[177Lu]Lu-DPI-4452 & [68Ga]Ga-DPI-4452, a new radiopeptide tandem targeting Carbonic Anhydrase IX displays strong theranostic potential in CRC and ccRCC tumors

Norbert Wiedemann[1], Frédéric Massière[1], Franck Brichory[1], Aileen Hoehne[2], Frank Osterkamp[2], Inês Borrego[1], Antoine Attinger[1]

[1]Debiopharm International SA, Chemin Messidor 5-7, CH-1002 Lausanne, Switzerland. [2] 3B Pharmaceuticals GmbH, Magnusstrasse 11, D-12489 Berlin, Germany

Debiopharm™ WE DEVELOP FOR PATIENTS

POSTER #4048

INTRODUCTION

- The transmembrane metalloprotease Carbonic Anhydrase IX (CAIX) represents an attractive diagnostic and therapeutic target in hypoxic solid tumors such as Colorectal Cancer (CRC) and clear cell Renal Cell Carcinoma (ccRCC), with limited expression in healthy organs
- DPI-4452 is a CAIX-targeting cyclic peptide with a DOTA cage that can be labelled with different radionuclides for theranostic purposes
- Here we report CAIX expression in CRC and ccRCC human tumor samples and evaluate for the first time the *in vitro* and *in vivo* therapeutic and diagnostic performance of [177Lu]Lu-DPI-4452 and [68Ga]Ga-DPI-4452

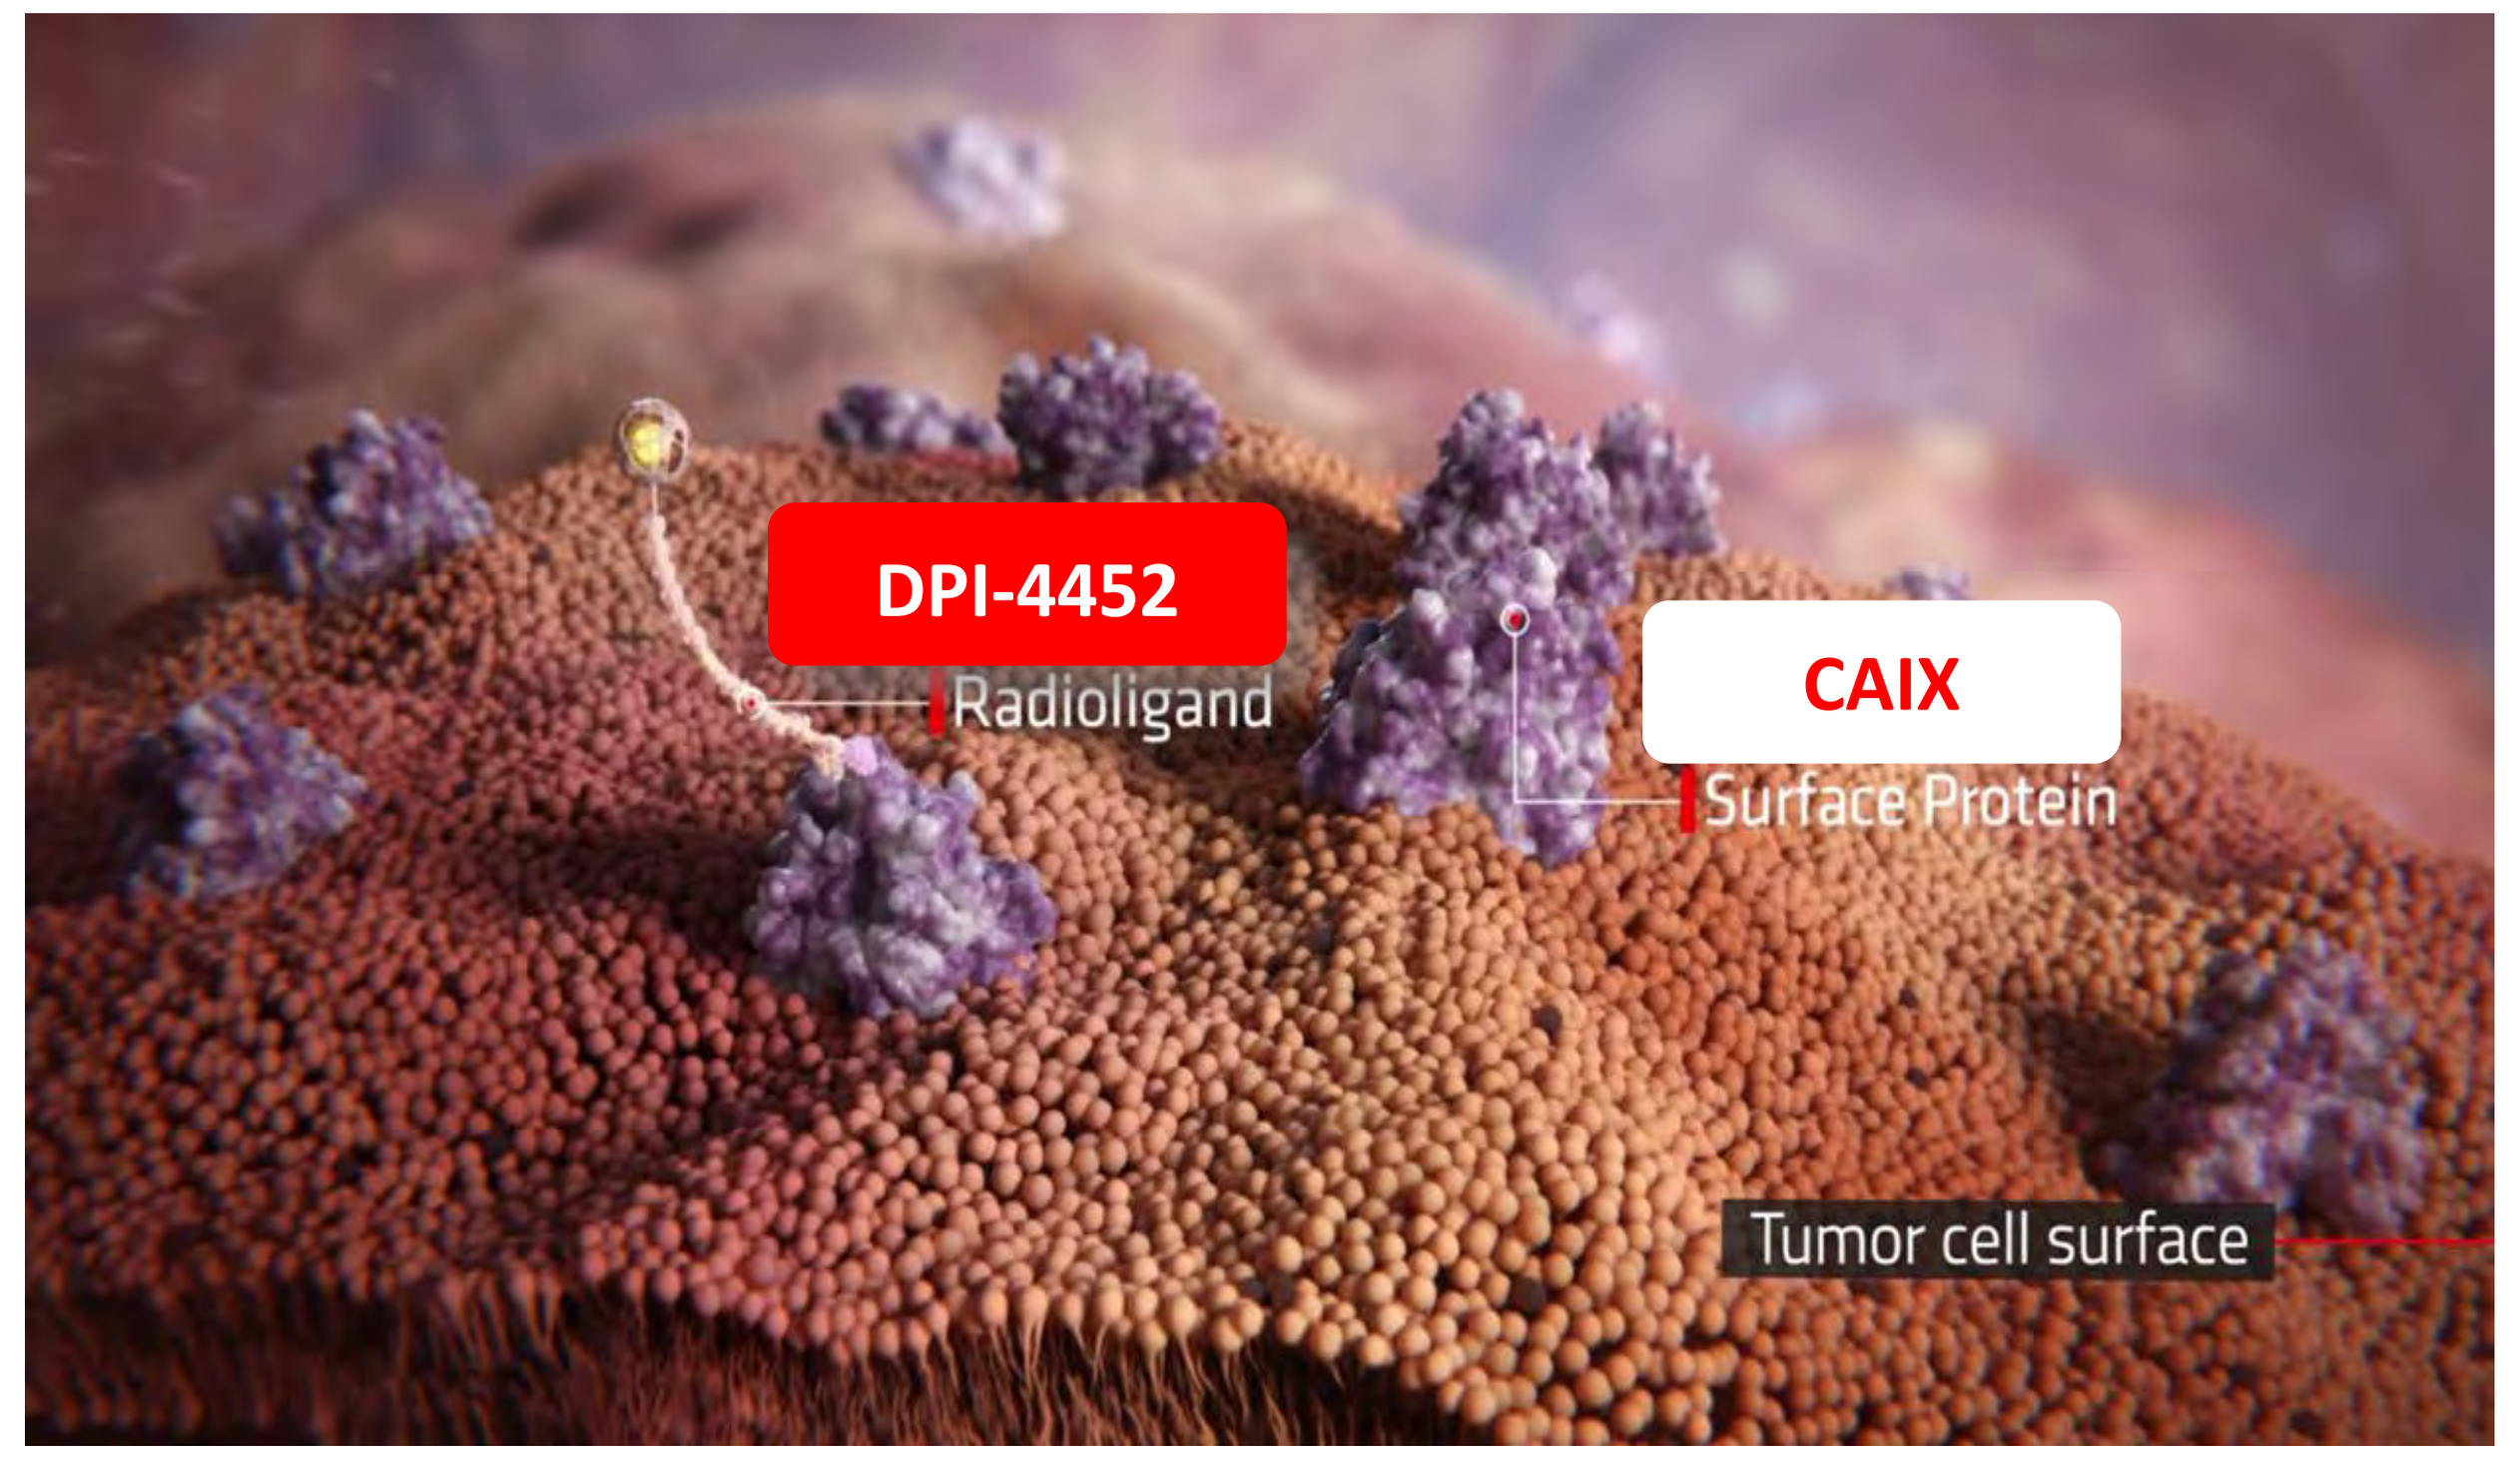
DPI-4452
Radioligand
CAIX
Surface Protein
Tumor cell surface

MATERIALS AND METHODS

Immunohistochemistry (IHC). Formalin-fixed paraffin-embedded tissue microarrays (US Biomax) were stained with the anti-CAIX antibody (mouse clone M75) at a 0.125µg/ml dilution. H-score were assessed by a pathologist.

Surface plasmon resonance (SPR): Recombinant soluble Fc-tagged human CAIX protein (Sino Biological) was captured on the sensor chip through an anti-human Fc antibody. DPI-4452 compounds were then injected on the captured CAIX and the association and the dissociation constants were measured.

In vivo efficacy study: Studies were conducted in accordance with local institutional animal welfare guidelines. CAIX-positive human cancer cell line HT-29 (CRC) or SK-RC-52 (ccRCC) were subcutaneously implanted with matrigel ($2x10^6$ cells for each model) into NMRI nude mice. Animals for each tumor type were divided in 4 treatment groups of 10 mice each: a) Single administration (day 1) of vehicle b) Single administration (day 1) of 100MBq of [177Lu]Lu-DPI-4452 c) Single administration (day 1) of 33MBq of [177Lu]Lu-DPI-4452 d) Three administrations (day 1, 8, 15) of 33MBq of [177Lu]Lu-DPI-4452. To assess the % of injected dose (ID)/gram of tissue in the tumor, kidney and liver, 3 animals per treatment group were imaged by SPECT 4h after each [177Lu]Lu-DPI-4452 administration. In both models, tumor volume and body weight were monitored for 43 days from treatment initiation, and blood sampling for hematology was performed on all animals at study day -1, 7, 14 and at study end. Kidney function markers creatinine and urea were assessed in blood on day 14 and 43 only in the SK-RC-52 model. To correlate the [68Ga]Ga-DPI-4452 signal with the [177Lu]Lu-DPI-4452 signal in tumors, a satellite group of animals received a first injection of 10MBq [68Ga]Ga-DPI-4452 (for PET imaging 1h later) followed 7 days later by an injection of 33MBq [177Lu]Lu-DPI-4452 (for SPECT imaging 4h later).

ACKNOWLEDGEMENTS

The authors would like to thank: Minerva Imaging ApS (Oelstykke, DK) for conducting the *in vivo* studies, Mosaic Laboratories (Lake Forest, USA) for conducting the IHC, Agro-Bio (La Ferté Saint Aubin, F) for conducting the SPR study and MSKCC for granting access to the SK-RC-52 cell line

RESULTS

CAIX expression in CRC and ccRCC human tumor samples

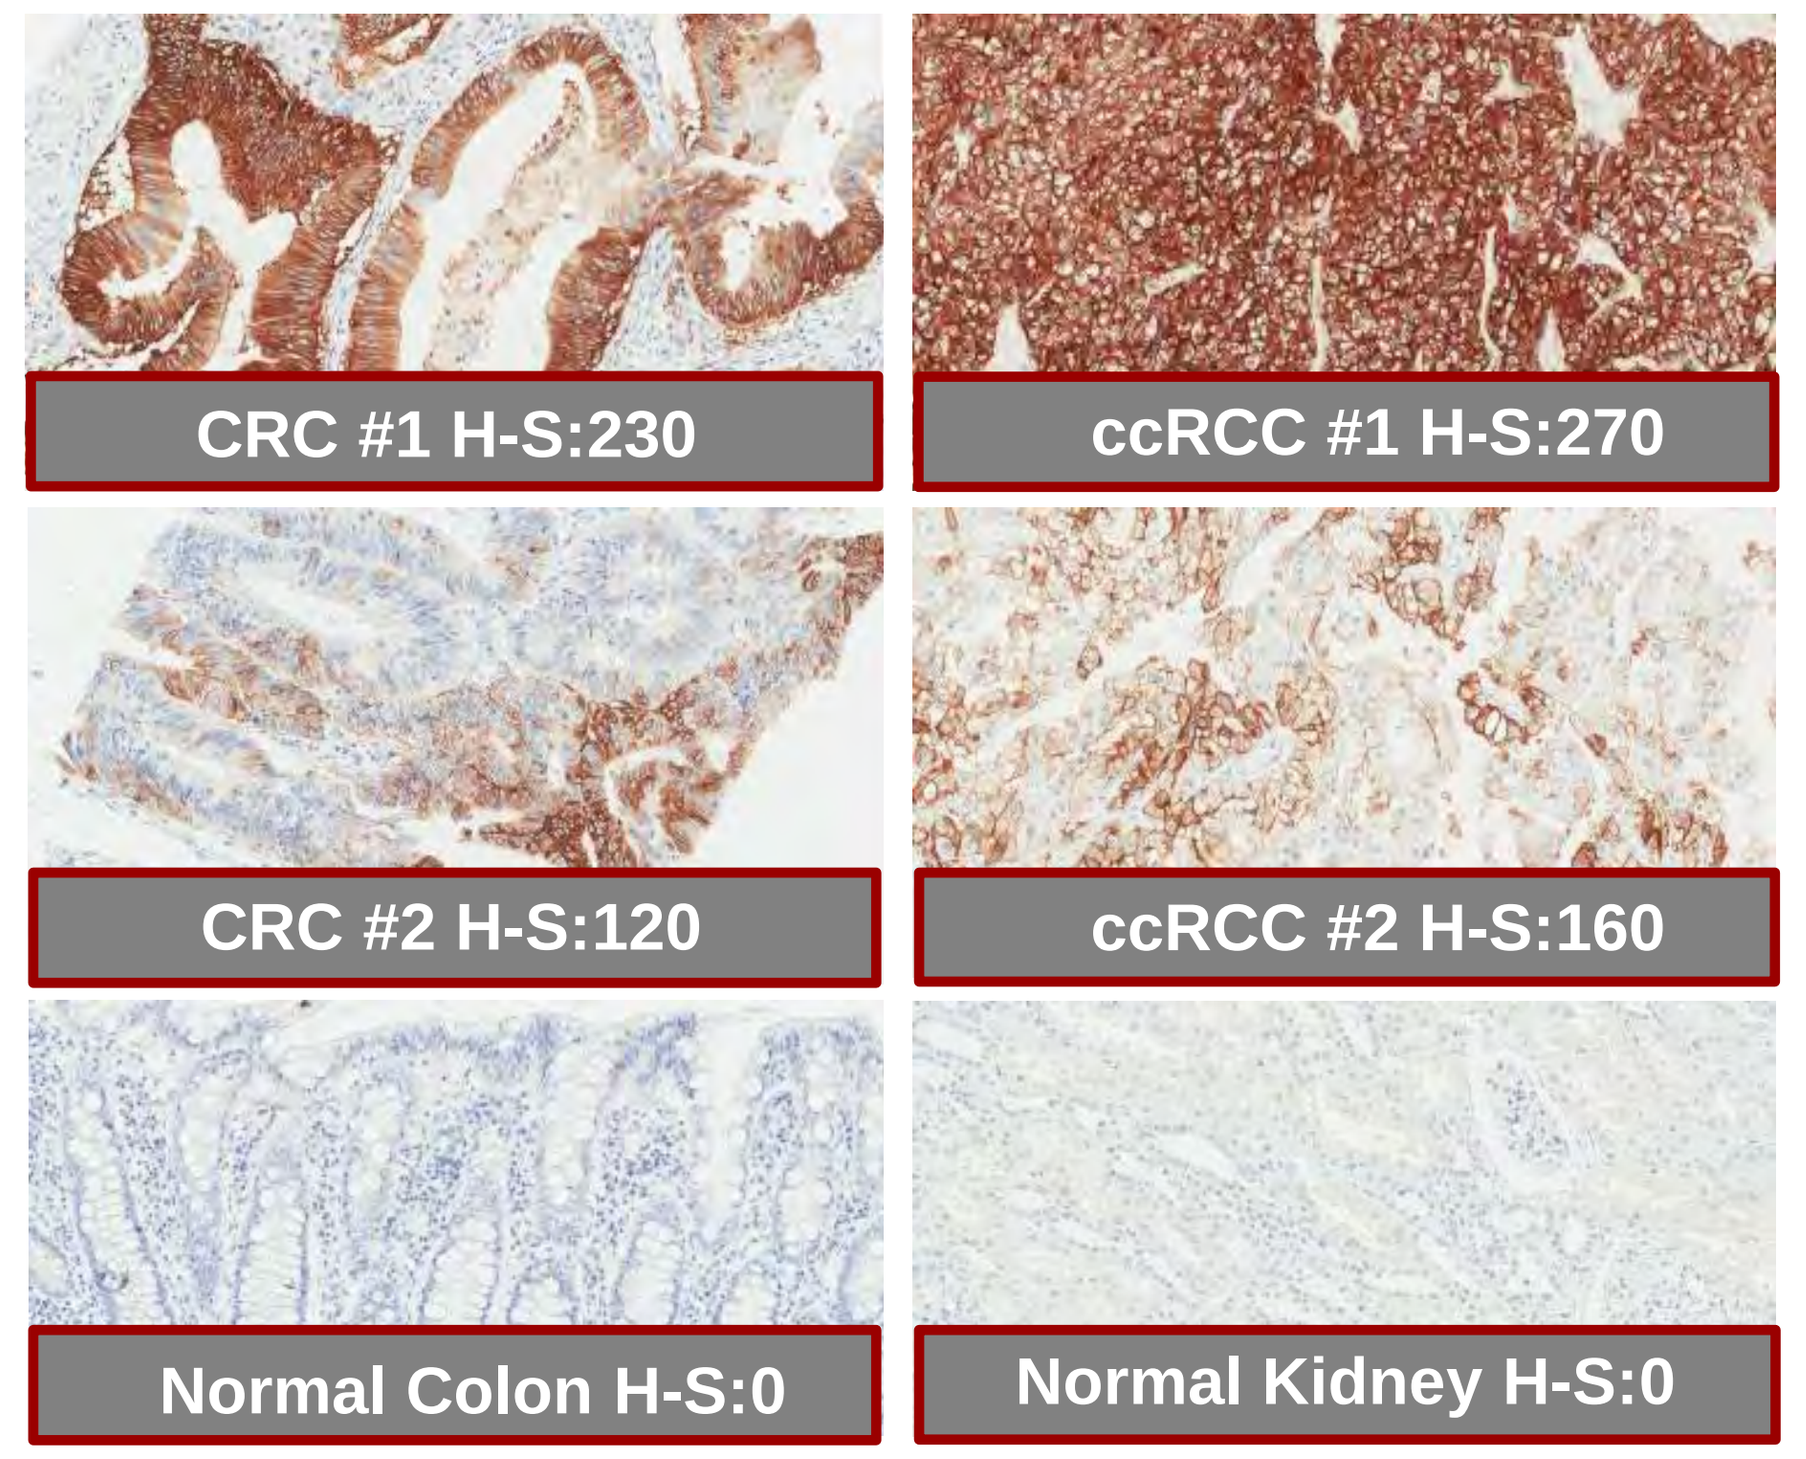
CRC #1 H-S:230
ccRCC #1 H-S:270
CRC #2 H-S:120
ccRCC #2 H-S:160
Normal Colon H-S:0
Normal Kidney H-S:0

Figure 1: Representative example of CAIX expression detected by IHC using the M75 antibody.

CRC: colorectal cancer

ccRCC: clear cell Renal cell Carcinoma

H-S: H-score (min:0 max:300)

Tissue	H-Score (max=300)	CRC	ccRCC
Malignant	> 150	29% (25/85)	83% (25/30)
	100-150	41% (35/85)	83% (25/30)
	50-100	47% (40/85)	87% (26/30)
Healthy control	> 50	0% (0/21)	0% (0/30)

Table 1: Summary of CAIX expression in CRC, ccRCC, healthy colon and healthy kidney tissue microarrays (based on IHC).

➢ **High expression of CAIX in CRC and ccRCC human tumors and no expression in healthy control tissue**

Characterisation of DPI-4452 binding to human CAIX

Compound	K_D (nM)	$T_{1/2}$ (min)
DPI-4452	0.25	99
[natLu] Lu-DPI-4452	0.16	123
[natGa] Ga-DPI-4452	0.20	112

Table 2: CAIX binding of DPI-4452 with free chelator, or DPI-4452 as complex with natLu or natGa assessed by Surface Plasmon Resonance.

K_D: dissociation constant

$T_{1/2}$: dissociation half-life

➢ **DPI-4452 binds human CAIX with high affinity and long residence time, independent of the complexation with natLu or natGa**

In vivo [177Lu]Lu-DPI-4452 uptake over weekly dosing in the HT-29 xenograft model

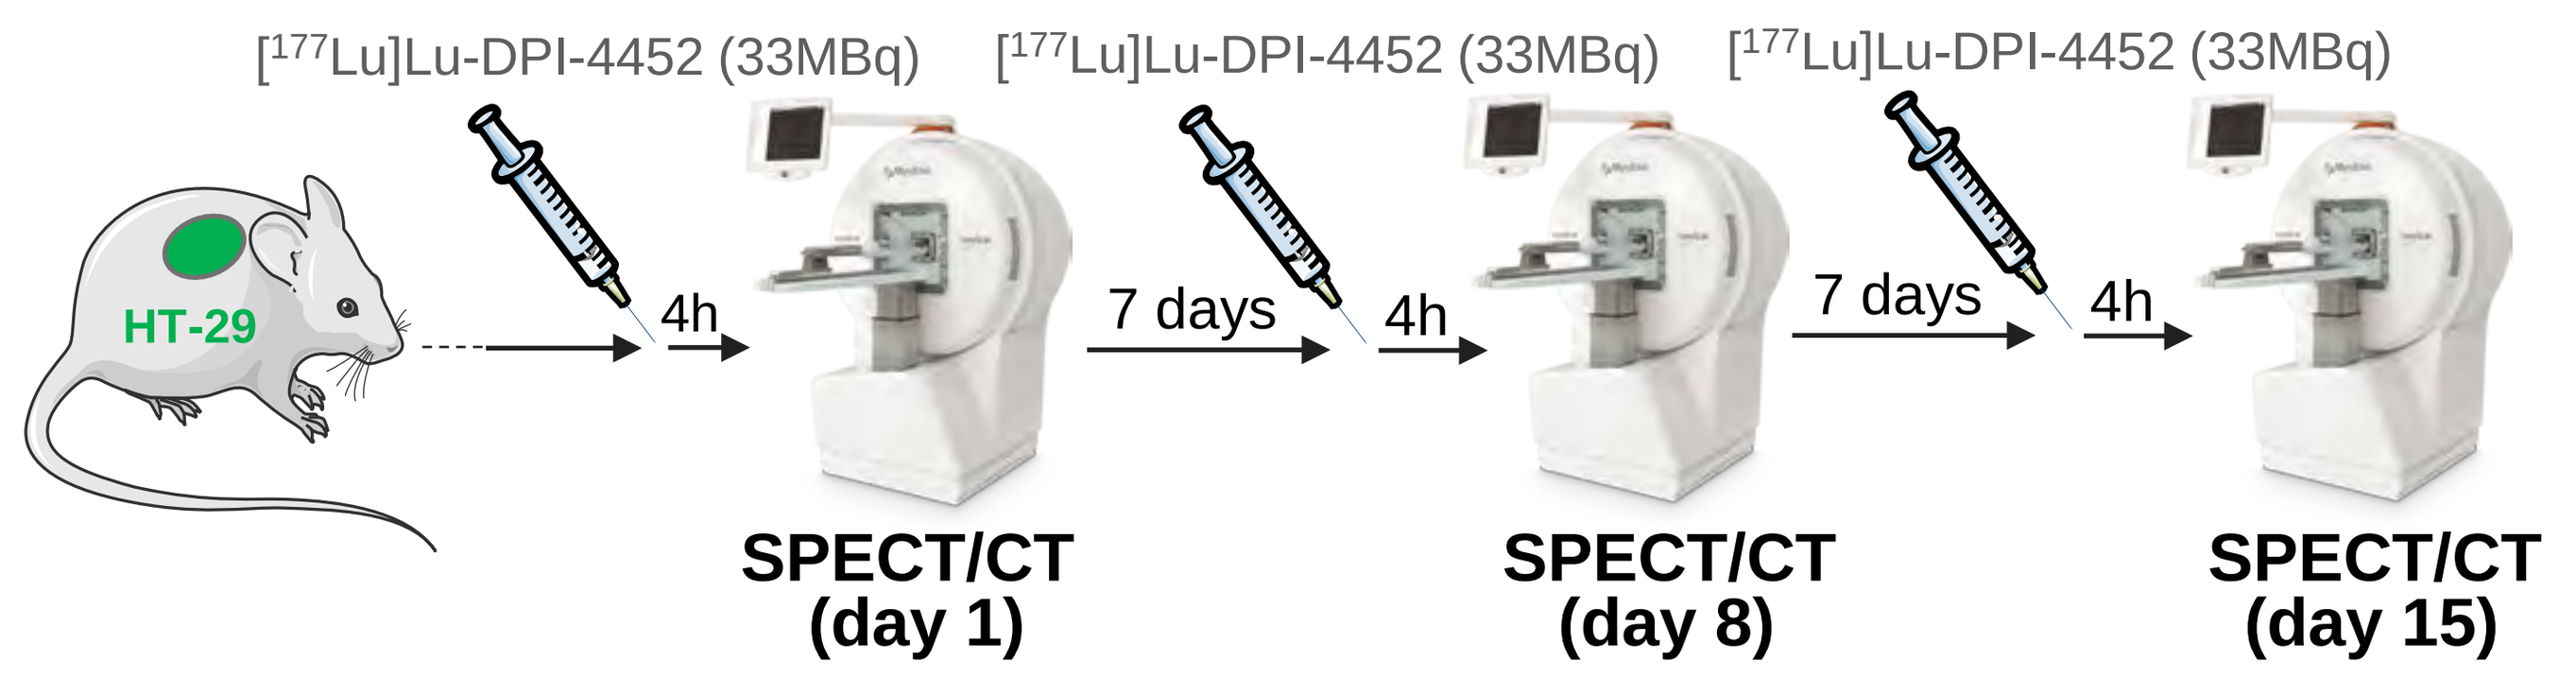
[177Lu]Lu-DPI-4452 (33MBq)
[177Lu]Lu-DPI-4452 (33MBq)
[177Lu]Lu-DPI-4452 (33MBq)
HT-29
4h
7 days
4h
7 days
4h
SPECT/CT (day 1)
SPECT/CT (day 8)
SPECT/CT (day 15)

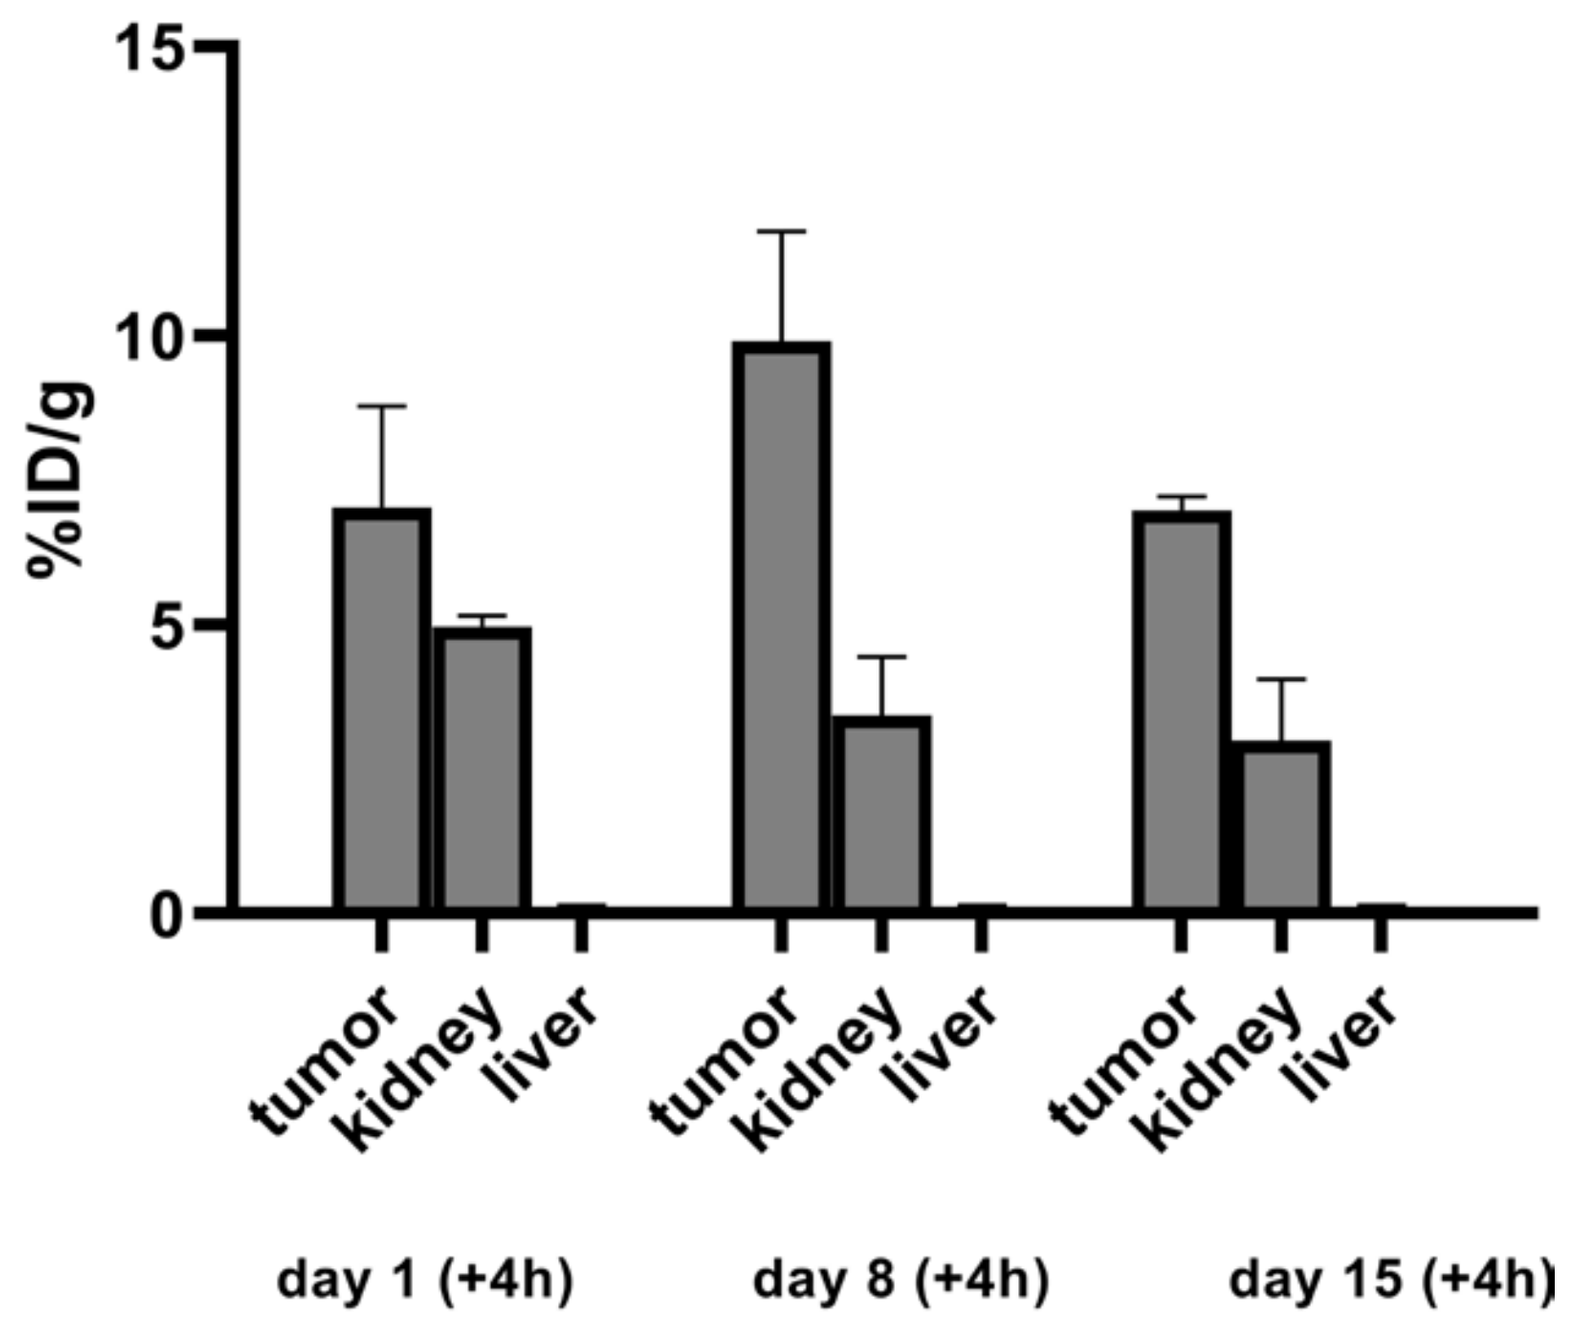
15
10
5
0
%ID/g
tumor
kidney
liver
day 1 (+4h)
day 8 (+4h)
day 15 (+4h)

Figure 2: [177Lu]Lu-DPI-4452 uptake in the tumor kidney and liver over the three weeks of dosing in the HT-29 model (comparable results were obtained in the SK-RC-52 model, not shown). N=3 animals monitored.

➢ **Stable uptake of [177Lu]Lu-DPI-4452 over the three weeks of dosing, suggesting CAIX expression is not downregulated upon treatment**

Comparison of [68Ga]Ga-DPI-4452 and [177Lu]Lu-DPI-4452 *in vivo* uptake in the SK-RC-52 model

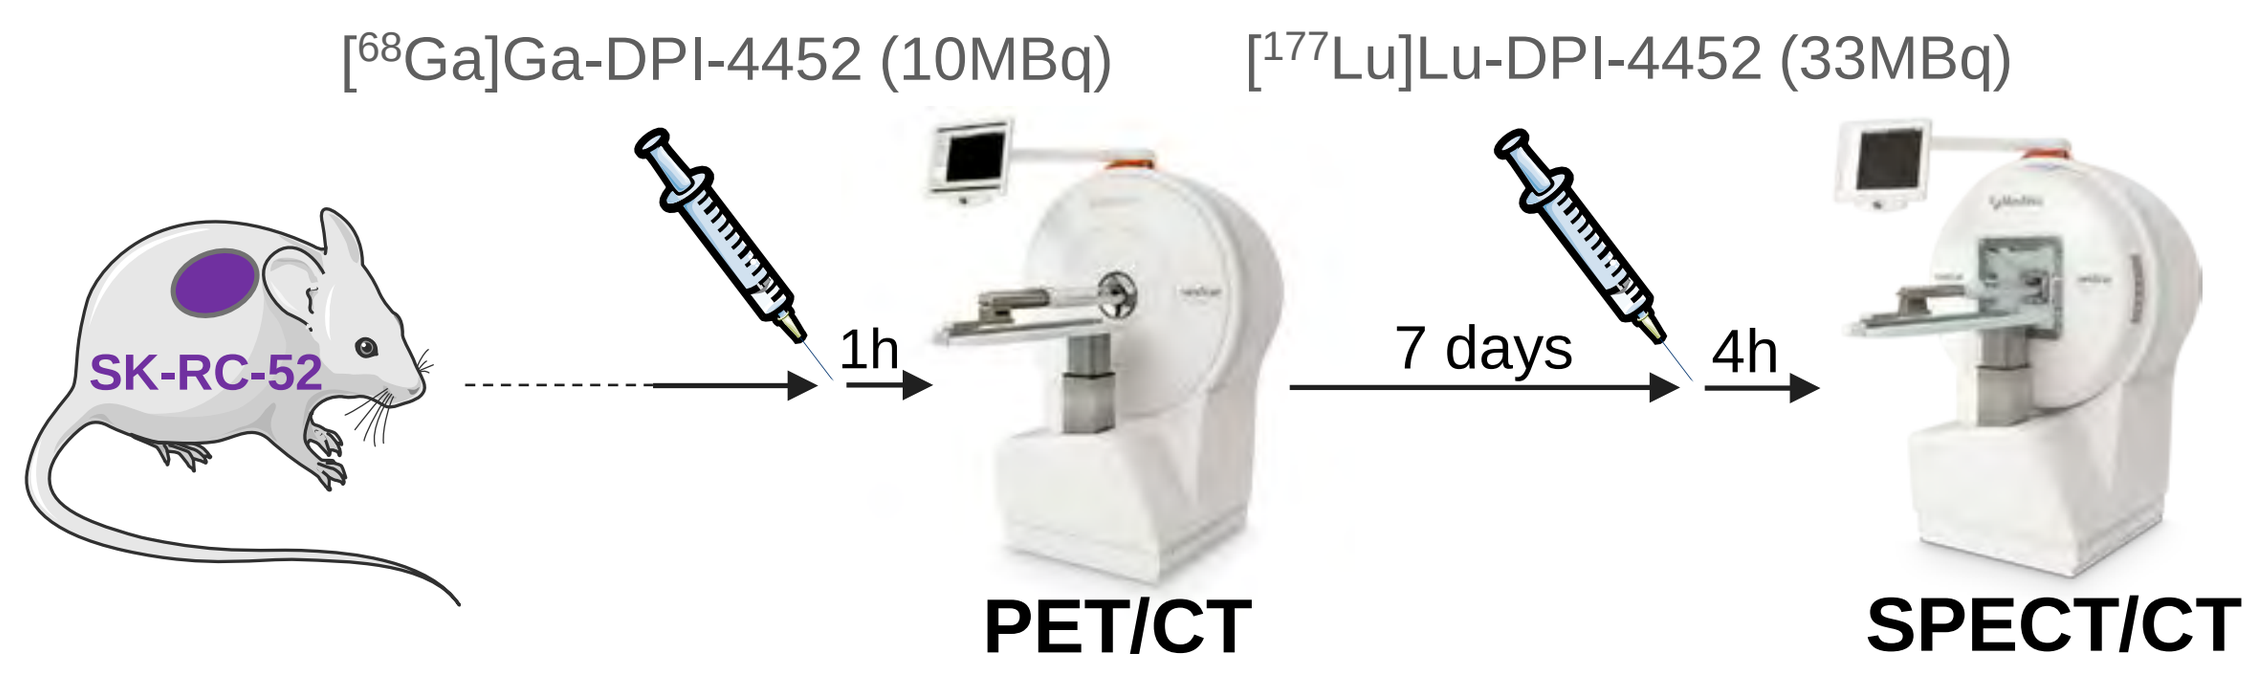
[68Ga]Ga-DPI-4452 (10MBq)
[177Lu]Lu-DPI-4452 (33MBq)
SK-RC-52
1h
7 days
4h
PET/CT
SPECT/CT

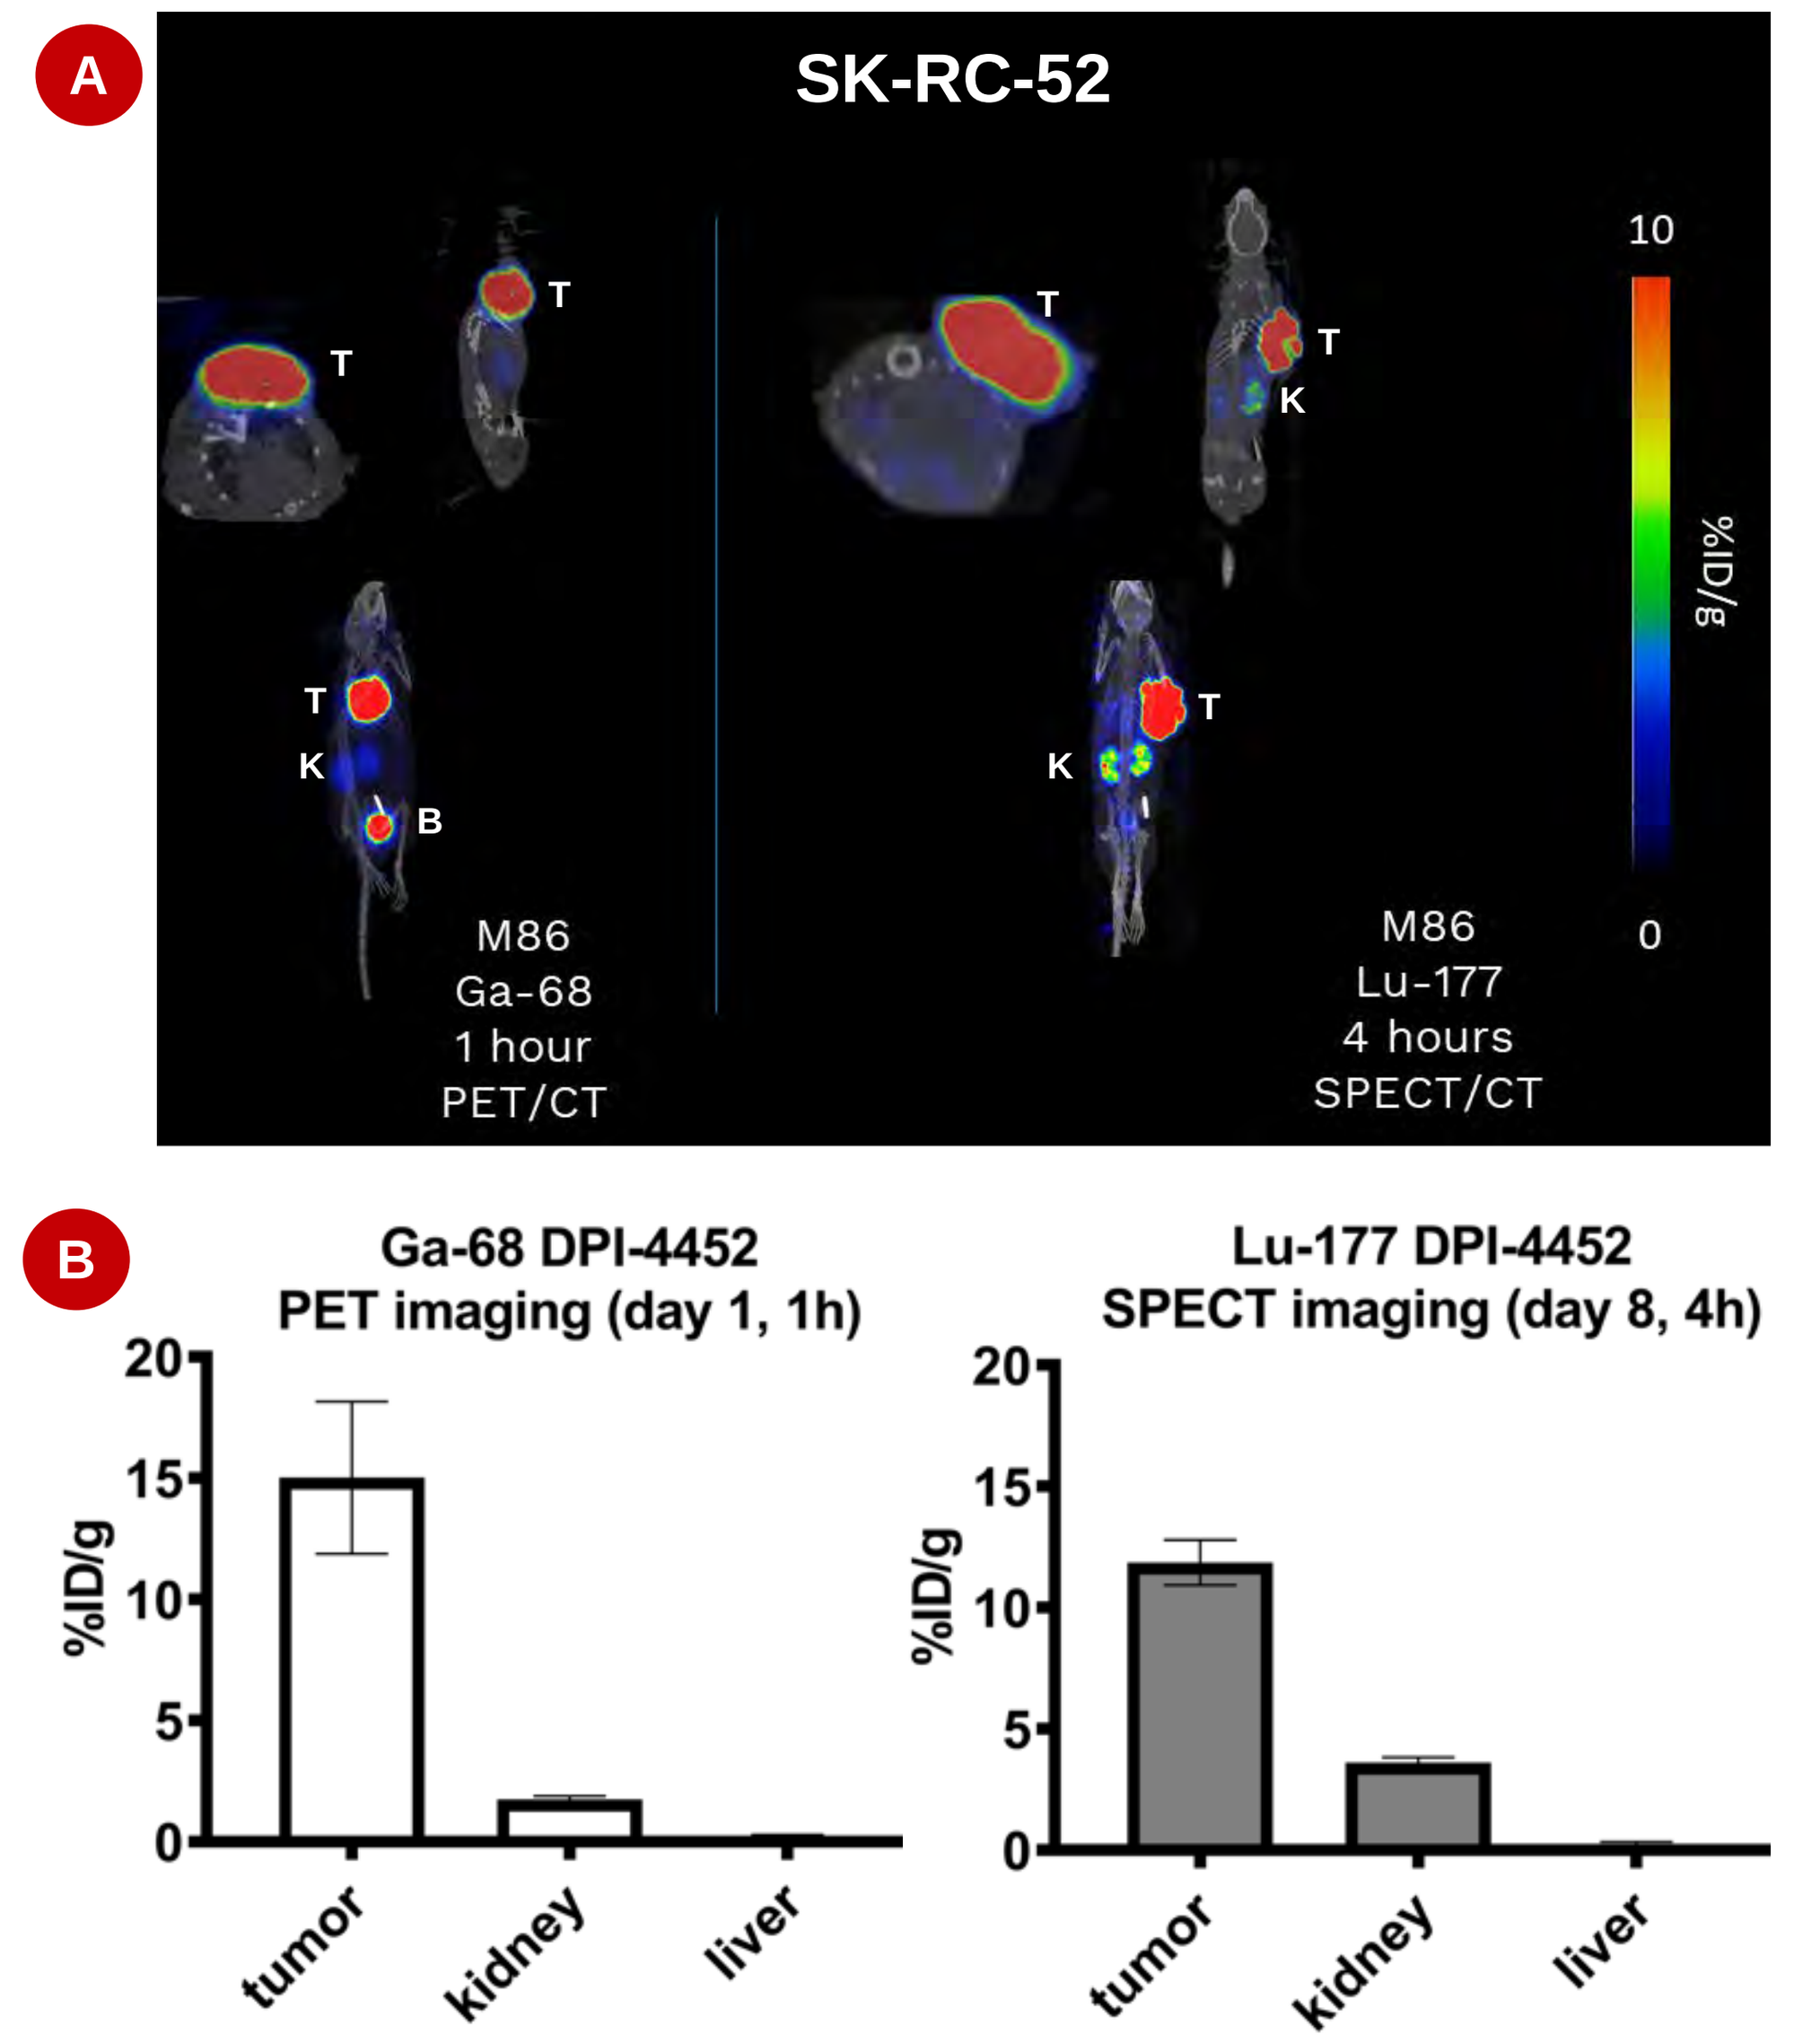
A
SK-RC-52
T
K
B
M86
Ga-68
1 hour
PET/CT
M86
Lu-177
4 hours
SPECT/CT
10
0
%ID/g
B
Ga-68 DPI-4452
PET imaging (day 1, 1h)
Lu-177 DPI-4452
SPECT imaging (day 8, 4h)
%ID/g
tumor
kidney
liver

Figure 3: Comparison of [68Ga]Ga-DPI-4452 and [177Lu]Lu-DPI-4452 uptake in the SK-RC-52 model (comparable results were obtained in the HT-29 model, not shown).

A) Representative PET/CT (day 1, 1h) and SPECT/CT (day 8, 4h) image from the same individual mouse. T: tumor; K: kidney; B: bladder

B) Average uptake (n=6 mice/group).

➢ **Comparable [68Ga]Ga-DPI-4452 and [177Lu]Lu-DPI-4452 uptake in the tumor indicates [68Ga]Ga-DPI-4452 uptake is predictive of [177Lu]Lu-DPI-4452 uptake, thus potentially allowing PET-based patient selection**

In vivo efficacy and tolerability in xenograft models

Strong and dose-dependent tumor growth inhibition (maximal T/C<20%) was observed in the 100MBq and the 3x33MBq treatment groups for the HT-29 model and the 100MBq, 3x33MBq, and the 33MBq treatment groups for the SK-RC-52 model. All [177Lu]Lu-DPI-4452 treatments were well tolerated as assessed by body weights and blood urea and creatinine levels.

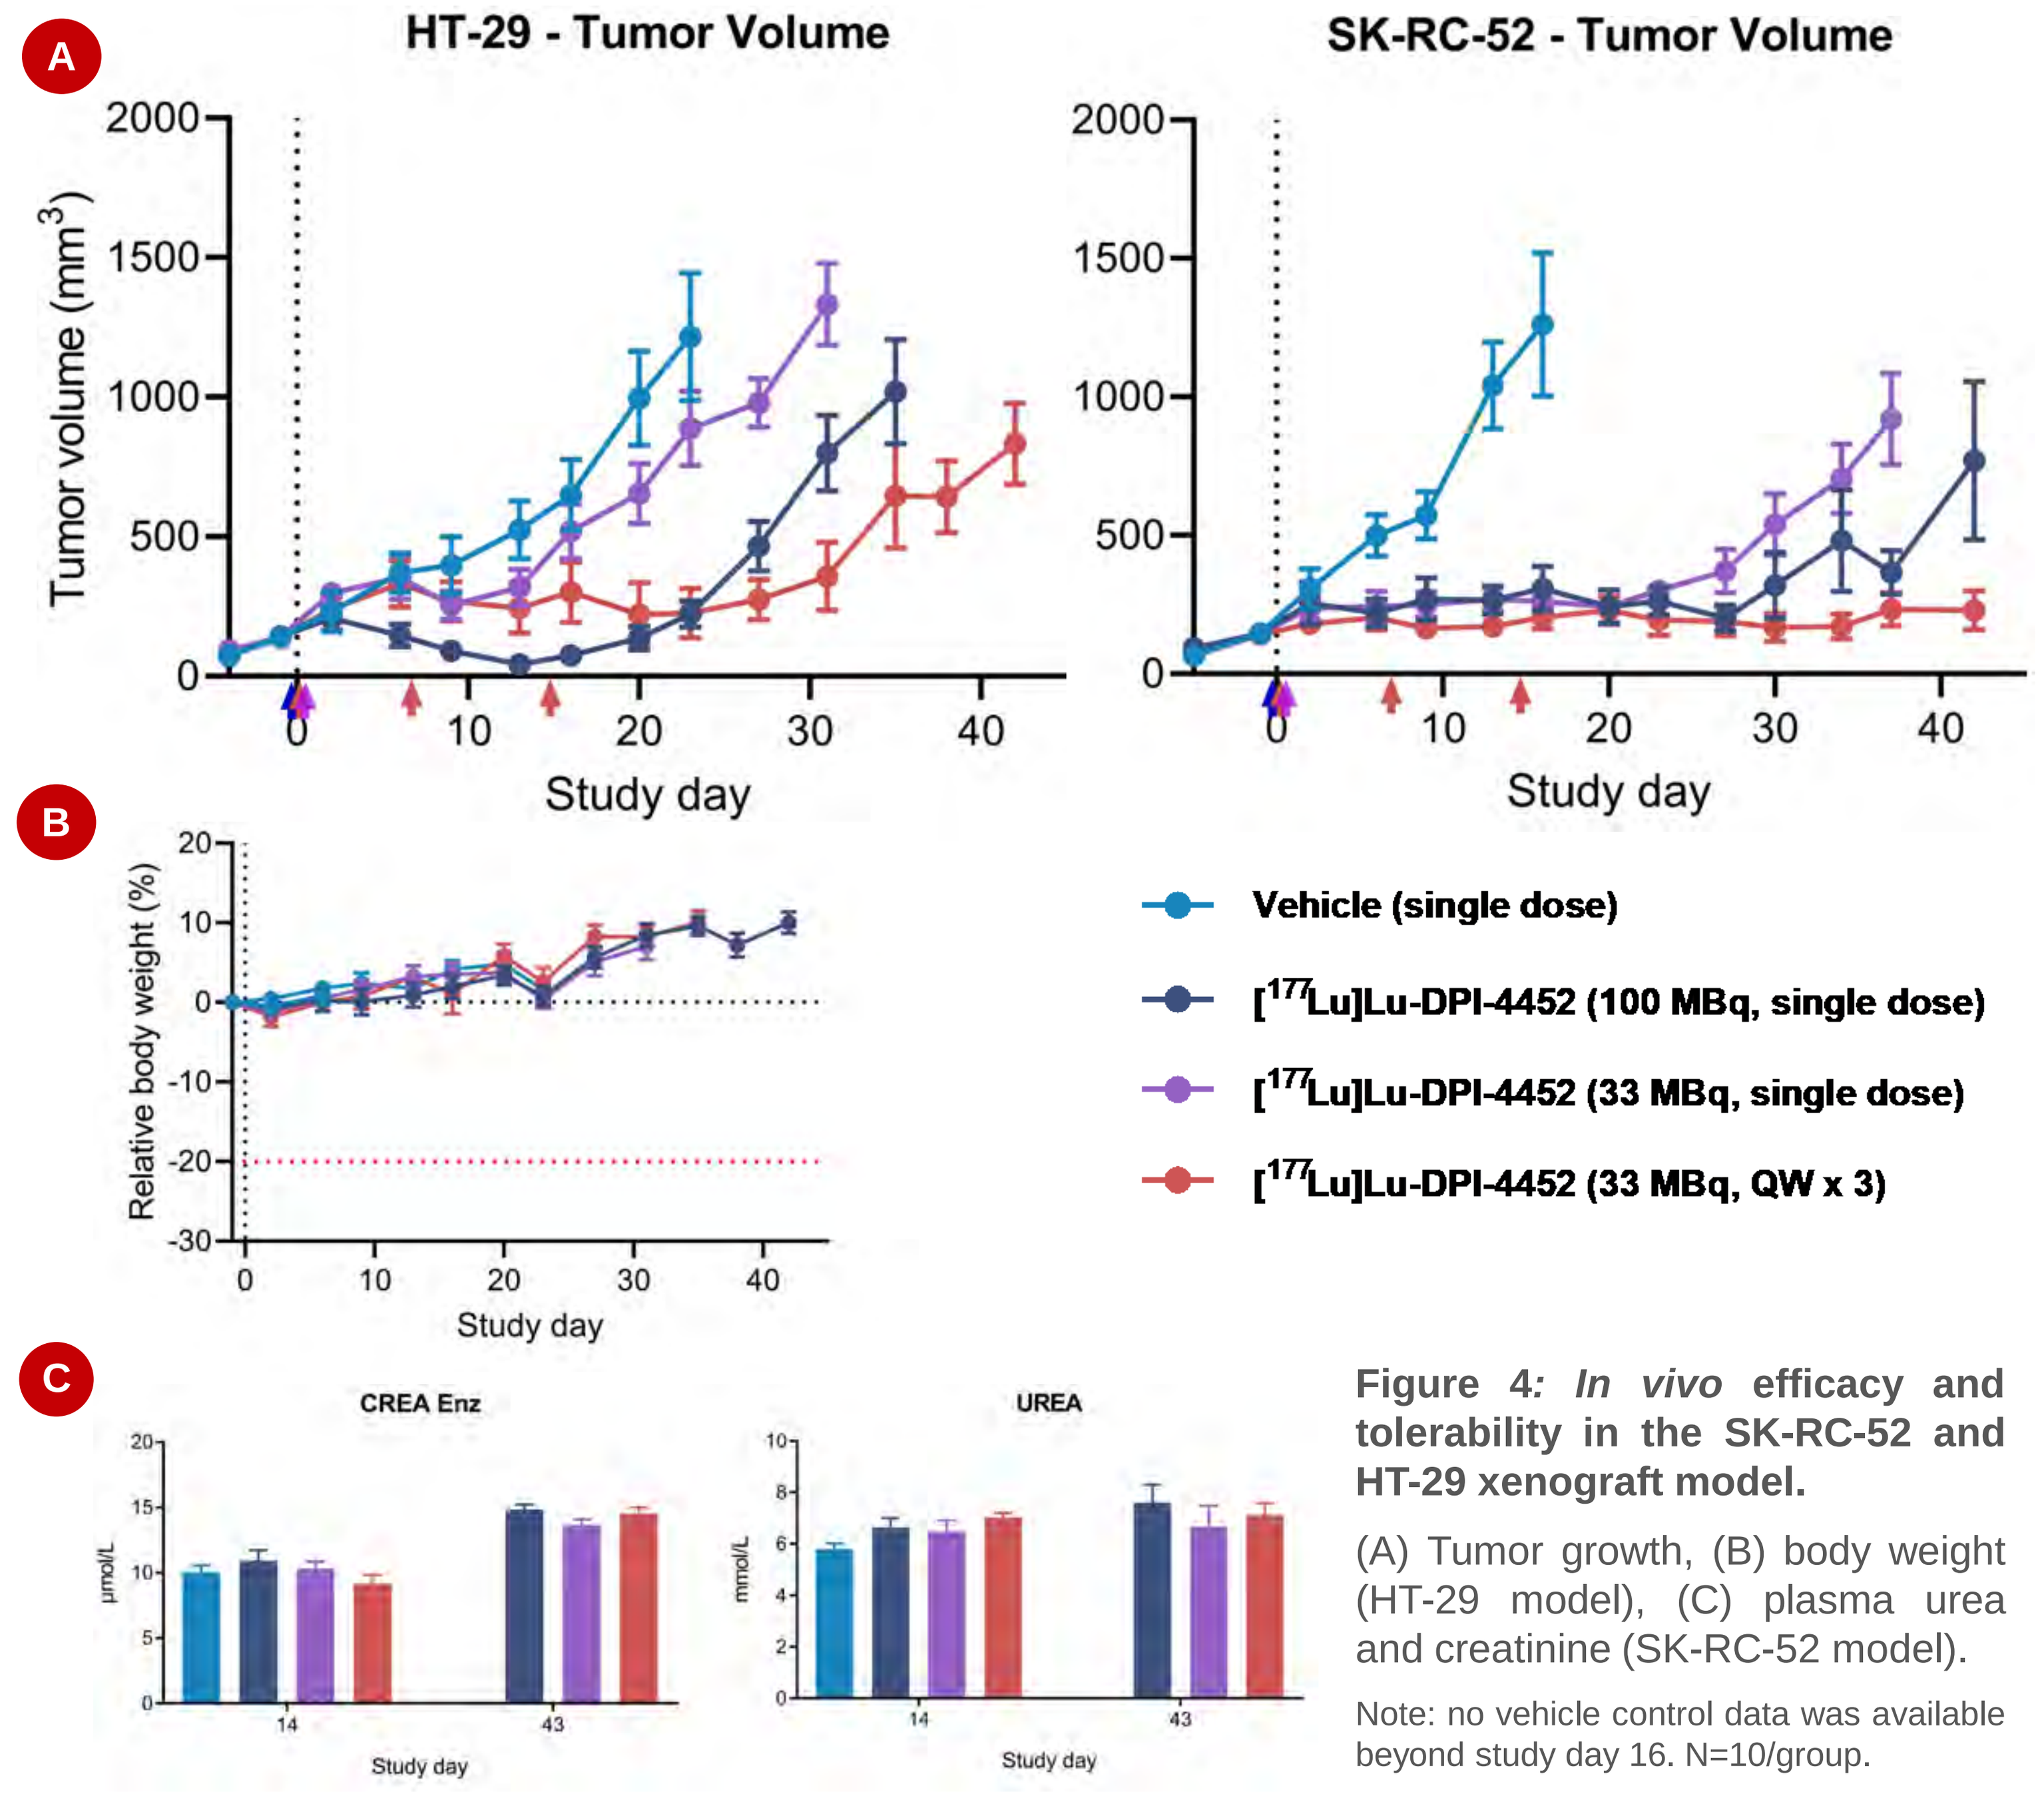
A
HT-29 - Tumor Volume
SK-RC-52 - Tumor Volume
Tumor volume (mm3)
Study day
B
Relative body weight (%)
Study day
Vehicle (single dose)
[177Lu]Lu-DPI-4452 (100 MBq, single dose)
[177Lu]Lu-DPI-4452 (33 MBq, single dose)
[177Lu]Lu-DPI-4452 (33 MBq, QW x 3)
C
CREA Enz
UREA
µmol/L
mmol/L
Study day

Figure 4: *In vivo* efficacy and tolerability in the SK-RC-52 and HT-29 xenograft model.

(A) Tumor growth, (B) body weight (HT-29 model), (C) plasma urea and creatinine (SK-RC-52 model).

Note: no vehicle control data was available beyond study day 16. N=10/group.

➢ **Strong tumor growth inhibition and good tolerability observed in both xenograft models**

CONCLUSIONS

- CAIX shows high and selective expression in ccRCC and CRC human tumor samples
- DPI-4452 binds CAIX with high affinity and a long residence time
- Strong accumulation of [177Lu]Lu-DPI-4452 and [68Ga]Ga-DPI-4452 in SK-RC-52 (ccRCC) and HT-29 (CRC) xenograft tumors, greatly exceeding both kidney and liver exposures
- Comparable [68Ga]Ga-DPI-4452 and [177Lu]Lu-DPI-4452 uptake suggests potential for PET-based patient selection
- [177Lu]Lu-DPI-4452 showed strong tumor growth inhibition and good tolerability in the SK-RC-52 and the HT-29 xenograft models
- DPI-4452 mouse and dog dosimetry results are displayed in the poster #4058

CONTACT

Debiopharm International S.A.,
Lausanne, Switzerland.
www.debiopharm.com
antoine.attinger@debiopharm.com

DOWNLOAD

This poster is available via:
www.debiopharm.com/medias/publications