.

Debio 0123 improves SCLC response to carboplatin or etoposide *in vivo*

Debio 0123 in combination with carboplatin induced significant anti-tumor effects across 2 different cell line-derived xenograft (CDX) models of SCLC. In the NCI-H466 model, combination treatment resulted in a significant tumor growth inhibition (TGI 67% vs vehicle, p<0.05) and significantly improved TGI compared to single agent Debio 0123 or carboplatin (TGI 59%, p<0.05). In the NCI-H1048 model, a moderate but significant single agent activity was observed for both Debio 0123 (TGI 54%, p<0.05) and carboplatin and combination treatment resulted in significant antitumor activity (TGI 68% vs vehicle, p<0.01) but represented a non-significant improvement over either monotherapy.

Debio 0123 in combination with etoposide resulted in significant anti-tumor activity in both a CDX (TGI 78% vs vehicle, p<0.01) and PDX (TGI 61% vs vehicle, p<0.05) model of SCLC. In the NCI-H1048 model, combination treatment also resulted in significantly improved tumor response when compared to either monotherapy treatment (TGI 60% vs Debio 0123 and 77% vs etoposide, p<0.01).

In each study all monotherapy and combination therapies were well tolerated.

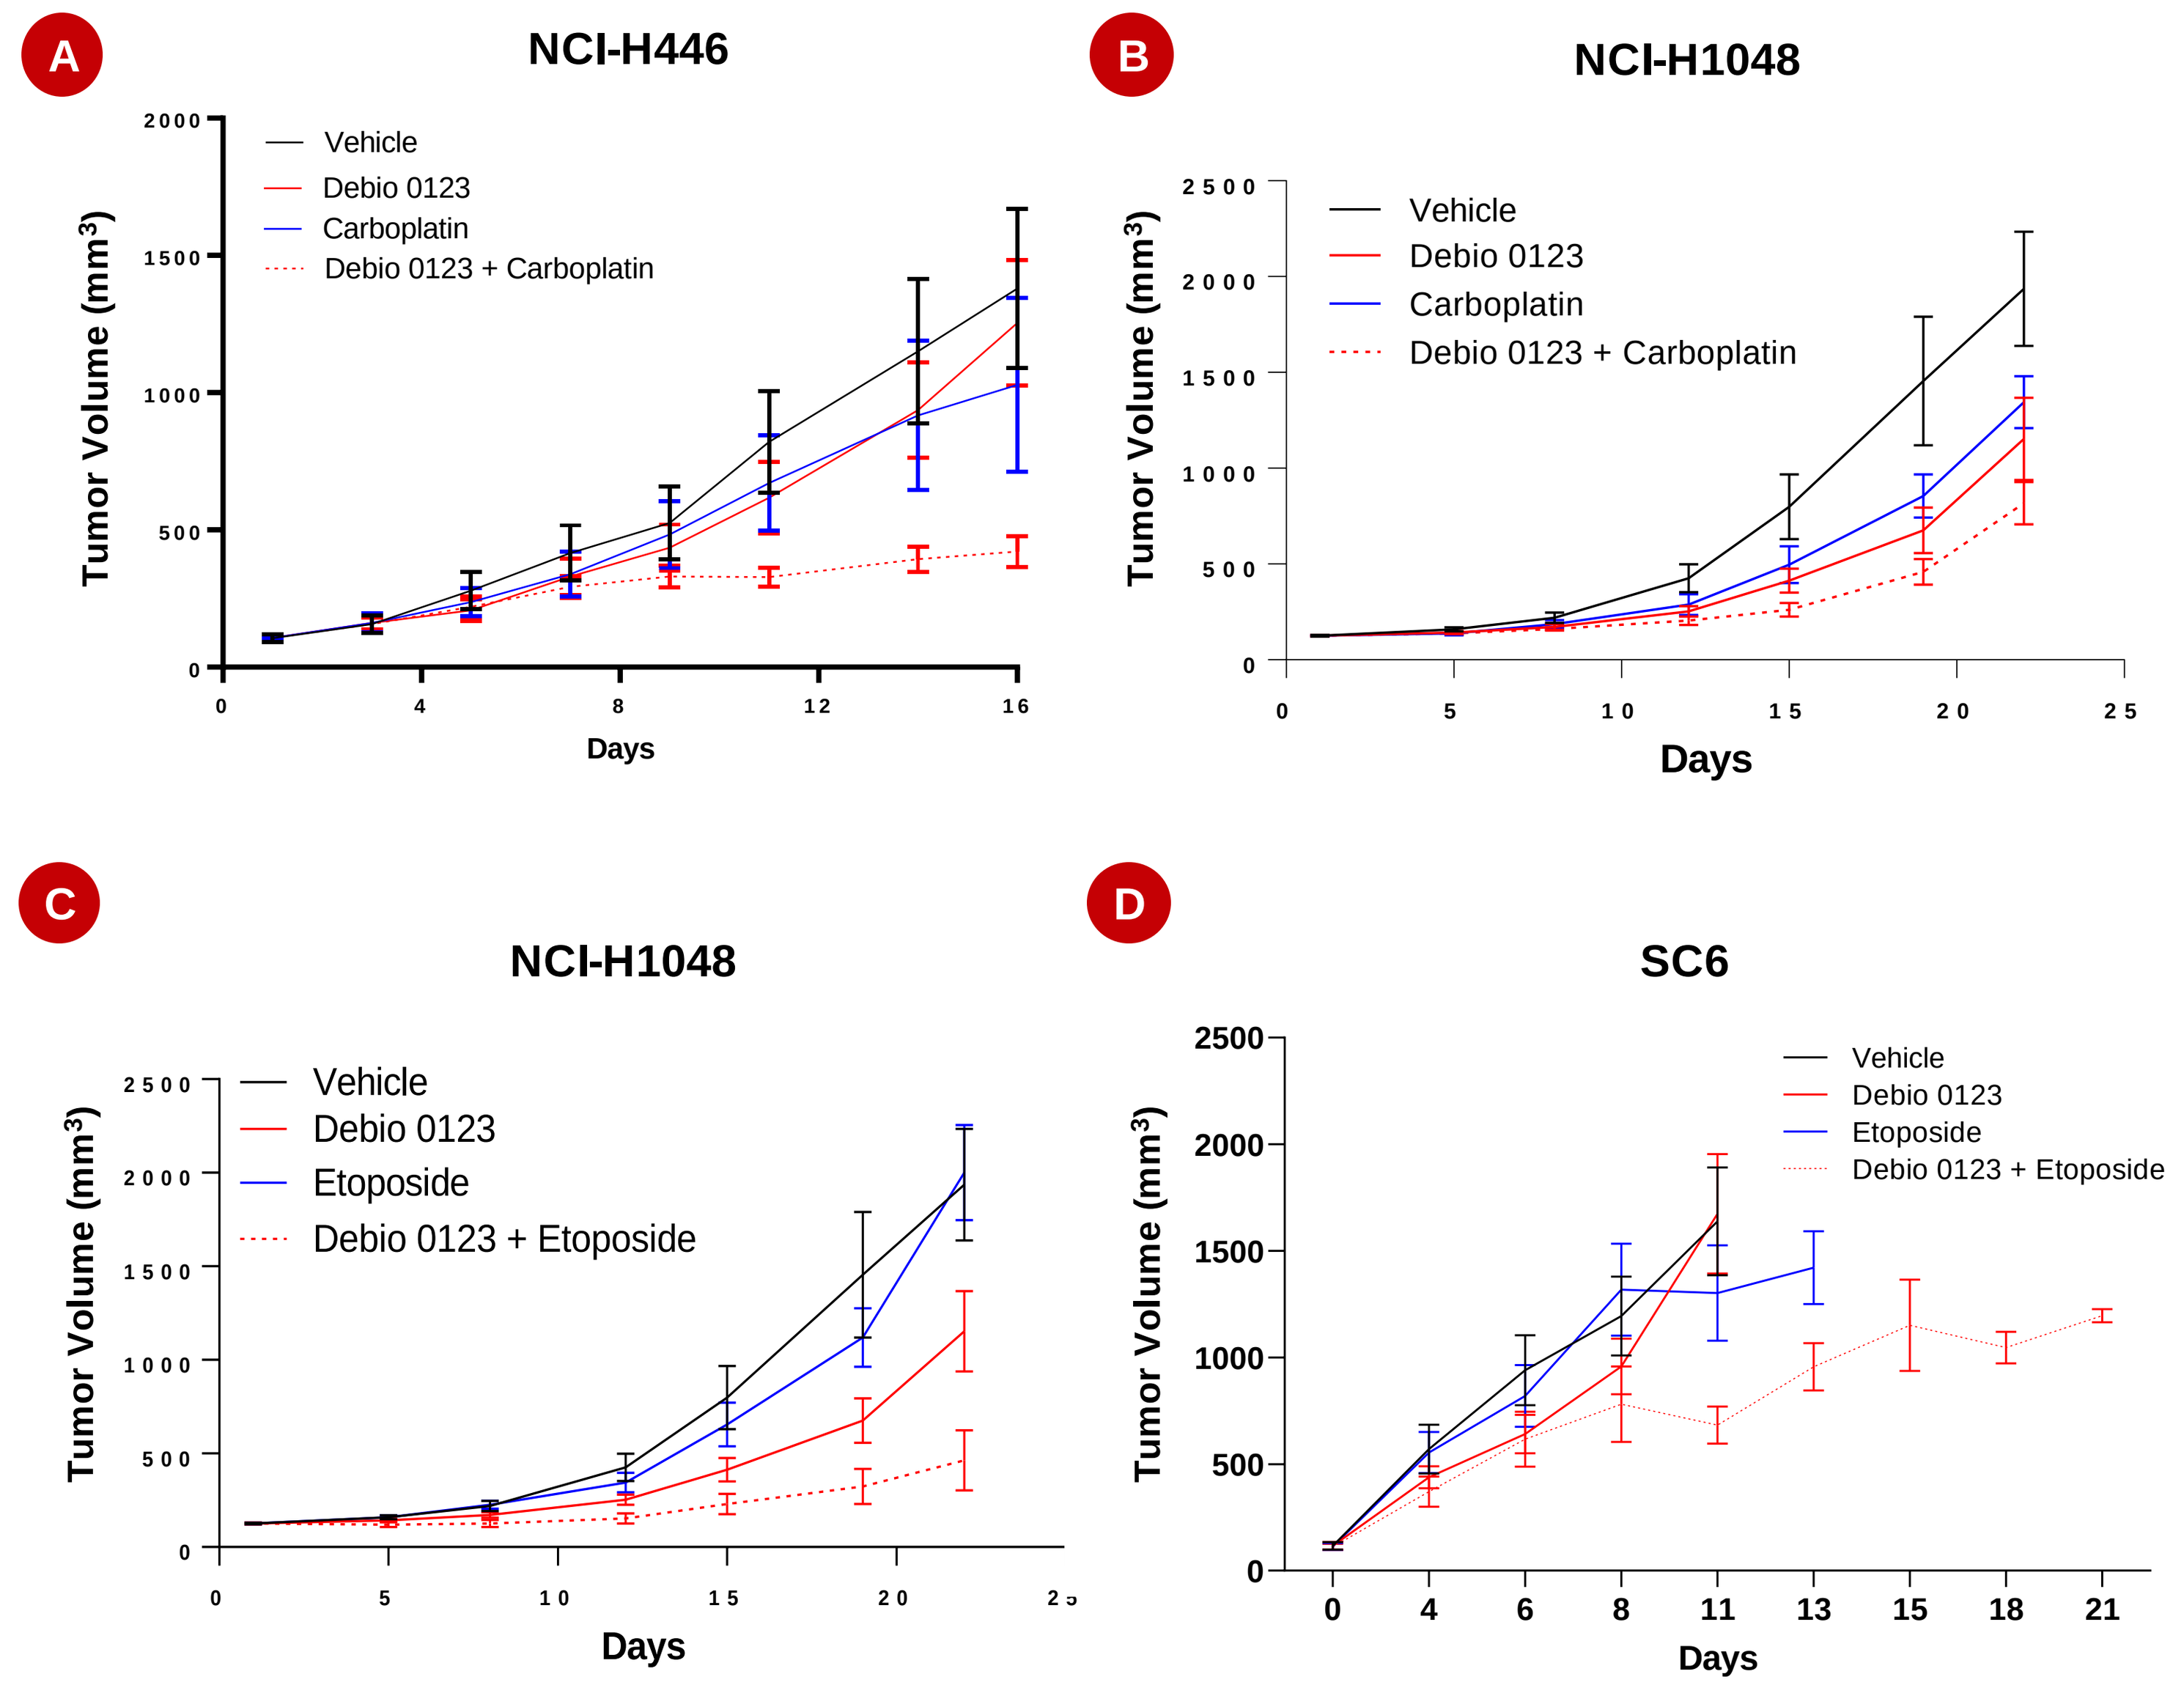
A
NCI-H446
B
NCI-H1048
Vehicle
Debio 0123
Carboplatin
Debio 0123 + Carboplatin
C
NCI-H1048
D
SC6
Etoposide
Debio 0123 + Etoposide
Tumor Volume (mm³)
Days

Figure 4. Debio 0123 demonstrates potent anti-tumor activity in combination with carboplatin or etoposide. (A) NCI-H446 SCLC CDX tumors treated with 30mg/kg QD Debio 0123, 50mg/kg QW carboplatin or in combination (n=5). **(B)** NCI-H1048 SCLC CDX tumors treated with 30mg/kg QD Debio 0123, 50mg/kg QW carboplatin or in combination (n=8). **(C)** NCI-H1048 SCLC CDX tumors treated with 30mg/kg QD Debio 0123, 25mg/kg QW etoposide or in combination (n=8). **(D)** SC6 SCLC PDX tumors treated with 30mg/kg QD Debio 0123, 6mg/kg QW 3 on 4 off etoposide or in combination (n=7).

CLINICAL TRIALS

Debio 0123 is currently under phase I clinical investigation as a monotherapy (NCT05109975) and in combination with carboplatin in patients with advanced solid tumors (NCT03968653).

REFERENCES

(1) Do K. et al., Cell Cycle. 2013 Oct 1;12(19):3159-64
(2) O'Dowd et al., Antitumor activity of the novel oral highly selective WEE1 inhibitor Debio 0123, AACR 2019 abstract #4423
(3) Workman et al., British Journal of cancer. (2010) 102, 1555-1577

Debio 0123 in combination with carboplatin and etoposide significantly improves SCLC tumor response *in vivo*

The NCI-H1048 model was used to further evaluate the efficacy and tolerability of Debio 0123 in combination with carboplatin and etoposide as a triple combination. Debio 0123 monotherapy resulted in a moderate anti-tumor response and SOC carboplatin/etoposide treatment resulted in a significant inhibition of tumor growth. Triple combination of Debio 0123, carboplatin and etoposide resulted in a significant anti-tumor response compared to vehicle (TGI 72%, p<0.01), Debio 0123 monotherapy (TGI 68%, p<0.01) and carboplatin/etoposide (TGI 44%, p<0.01) treatments. All treatments were well tolerated with no significant body weight loss in the treated animals.

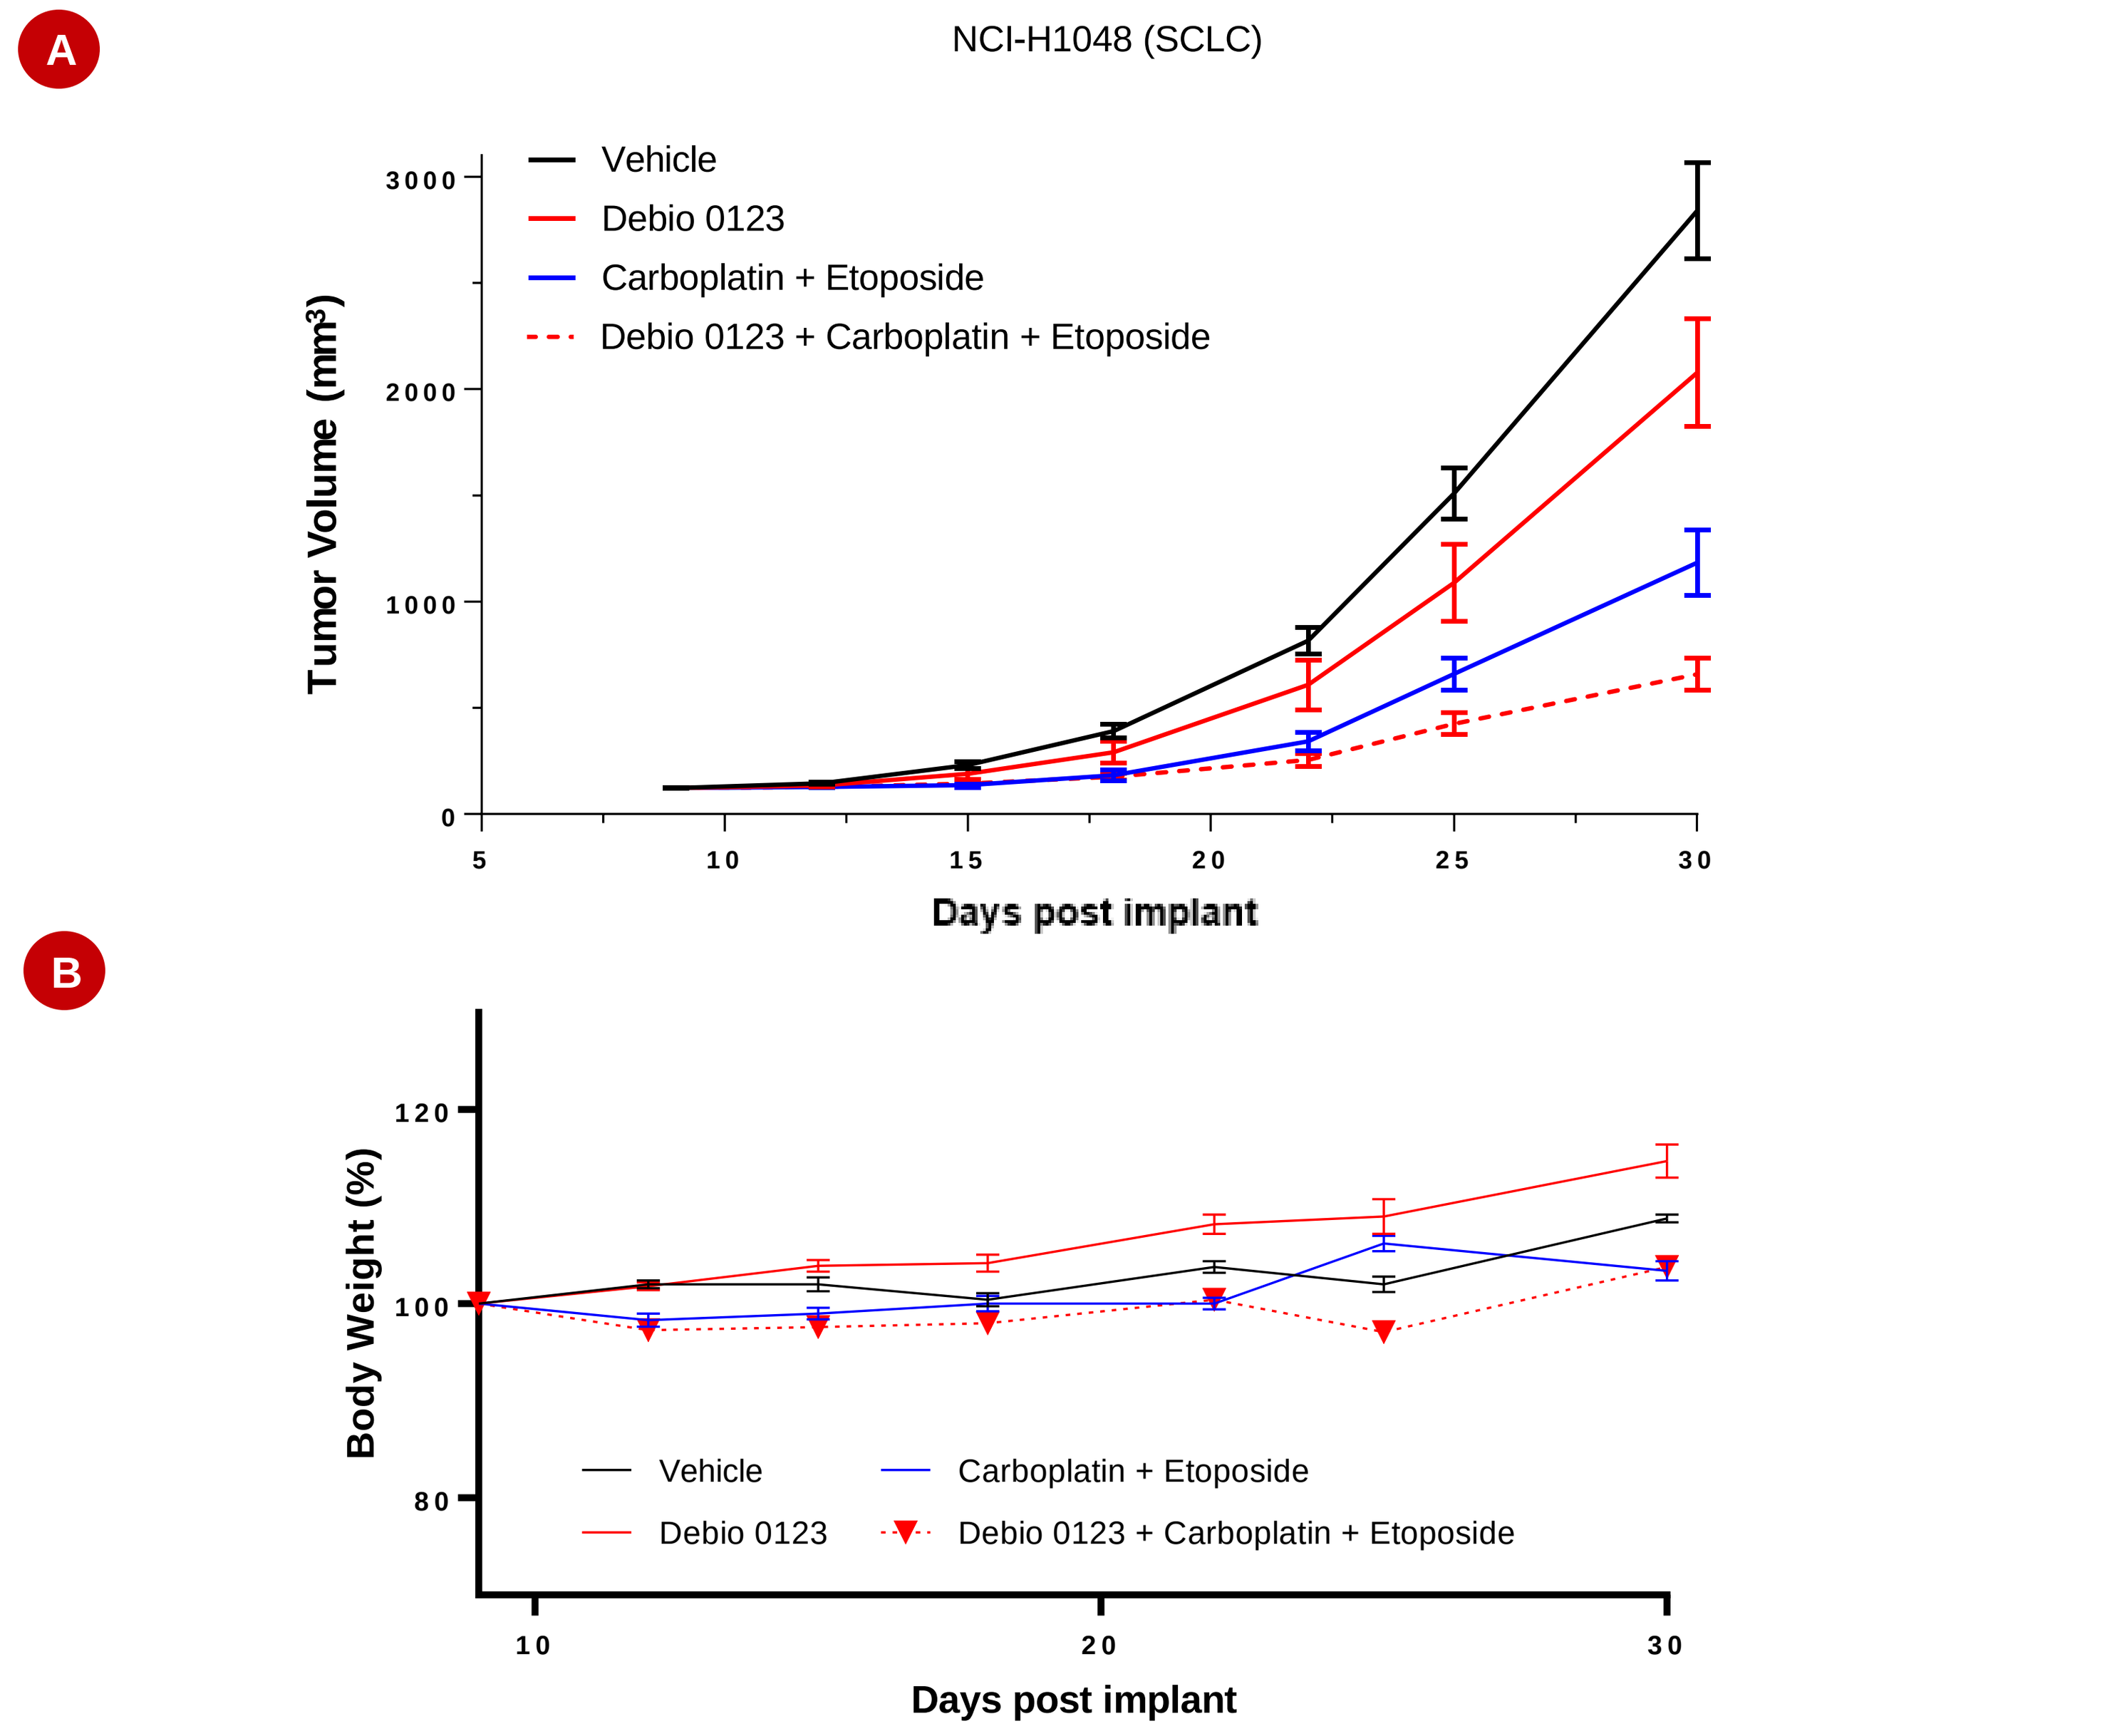
A
NCI-H1048 (SCLC)
Vehicle
Debio 0123
Carboplatin + Etoposide
Debio 0123 + Carboplatin + Etoposide
Tumor Volume (mm³)
Days post implant
B
Body Weight (%)
Days post implant

Figure 5. Debio 0123 shows potent antitumor efficacy *in vivo* in combination with carboplatin/etoposide. (A) NCI-H1048 tumors were treated with 30mg/kg QD Debio 0123, 12.5mg/kg QW etoposide/ 50mg/kg QW carboplatin alone or in combination (n=10). **(B)** Relative mean body weight during treatment. All treatments in all groups were well tolerated with no significant body weight loss recorded.

CONCLUSIONS

- Debio 0123 demonstrates synergistic anti-tumor effects in combination with carboplatin *in vitro* in SCLC cell lines.
- Debio 0123 enhances the anti-tumor effects of both carboplatin and etoposide *in vivo* in a PDX and CDX models of SCLC.
- Debio 0123 significantly improves response to standard of care carboplatin/etoposide treatment *in vivo* in a model of SCLC and is well tolerated, providing the foundation for future clinical exploration of Debio 0123 in SCLC.
- Debio 0123 may improve responses to standard of care therapy carboplatin/etoposide in patients with SCLC.

CONTACT

Debiopharm International S.A., Lausanne, Switzerland.
www.debiopharm.com
Luke.piggott@debiopharm.com

DOWNLOAD

This poster is available via:
www.debiopharm.com/medias/publications