Safety assessment of DPI-4452, a cyclic peptide targeting Carbonic Anhydrase IX

Inês Borrego[1], Frédéric Massière[1], Antoine Attinger[1], Anne Schumann [2], Aileen Hoehne [2], Frank Osterkamp [2], Ulf Andersson[1]

[1]Debiopharm International SA, Chemin Messidor 5-7, CH-1002 Lausanne, Switzerland. [2] 3B Pharmaceuticals GmbH, Magnusstrasse 11, D-12489 Berlin, Germany

Debiopharm
WE DEVELOP FOR PATIENTS

POSTER #P525

INTRODUCTION

- **The transmembrane metalloprotease Carbonic Anhydrase IX (CAIX) represents an attractive diagnostic and therapeutic target in hypoxic solid tumors such as Colorectal Cancer (CRC) and clear cell Renal Cell Carcinoma (ccRCC), with limited expression in healthy organs.**
- **DPI-4452 is a CAIX-targeting cyclic peptide with a DOTA cage that can be labelled with different radionuclides for theranostic purposes.**
- **Here, we report a brief overview of the preclinical data and the preclinical toxicology package.**

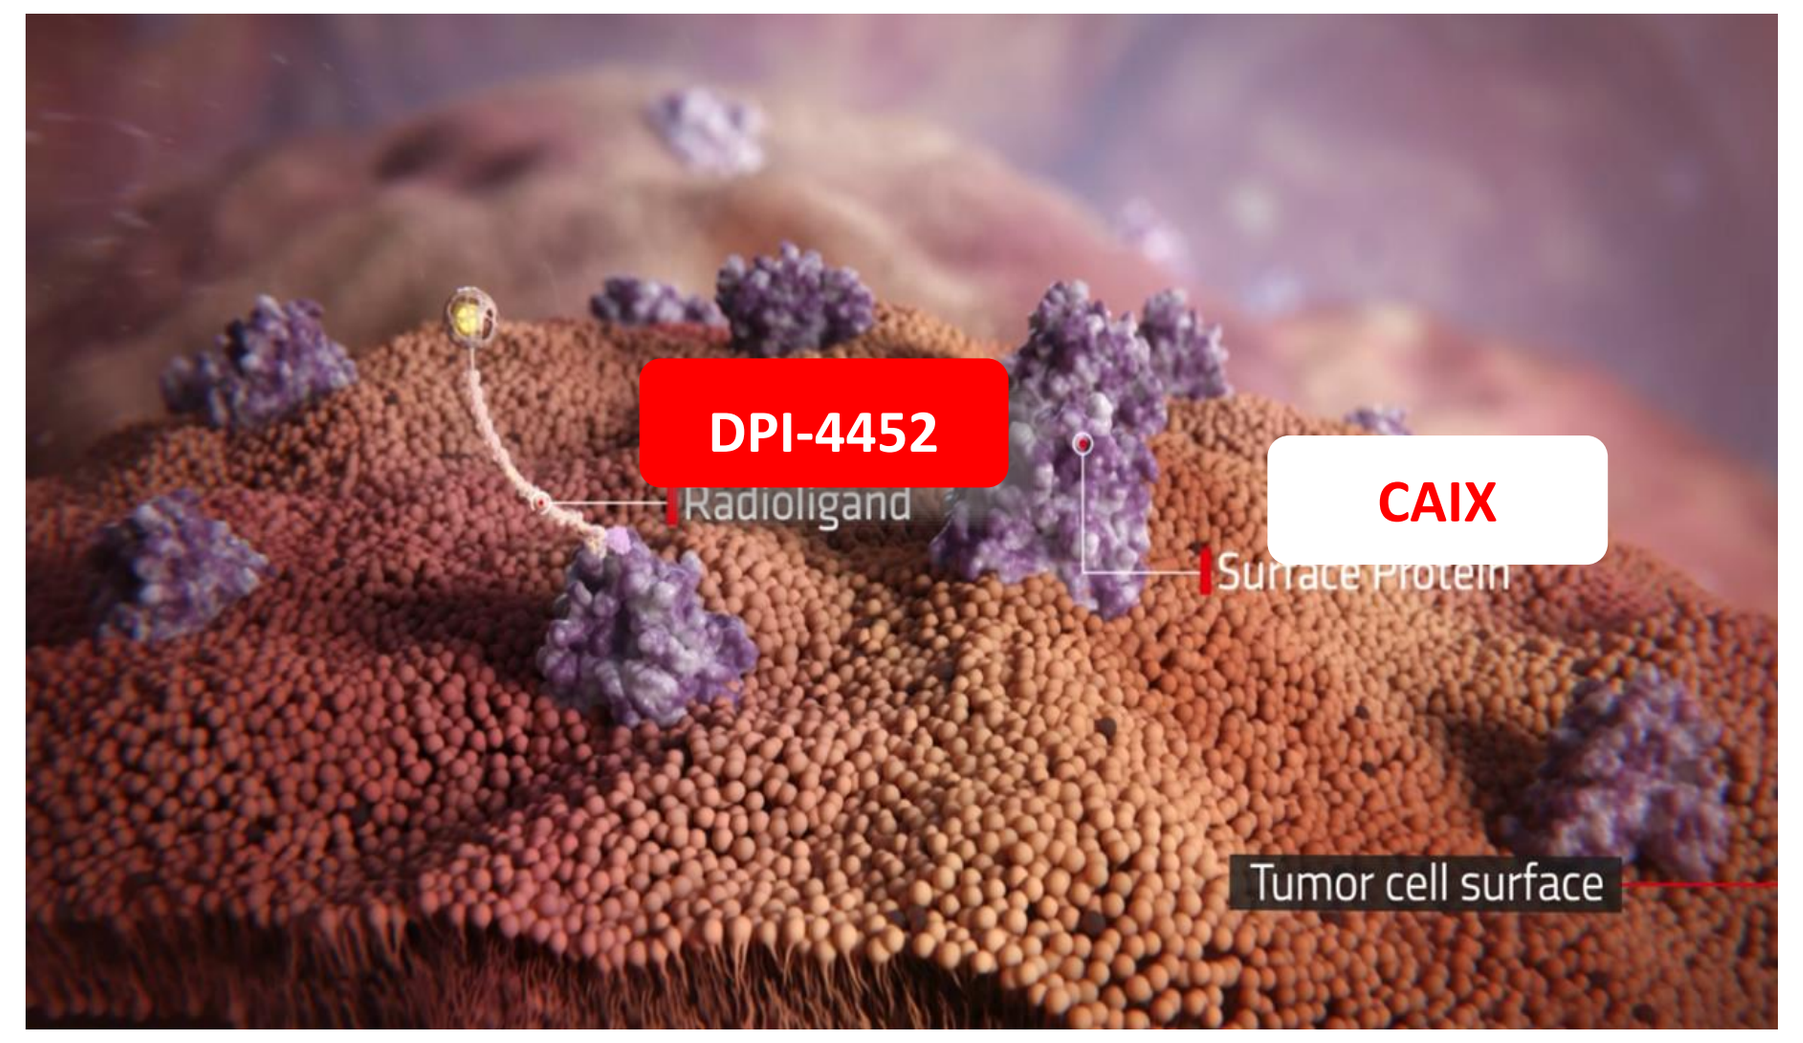

DPI-4452
Radioligand
CAIX
Surface Protein
Tumor cell surface

MATERIALS AND METHODS

***In-vitro* binding to CAIX and species cross-reactivity.** Different concentrations of [^{111}In]In-DPI-4452 were incubated with CHO cells transfected with human, dog or mouse CAIX in a radioligand binding assay.

SPECT/CT-based *in vivo* biodistribution in healthy dogs and dosimetry. Beagle dogs (2 males and 2 females) were injected intravenously with ^{111}In-labeled compound (single bolus, 250 MBq, ~40 nmol ligand). Biodistribution was evaluated using whole-body SPECT/CT scanning under anesthesia (3 mg/kg IV propofol). Radioactivity uptake was quantified in regions of interest drawn over 8 organs: kidney, liver including gallbladder, gonads, bone marrow, lung with pleura, stomach, small intestine, and colon. Uptake was expressed as % injected dose per gram tissue (%ID/g) and as SUV (standardized uptake value: ratio of tissue radioactivity concentration, e.g., in kBq/mL, to administered dose per body weight, e.g., in MBq/kg). Dosimetry was conducted as follows: from the time-activity data, the residence time of radiation in each organ of interest was calculated; those values were input in OLINDA/EXM 2.0 to calculate the radiation dose absorbed in each organ and in the whole-body. Estimates of the radiation dose absorbed in human organs after injection of [^{177}Lu]Lu-DPI-4452 were obtained with dosimetry adaptation to the radionuclide characteristics of ^{177}Lu and extrapolation to ICRP89 male and female human phantoms using the %kg/g method (Kirschner A.S. *et al.*, 1975).

In vivo efficacy study. CAIX-positive human cancer cell line HT-29 (CRC) or SK-RC-52 (ccRCC) were subcutaneously implanted with matrigel (2x10^6 cells for each model) into NMRI nude mice. Animals for each tumor type were divided in 4 treatment groups of 10 mice each: a) Single administration of vehicle b) Single administration of 100 MBq of [^{177}Lu]Lu-DPI-4452 c) Single administration of 33 MBq of [^{177}Lu]Lu-DPI-4452 d) Three administrations (QW) of 33 MBq of [^{177}Lu]Lu-DPI-4452.

Dose-range finding study incl TK. DPI-4452 was administered by IV bolus injection at ascending dose levels up to 800 µg/kg/day to one group of two Beagle dogs as one single-dose followed by 3 days of wash-out period. The following parameters and endpoints were evaluated: mortality, clinical observations, body weights, food consumption, local reactions, clinical pathology parameters (hematology, coagulation, and clinical chemistry), organ weights, and macroscopic examinations.

GLP Extended Single-Dose toxicity study with Safety Pharmacology assessment. DPI-4452 was administered in an extended two subsets single IV dose in Beagle dogs, including safety pharmacology endpoints at 16, 80, or 400 µg/kg. Subset A of 3 males and 3 females per group was dedicated to the Functional Observational Battery (FOB) assay and toxicokinetic investigations and terminated after 24h. Subset B of 2 males and 2 females per group was dedicated to the telemetry recording on Days 1 and 14. The following parameters and endpoints were evaluated: mortality, clinical observations, body weights, food consumption, body temperature, local reactions, ophthalmology, clinical pathology parameters (hematology, coagulation, clinical chemistry, and urinalysis), cardiovascular and respiratory safety pharmacology endpoints along with FOB evaluation, organ weights, macroscopic and microscopic examinations, and TK parameters.

RESULTS

Binding characteristics to mouse, dog and human CAIX

Table 1: *In vitro* binding of [^{111}In]In-DPI-4452 to mouse, dog and human CAIX expressed in CHO cells

Compound	Human CAIX		Dog CAIX		Mouse CAIX	
	pK_D	B_{max} (fmol/µg prot)	pK_D	B_{max} (fmol/µg prot)	pK_D	B_{max} (fmol/µg prot)
[^{111}In]In-DPI-4452 (n=2)	**9.5**	12.8	**9.6**	9.3	**7.2**	5.6

pK_D: -log of dissociation constant. B_{max}: concentration of specific binding sites

- **[^{111}In]In-DPI-4452 low affinity for rodent CAIX, however, the binding affinity to canine CAIX was similar to humans**
- **Mouse biodistribution does not enable to investigate the compound accumulation in organs naturally expressing CAIX**
- **Dog is a relevant species for safety assessments**

In vivo efficacy and tolerability in xenograft models

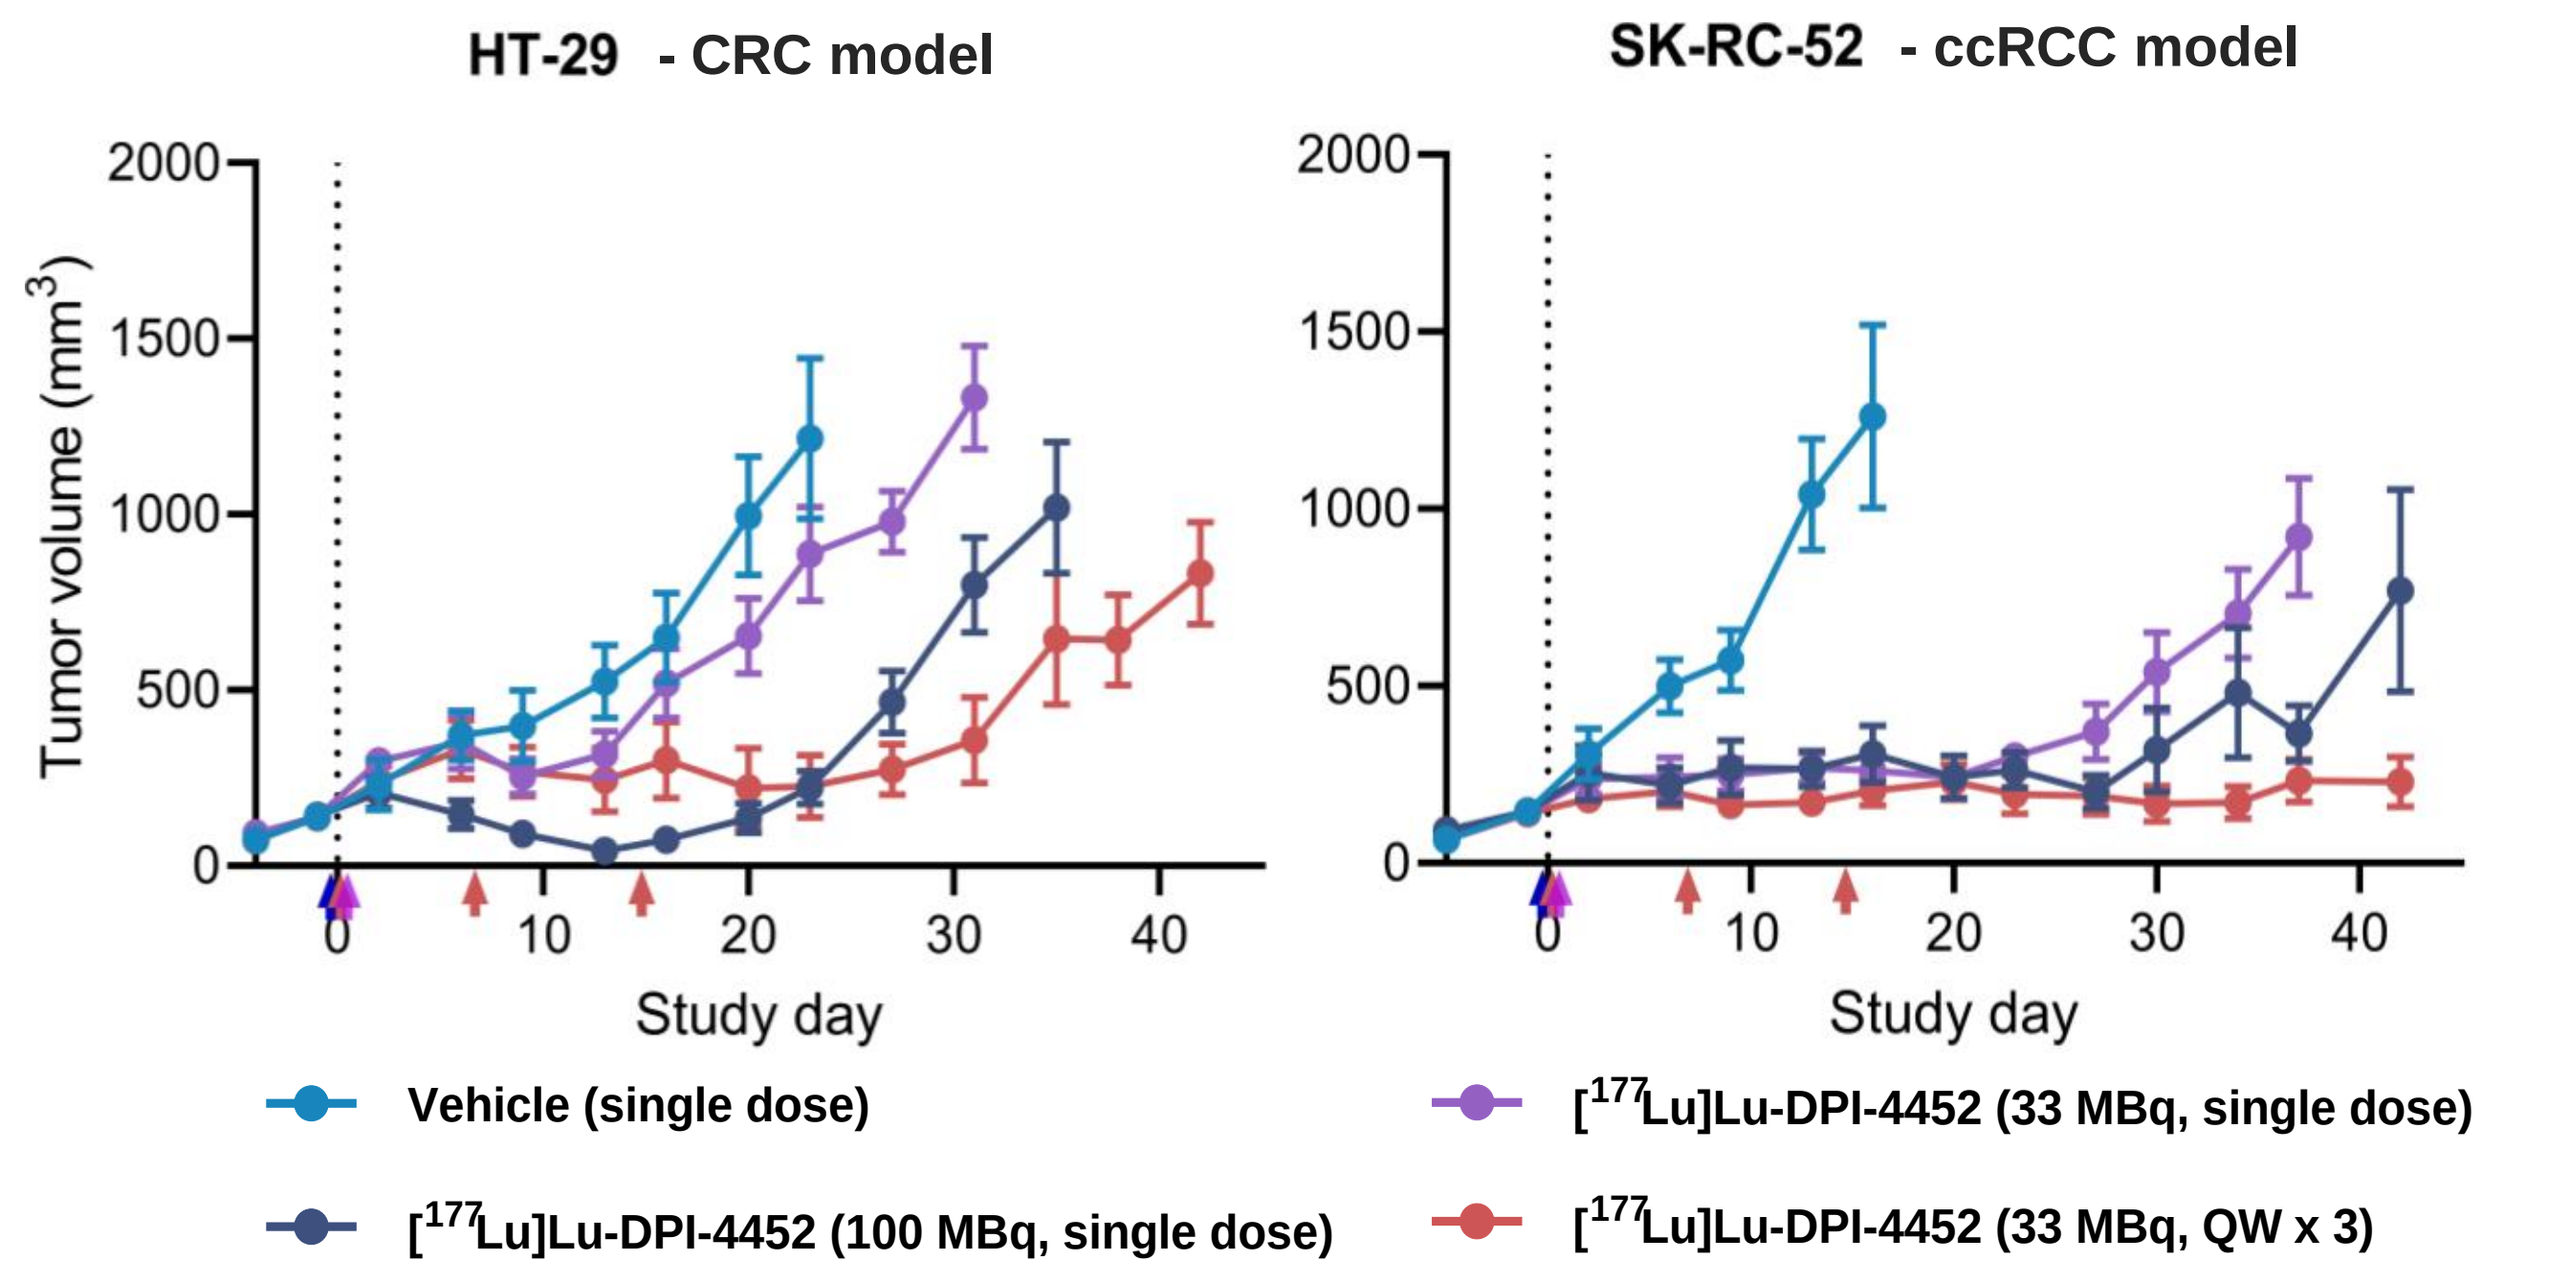

HT-29 - CRC model
SK-RC-52 - ccRCC model
Tumor volume (mm³)
Study day
Vehicle (single dose)
[177Lu]Lu-DPI-4452 (100 MBq, single dose)
[177Lu]Lu-DPI-4452 (33 MBq, single dose)
[177Lu]Lu-DPI-4452 (33 MBq, QW x 3)

Figure 1: *In vivo* efficacy in the SK-RC-52 and HT-29 xenograft model (good tolerability and no BW alterations were observed)

- **Strong tumor growth inhibition and good tolerability observed in both xenograft models**

Biodistribution in healthy dog

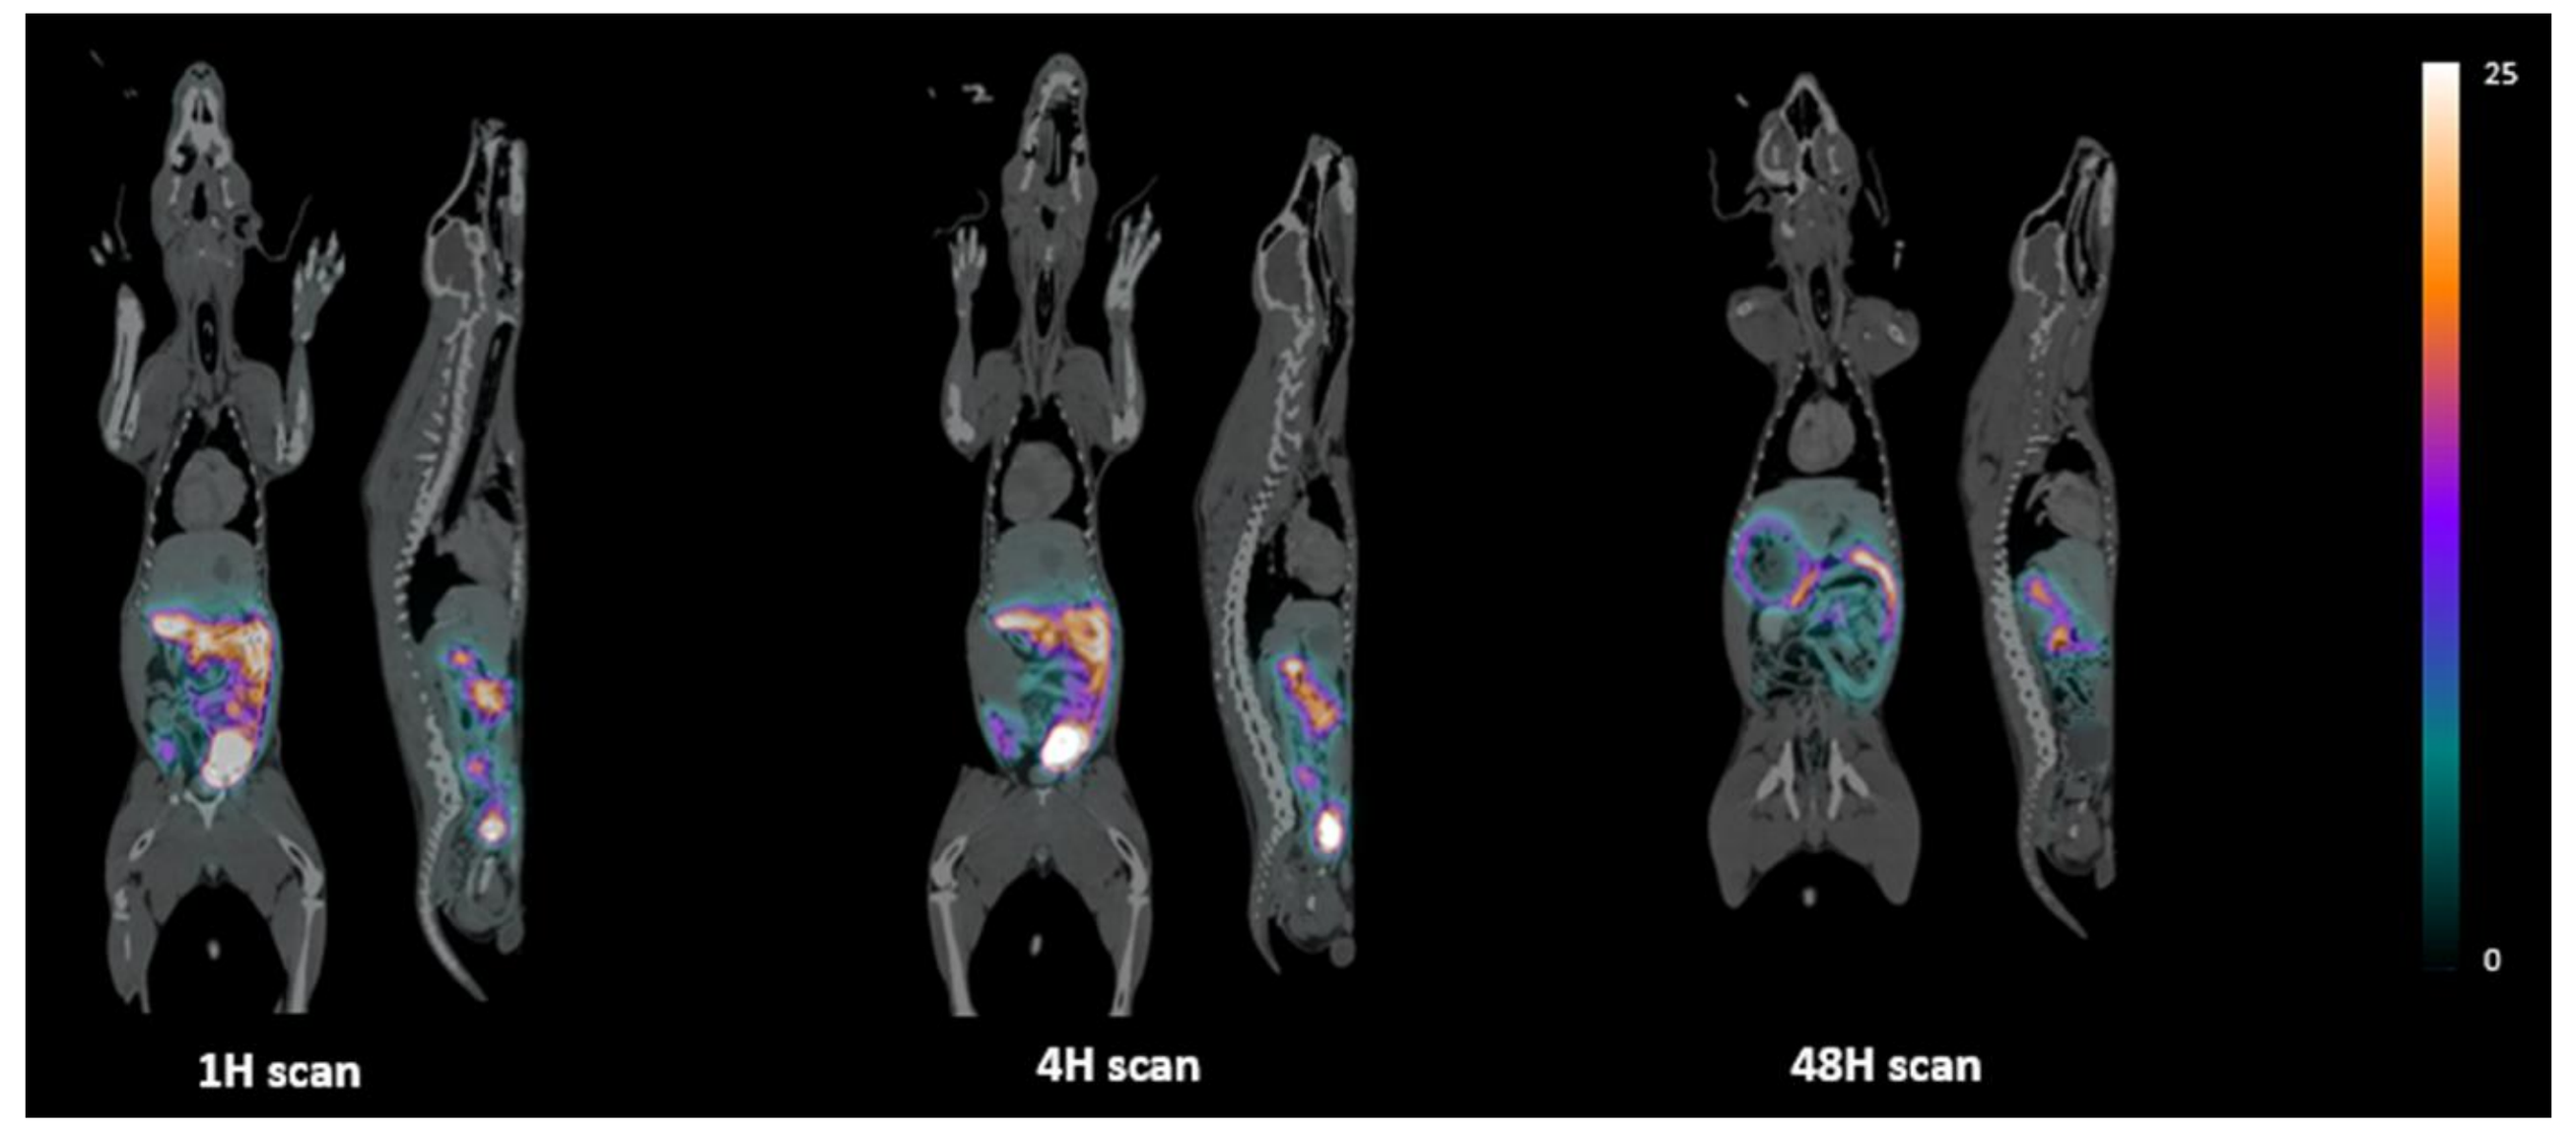

1H scan
4H scan
48H scan
25
0

Figure 2: SPECT/CT representative images of biodistribution in dogs: one male Beagle dog after injection of [^{111}In]In-DPI-4452. Scalebar represents SUV values.

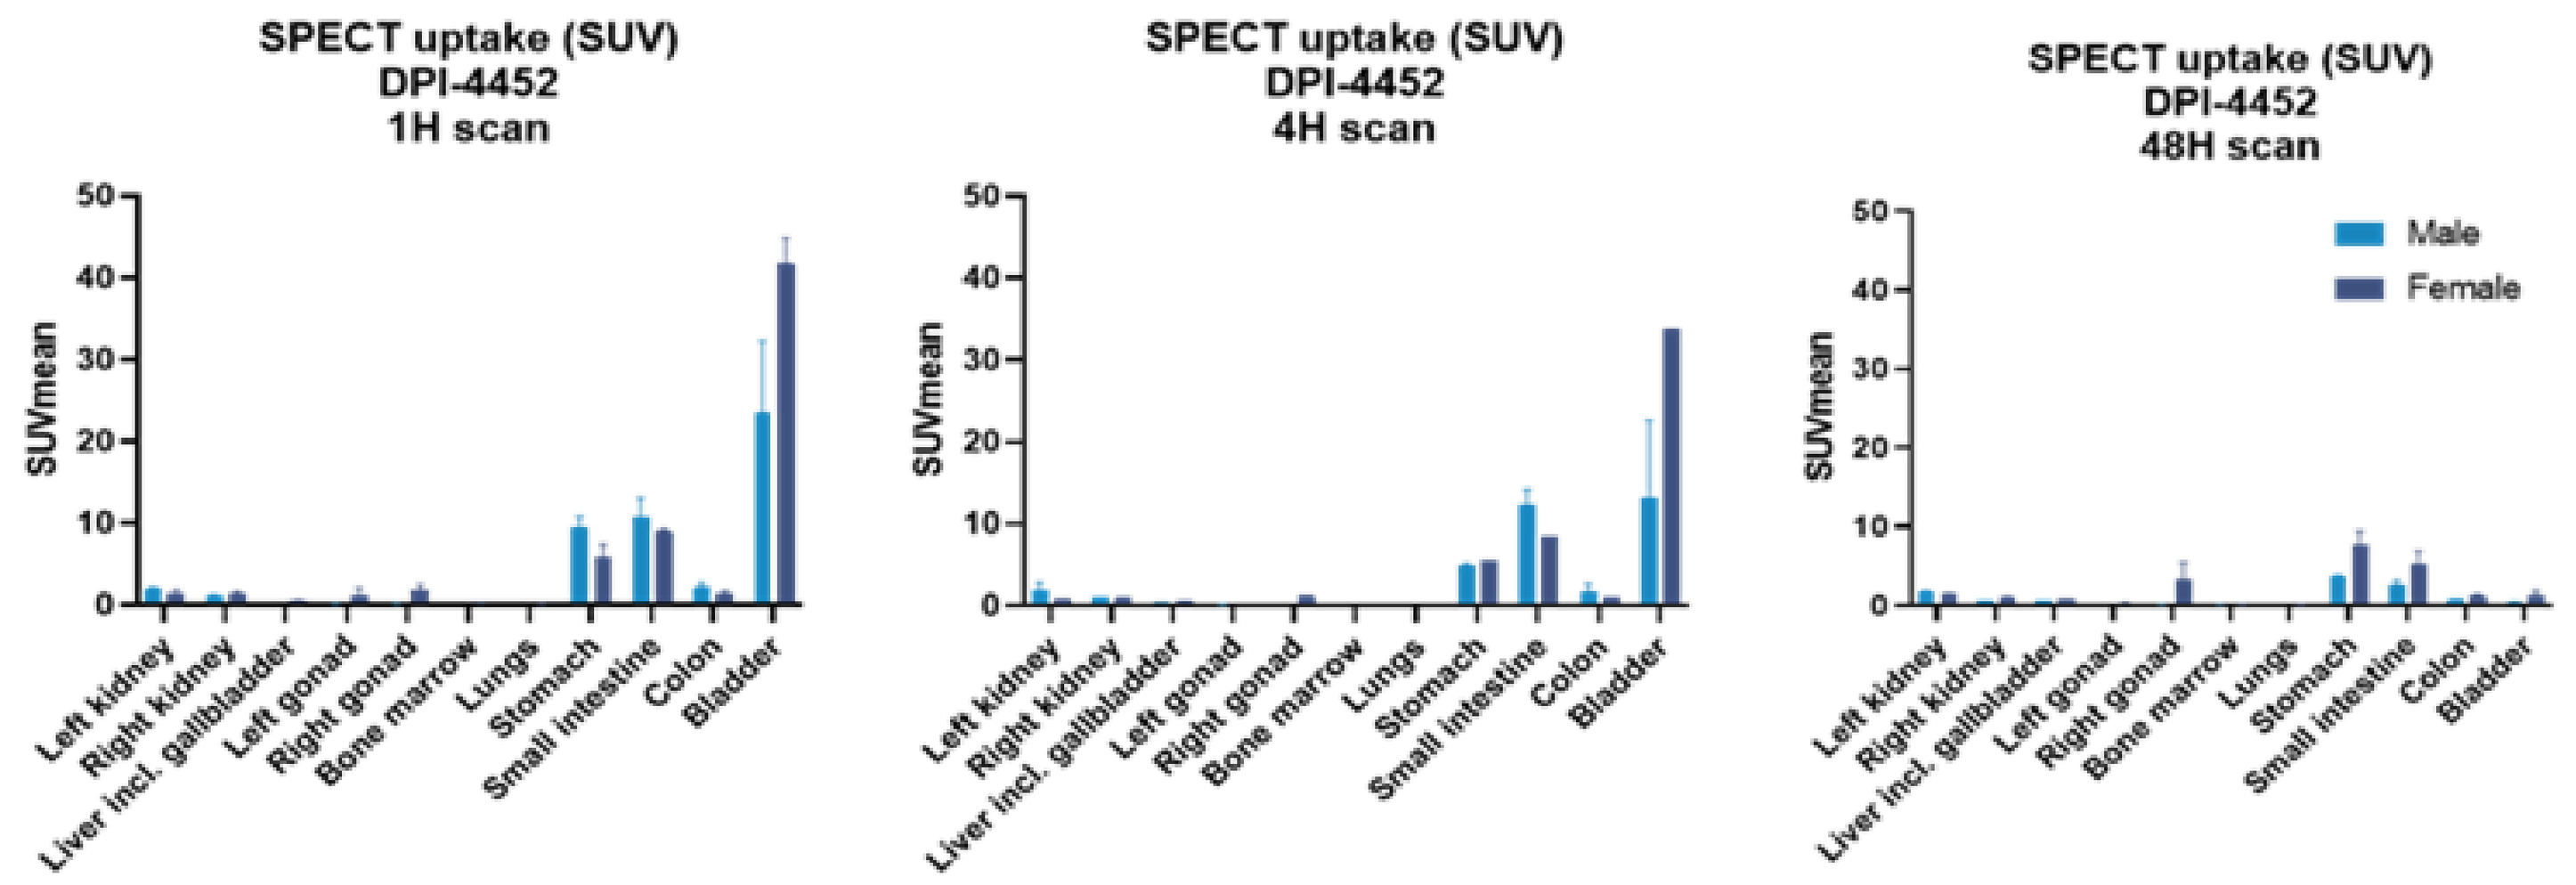

SPECT uptake (SUV) DPI-4452 1H scan
SPECT uptake (SUV) DPI-4452 4H scan
SPECT uptake (SUV) DPI-4452 48H scan
SUVmean
Male
Female
Left kidney
Right kidney
Liver incl. gallbladder
Left gonad
Right gonad
Bone marrow
Lungs
Stomach
Small intestine
Colon
Bladder

Figure 3: SPECT/CT-derived dog biodistribution data (as SUV) of [^{111}In]In-DPI-4452 N=2/group. Plots represent mean ± SEM.

- **Highest uptake is in stomach and small intestine, consistent with reported expression of CAIX in these organs**
- **Significant signal in the bladder, primarily due to rapid renal elimination**
- **Low uptake in the kidney, bone marrow, blood, heart wall and skin**

Dog dosimetry supports intended human starting dose

Table 2: Dosimetry based on dog biodistribution data after IV injection of [^{111}In]In-DPI-4452, adaptation to ^{177}Lu radionuclide and extrapolation to human

Organ	Absorbed radiation dose (mGy/MBq) (dosimetry output)	Dose limit (Gy) *	Allowed dose (GBq)
Small intestine	0.609	18	29.6
Stomach wall	0.654	28	42.8
Kidneys	0.333	23	69.1
Colon	0.234	25	106
Rectum	0.233	25	106
Urinary bladder wall	0.353	18.3	51.8

* Emami, B. (2013). Tolerance of normal tissue to therapeutic radiation. Reports Radiother Oncol. 1. 35-48.

- **Similar expression of CAIX in dogs and humans is assumed**
- **Human estimated effective dose (average of male and female): 0.234 mSv/MBq (i.e., 866 mSv for a 3.7 GBq dose)**
- **Small intestine is the potential dose-limiting organ. Starting dose for humans is covered by conservative estimate of allowed dose (29.6 GBq) based on organ radiation dose limits set for EBRT**

Dose-Range Finding study

Figure 4: Mean total plasma concentration of DPI-4452 versus time profiles following a single IV bolus injection of DPI-4452 in male Beagle Dogs. N=6/group, Mean ± SD

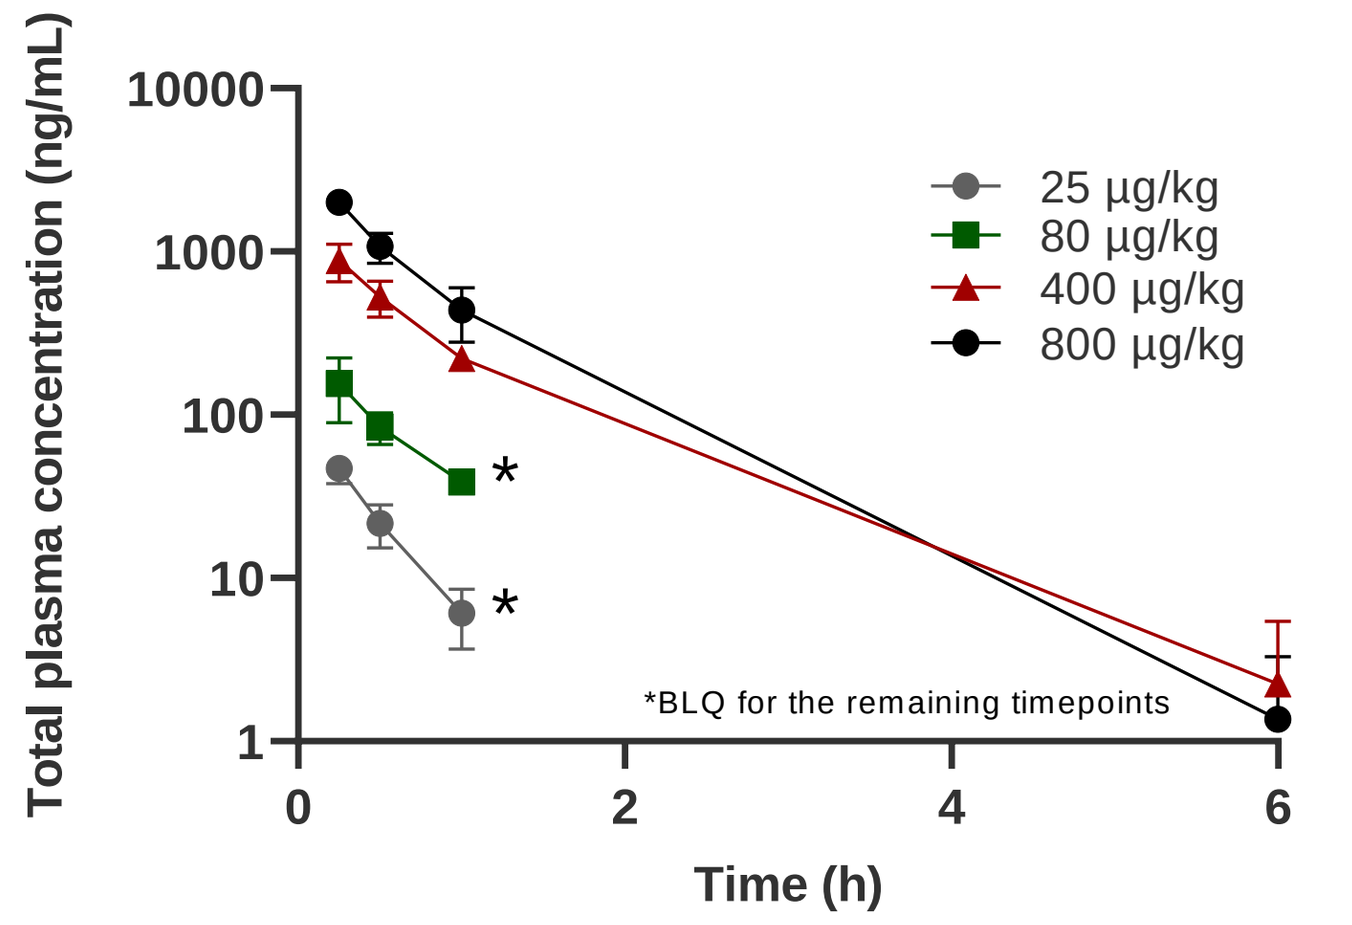

Total plasma concentration (ng/mL)
Time (h)
25 µg/kg
80 µg/kg
400 µg/kg
800 µg/kg
*BLQ for the remaining timepoints

- **Administrations well tolerated up to 800 µg/kg with no treatment-related findings (in-life parameters, clinical pathology and macroscopic observations)**
- **TK data suggested that exposure increased dose-proportionally**

GLP Extended Single-Dose toxicity study with Safety Pharmacology assessment (CV, Resp, CNS)

Figure 5: Mean total plasma concentration of DPI-4452 versus time profiles following a single IV bolus injection of DPI-4452 in Beagle dogs. No difference in exposure between male and female. N=2, Mean ± SD

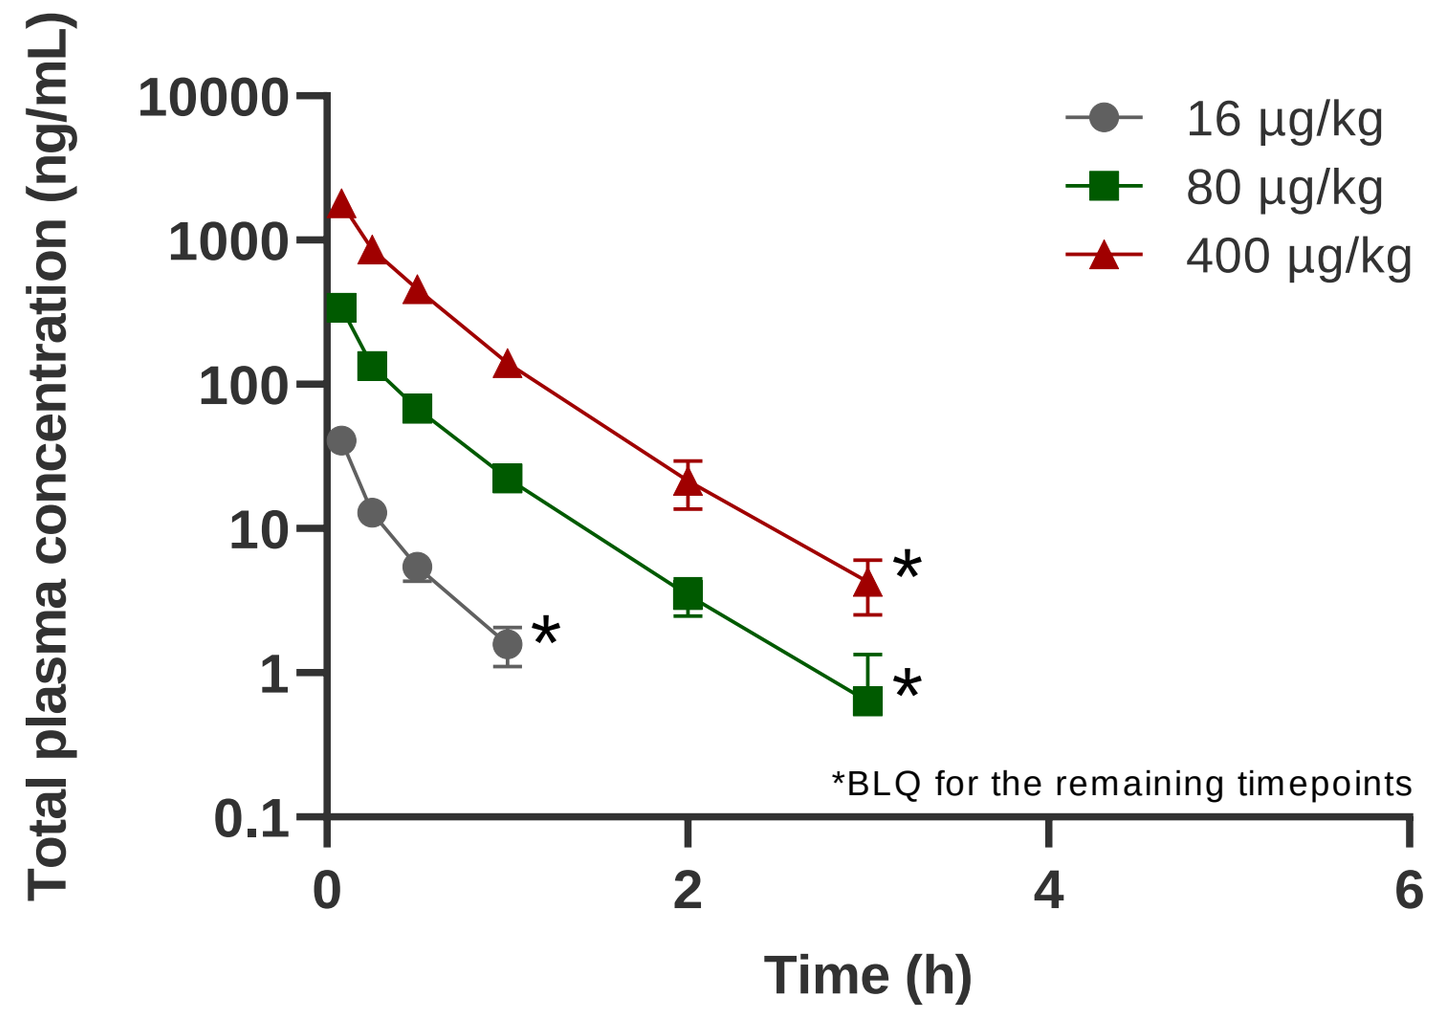

Total plasma concentration (ng/mL)
Time (h)
16 µg/kg
80 µg/kg
400 µg/kg
*BLQ for the remaining timepoints

- **Administrations well tolerated up to 400 µg/kg with no treatment-related findings at early and late necropsies (in-life parameters, clinical pathology, histopathology and safety pharmacology assessment)**
- **TK data suggested that exposure increased dose-proportionally**
- **NOAEL/NOEL: 400 µg/kg**

Human PK Prediction

Table 3: Estimated human PK parameters of DPI-4452, based on allometric extrapolation from Mouse and Dog PK data

PK parameters	Mouse (0.7 mg/kg)	Dog (0.1 mg/kg)	Human (prediction)
$T_{1/2}$ (h)	0.28	0.38	0.17-1.47
V_d (L/kg)	0.41	0.26	0.19-0.46
CL (mL/min/kg)	27.5	12.7	3.57-13.0

- **Predicted human C_{5min} = 15.8-38.0 ng/mL at the dose currently planned in clinic**
- **Highest observed concentration in the dog at the NOAEL (400 µg/kg): C_{5min} = 1840 ng/mL = 50-fold higher than expected concentration in humans**

CONCLUSIONS

- **[^{111}In]In-DPI-4452 is a potent binder of dog and human CAIX, but does not cross-react with mouse CAIX to similar extent. Dog is a relevant species for safety assessments.**
- **[^{177}Lu]Lu-DPI-4452 showed strong tumor growth inhibition and good tolerability in the SK-RC-52 and the HT-29 xenograft models**
- **Biodistribution in healthy dogs showed highest uptake in stomach and small intestine, and low uptake in kidney, consistent with expression of CAIX in these organs**
- **Small intestine is the potential dose-limiting organ.**
- **Administrations well tolerated up to 400 µg/kg with no treatment-related findings, at exposures 50-fold higher than predicted human exposure in the planned clinical trial.**

CONTACT

Debiopharm International S.A.,
Lausanne, Switzerland.
www.debiopharm.com
ines.borrego@debiopharm.com

DOWNLOAD

This poster is available via:
www.debiopharm.com/medias/publications