A phase 1 study of the IAP inhibitor xevinapant (Debio 1143) to evaluate food effect and drug-drug interactions with a proton pump inhibitor in healthy volunteers

V. Nicolas-Métral,[1] E. Rouits,[1*] C. Riff,[1*] D. Spaggiari,[1] N. Wiedemann,[1] P. Colin,[1*] E. Harrison-Moench,[2] R. Crabbé,[1] H. Nauwelaerts,[1] A. Schroeder,[2] C. Haefliger,[1] Y. Vugmeyster[3]

[1]Debiopharm International SA, Lausanne, Switzerland; [2]the healthcare business of Merck KGaA, Darmstadt, Germany; [3]EMD Serono, Billerica, MA, USA

*Affiliation at the time the study was conducted.

SCOPE

- We report results of this randomized, 2-stage, phase 1 study evaluating food effect, drug-drug interactions with a proton pump inhibitor (pantoprazole) and the relative bioavailability of 2 formulations of xevinapant

CONCLUSIONS

- Food and pantoprazole did not meaningfully alter the extent of absorption of xevinapant, as evidenced by no change in the AUC
 - Given xevinapant's mechanism of action, AUC is the most relevant exposure metric for efficacy
- Target engagement was not affected by food at steady state based on pharmacokinetic modeling
- In future studies xevinapant can be administered without regard to food or gastric acid–reducing agents, which will be advantageous for patient convenience and adherence
- These results have already informed dosing in the ongoing HyperlynX and XRay Vision studies that are evaluating xevinapant in combination with chemoradiotherapy and radiotherapy, respectively[1,2]; in these studies, xevinapant is being administered without regard to food

GET POSTER PDF

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from AACR and the authors of this poster

Correspondence: **Yulia Vugmeyster, yulia.vugmeyster@emdserono.com**

SUPPLEMENTARY DATA

Please scan this QR code to view supplementary data associated with this poster

BACKGROUND

- Xevinapant is a potentially first-in-class, potent, oral, small-molecule IAP (inhibitor of apoptosis protein) inhibitor that is thought to restore cancer cell sensitivity to apoptosis and increase anti-tumor immunity, thereby enhancing the effects of chemotherapy and radiotherapy[3–5] via:
 - Inhibiting X-linked IAP, which releases the blockade on downstream caspase activity in the intrinsic apoptotic pathway[3]
 - Inhibiting cellular IAP1/2 (cIAP1/2), which promotes proapoptotic TNF receptor signaling via the extrinsic pathway and induces TNF–α expression via the noncanonical NF–κB pathway[3,6–8]
- In a randomized phase 2 study of patients with unresected locally advanced squamous cell carcinoma of the head and neck, xevinapant + chemoradiotherapy (CRT) significantly improved locoregional control at 18 months after the end of CRT and halved the risk of death after 5 years of follow-up vs placebo + CRT[9,10]

METHODS

- This randomized, crossover, open-label, phase 1 study (**Figure 1**) included healthy volunteers aged 18–60 years who smoked ≤5 cigarettes per day, had a body mass index between 18 and 30 kg/m^2, and had no regular intake of any medication or vitamin, mineral, or dietary supplement within 2 weeks prior to administration of study treatment and during the study period
- This study was composed of 2 stages:
 - Stage 1: a single dose of oral xevinapant 200 mg formulated as Solution A or Solution B (with permeability regulator) in fasted and fed conditions
 - Stage 2: a single dose of oral xevinapant 200 mg formulated as Solution A with or without pantoprazole (40 mg for 6 days [twice daily on Days 2 to 5, once daily on Days 1 and 6])
- Stage 1 included 4 treatment periods lasting 4 days (**Supplementary Table 1**), and stage 2 included 2 treatment periods lasting 6 days

- The primary endpoint was the pharmacokinetics (PK) of xevinapant and its metabolite (D-1143-MET1)
- In both stages, PK samples were collected within 72 hours of the final dose in each treatment period
 - Nonparametric superposition was used to simulate steady-state concentration-time profiles based on observed (single-dose) profiles
 - PK parameters were derived by noncompartmental analyses
- Blood samples for cIAP1 (the PD biomarker in peripheral blood mononuclear cells [PBMCs]) were collected over 72 hours of the final dose in treatment period 1 in stage 1 only
 - cIAP1 and β-actin levels in PBMCs were determined by Western blot; cIAP1 levels were normalized to β-actin levels
- The bioequivalence margin used to assess bioavailability was 0.80–1.25

Figure 1. Study design*.

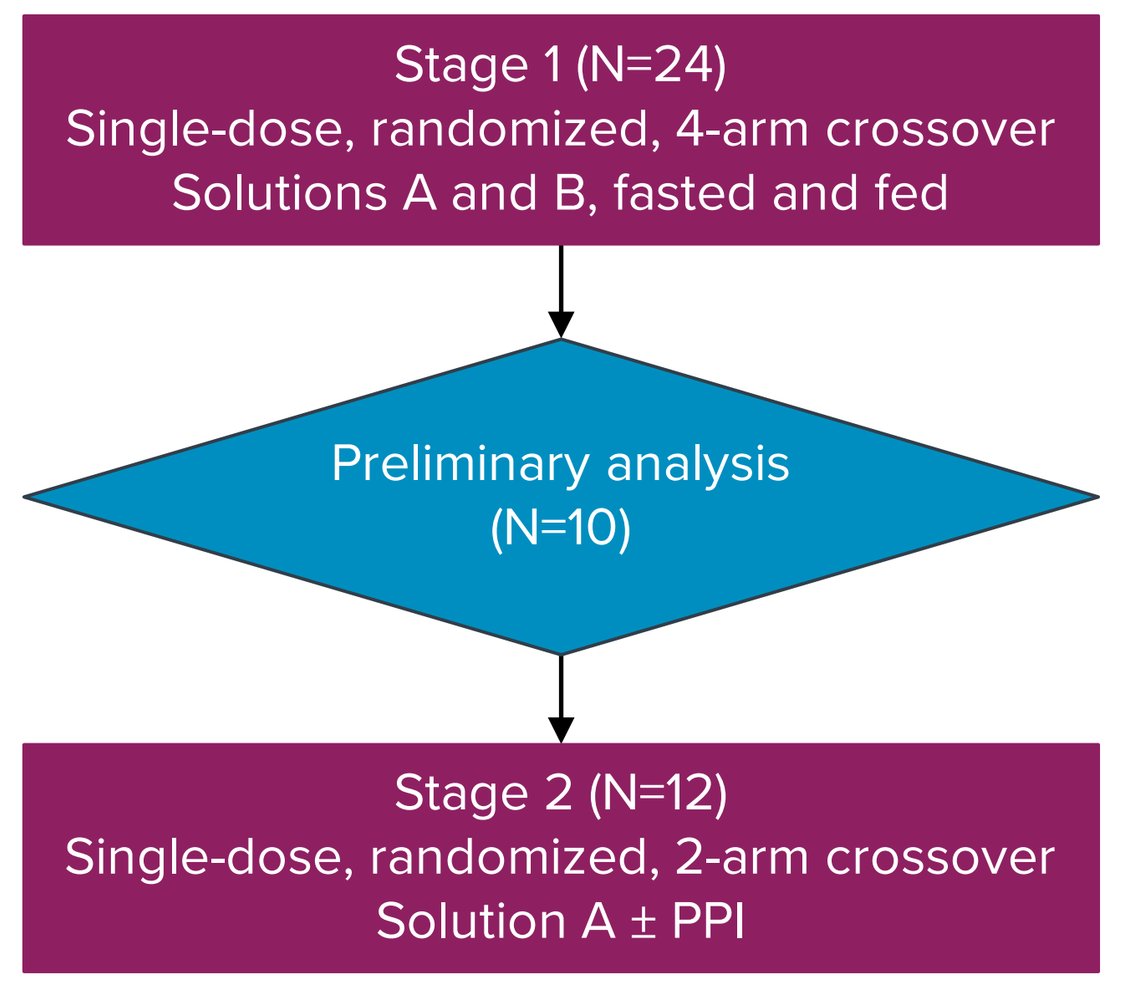
Stage 1 (N=24)
Single-dose, randomized, 4-arm crossover
Solutions A and B, fasted and fed
Preliminary analysis
(N=10)
Stage 2 (N=12)
Single-dose, randomized, 2-arm crossover
Solution A ± PPI

PPI, proton pump inhibitor.
*An additional stage (stage 1b) was to be performed in case results of an interim analysis at the end of stage 1 were inconclusive and suggested that a greater number of subjects was needed to prove exchangeability or a clinically relevant difference between formulations. Stage 1b was not required.

RESULTS

Volunteer disposition

- Stage 1 consisted of 26 volunteers (mean age, 44 years; 85% male)
 - 2 discontinuations were reported due to volunteer withdrawal and an issue with obtaining blood samples; however, all volunteers completed xevinapant treatment
- Stage 2 consisted of 12 volunteers (mean age, 55 years; 83% male)
 - All volunteers received the full dose of study treatment

PK: stage 1

- Following a single dose of xevinapant 200 mg, exposures in volunteers receiving Solution A under fasted conditions were consistent with those observed previously in patients with cancer (**Table 1**)
- Food (high-fat meal) did not meaningfully alter the extent of xevinapant absorption, as evidenced by no change in the area under the plasma concentration-time curve (AUC) in fed vs fasted volunteers; however, the rate of absorption was affected—the time to reach C_{max} (t_{max}) was delayed, and C_{max} was decreased by 39% (**Figure 2A** and **Table 2**)
- PK simulations indicated no relevant change in xevinapant AUC and no impact on target engagement at steady state with food (**Table 3**)
- The PK profile of xevinapant's metabolite, D-1143-MET1 (inactive on IAP1/2), was affected by food, as shown by a 64% decrease in C_{max} and 46% decrease in AUC_∞ in fed vs fasted volunteers (**Figure 2B** and **Supplementary Table 2**)

Bioavailability of Solution A vs Solution B

- Both xevinapant formulations had a comparable $AUC_{0–t}$, AUC_∞ and C_{max}, with ratios and 90% CIs within the bioequivalence margin (0.80–1.25) (**Supplementary Table 3**)
- Inter-individual variability was not impacted by the change in the excipients between Solution A and Solution B

Table 1. PK parameters in volunteers and patients with cancer receiving Solution A under fasted conditions in stage 1.

Geometric mean (GCV %)	Patients with cancer*	Volunteers
C_{max}, ng/mL	1,610 (45.8)	1,934 (42.6)
AUC_∞, ng•h/mL	11,900 (46.0)†	10,018 (34.8)

AUC, area under the plasma concentration-time curve; C_{max}, maximum plasma concentration; **GCV**, geometric coefficient of variation; **PK**, pharmacokinetic.
*Derived from the population PK modeling from NCT01078649 and NCT02022098.[11]
†AUC_∞ after the first dose is equivalent to $AUC_{0–24}$ at steady state.

Figure 2. Plasma concentrations of xevinapant (A) and D-1143-MET1 (B) over time for Solution A in fasted and fed conditions in stage 1.

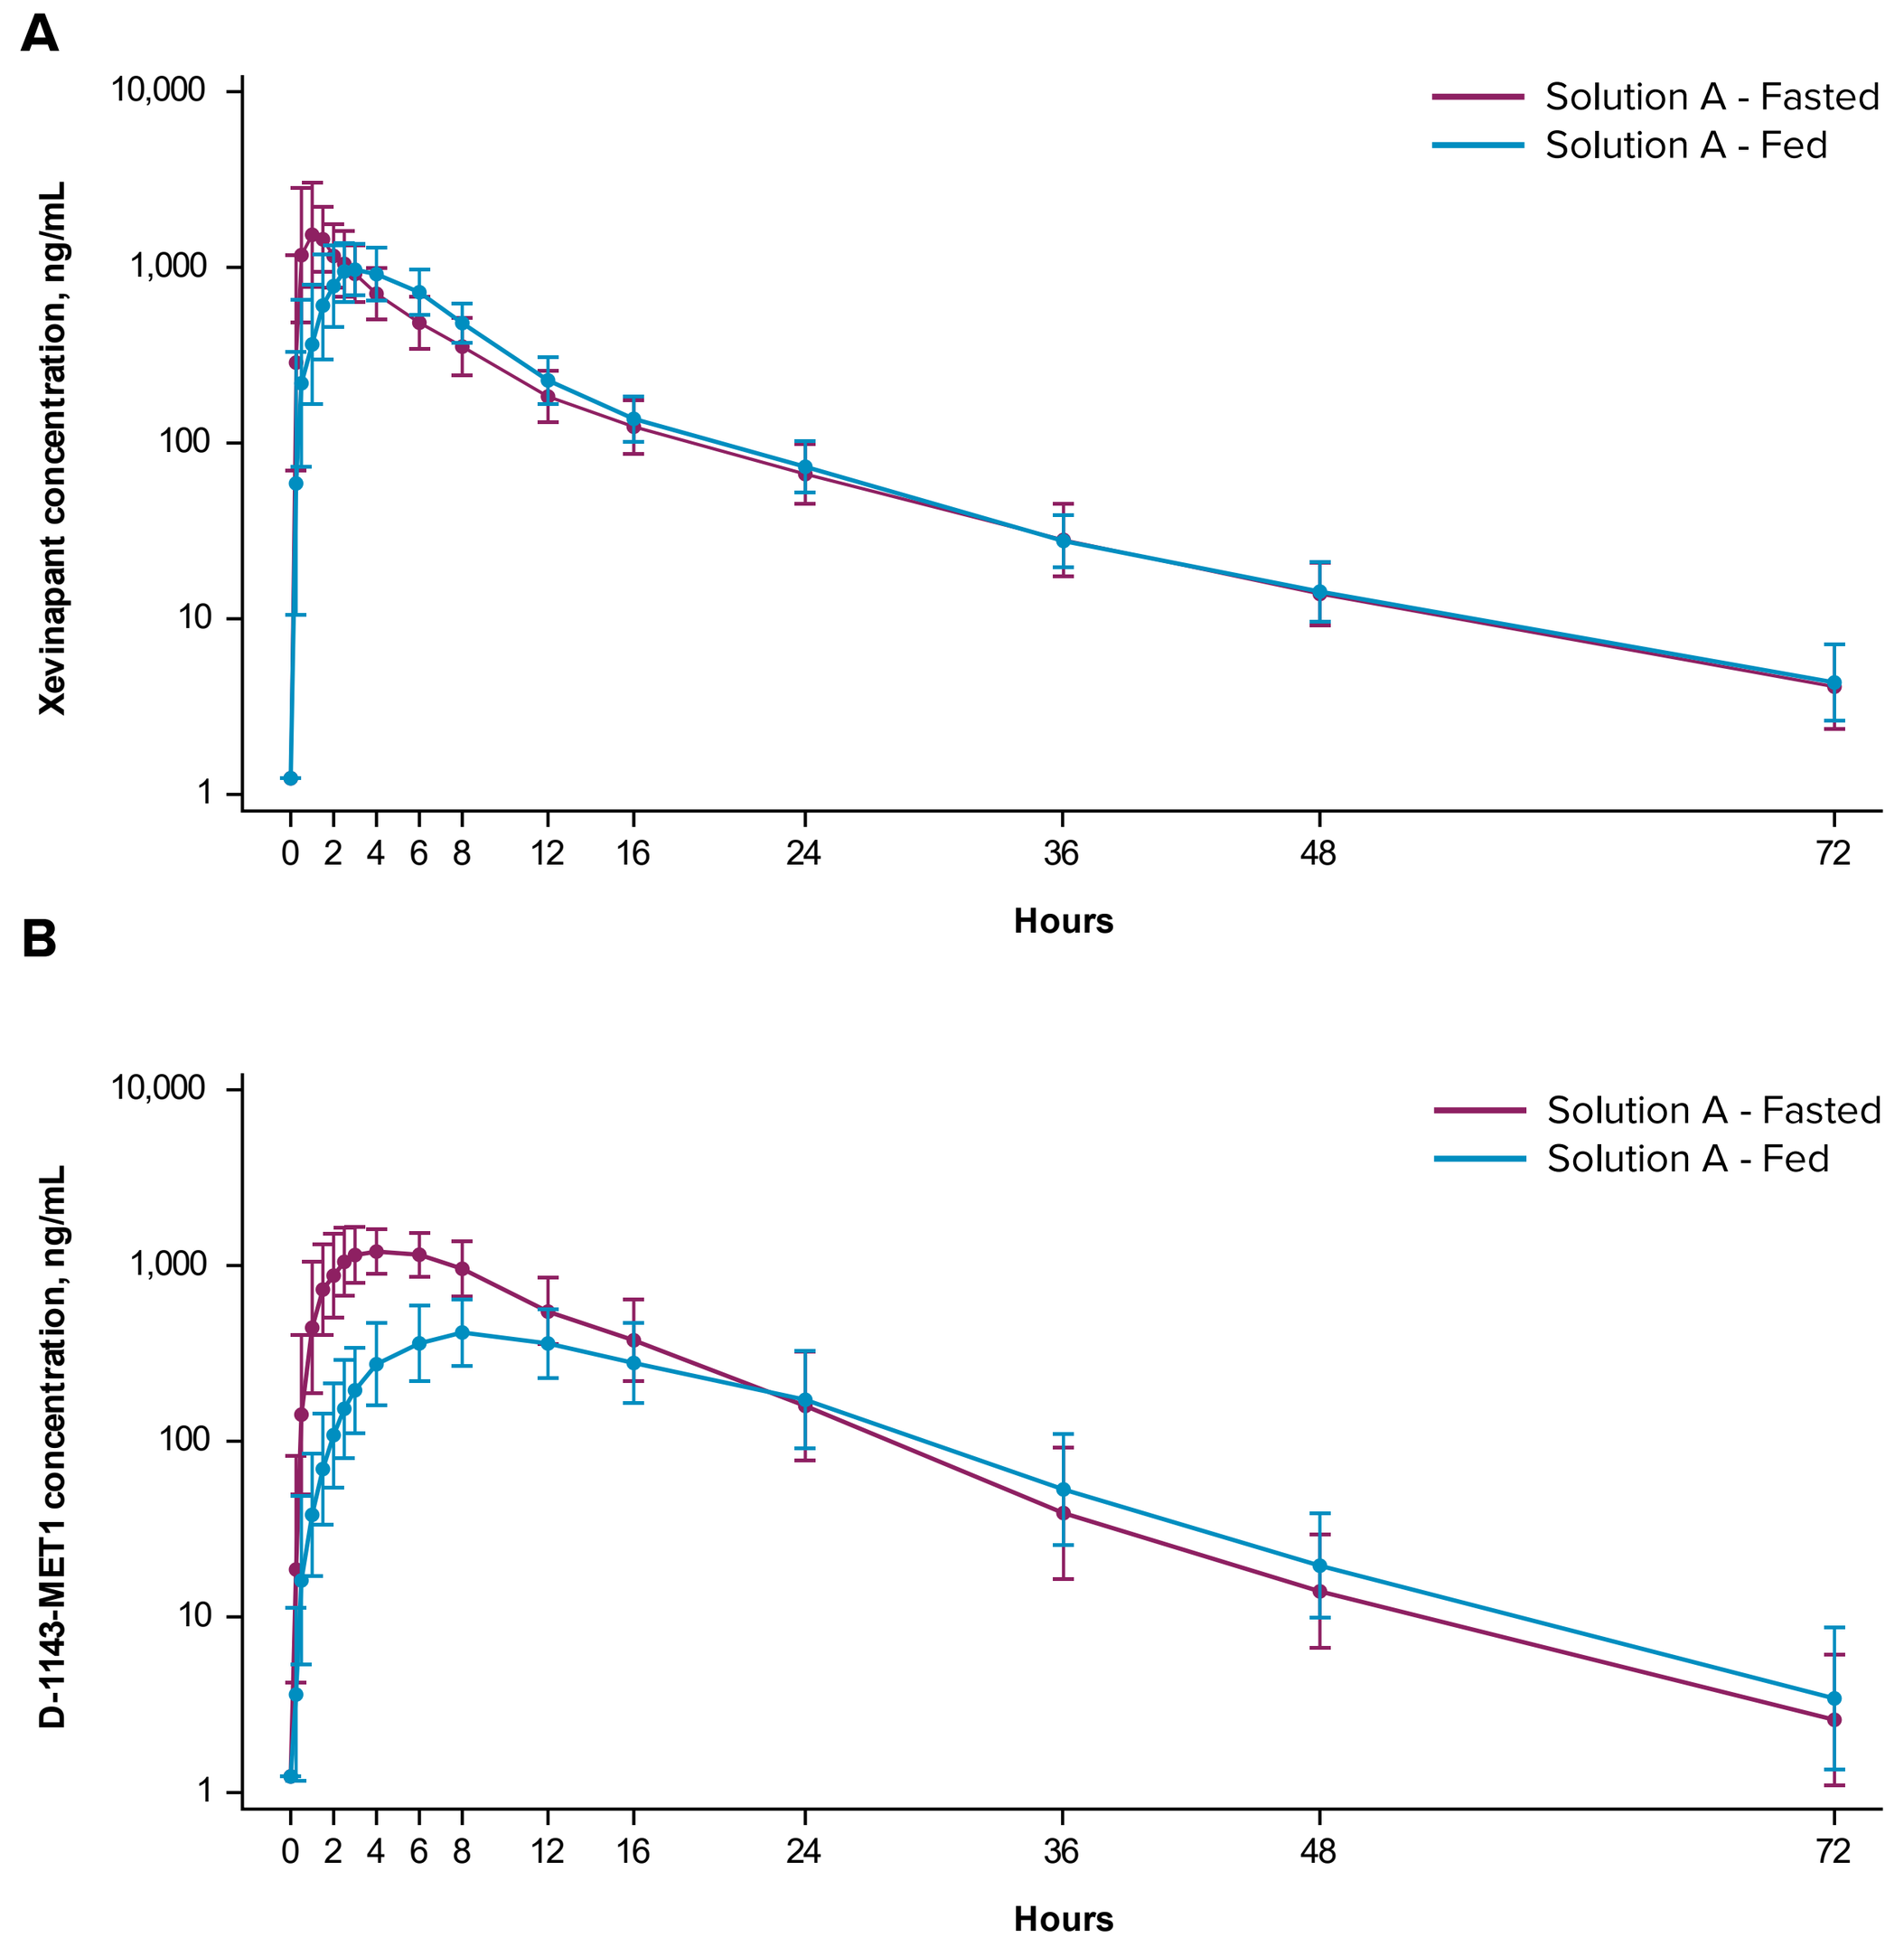
A
Xevinapant concentration, ng/mL
10,000
1,000
100
10
1
0 2 4 6 8 12 16 24 36 48 72
Hours
Solution A - Fasted
Solution A - Fed
B
D-1143-MET1 concentration, ng/mL
10,000
1,000
100
10
1
0 2 4 6 8 12 16 24 36 48 72
Hours
Solution A - Fasted
Solution A - Fed

Each data point represents geometric mean (geometric SD).

Table 2. Plasma xevinapant PK parameters after a single dose of Solution A.

Geometric mean (GCV %)	Fasted (n=25)	Fed (n=24)
$AUC_{0–t}$, ng•h/mL	9,909 (35.2)	9,733 (30.5)
AUC_∞, ng•h/mL	10,018 (34.8)	9,836 (30.4)
C_{max}, ng/mL	1,934 (42.6)	1,130 (37.9)
t_{max}, median (range), h	1.0 (0.5–4.0)	2.75 (1.5–6.0)
$t_{1/2}$, h	13.3 (18.8)	13.3 (16.8)
CL/F, L/h	20.0 (34.8)	20.3 (30.4)
V_z/F, L	382 (40.1)	389 (29.1)

AUC, area under the plasma concentration-time curve; **CL/F**, apparent total body clearance; C_{max}, maximum plasma concentration; **F**, bioavailability; **GCV**, geometric coefficient of variation; **PK**, pharmacokinetic; $t_{1/2}$, elimination half-life; t_{max}, time to reach C_{max}; V_z, apparent volume of distribution.

PD: stage 1

- No relevant differences in PD were observed across treatments in fasted and fed conditions (**Figure 3**)
- The mean cIAP1:β-actin ratio decreased after completion of each treatment, indicating robust target engagement and PD effect
- The cIAP1:β-actin ratio was lowest 4–24 hours after treatment
- cIAP1:β-actin levels tended to increase 24 hours after treatment; however, they had not returned to baseline by 72 hours after treatment, consistent with PK/PD model predictions based on patient data[11]

PK: stage 2

- The PK profiles of xevinapant and D-1143-MET1 were similar in the presence and absence of pantoprazole, with C_{max} observed approximately 1 and 4 hours after treatment, respectively (**Figure 4**)
- Xevinapant exposure was comparable with and without pantoprazole, as 90% CIs of AUC and C_{max} were within the bioequivalence margin (0.80–1.25)

Safety

- Stage 1: Both xevinapant formulations were safe and well tolerated in both fasted and fed volunteers
- Stage 2: Solution A was safe and well tolerated in the presence or absence of pantoprazole

Table 3. Plasma PK parameters of Solution A at steady state based on PK simulations.

Geometric mean (GCV %)	Fasted (n=25)	Fed (n=24)
$C_{max,SS}$, ng/mL	2,020 (42.2)	1,210 (37.5)
$C_{trough,SS}$, ng/mL	87.2 (1.48)	95.5 (1.40)
$AUC_{0–24,SS}$, ng•h/mL	10,000 (34.8)	9,830 (30.4)

$AUC_{0–24,SS}$, area under the plasma concentration-time curve at steady state; $C_{max,ss}$, maximum plasma concentration at steady state; $C_{trough,SS}$, predose trough concentration at steady state; **GCV**, geometric coefficient of variation; **PK**, pharmacokinetic.

Figure 3. cIAP1:β-actin ratio over time in stage 1.

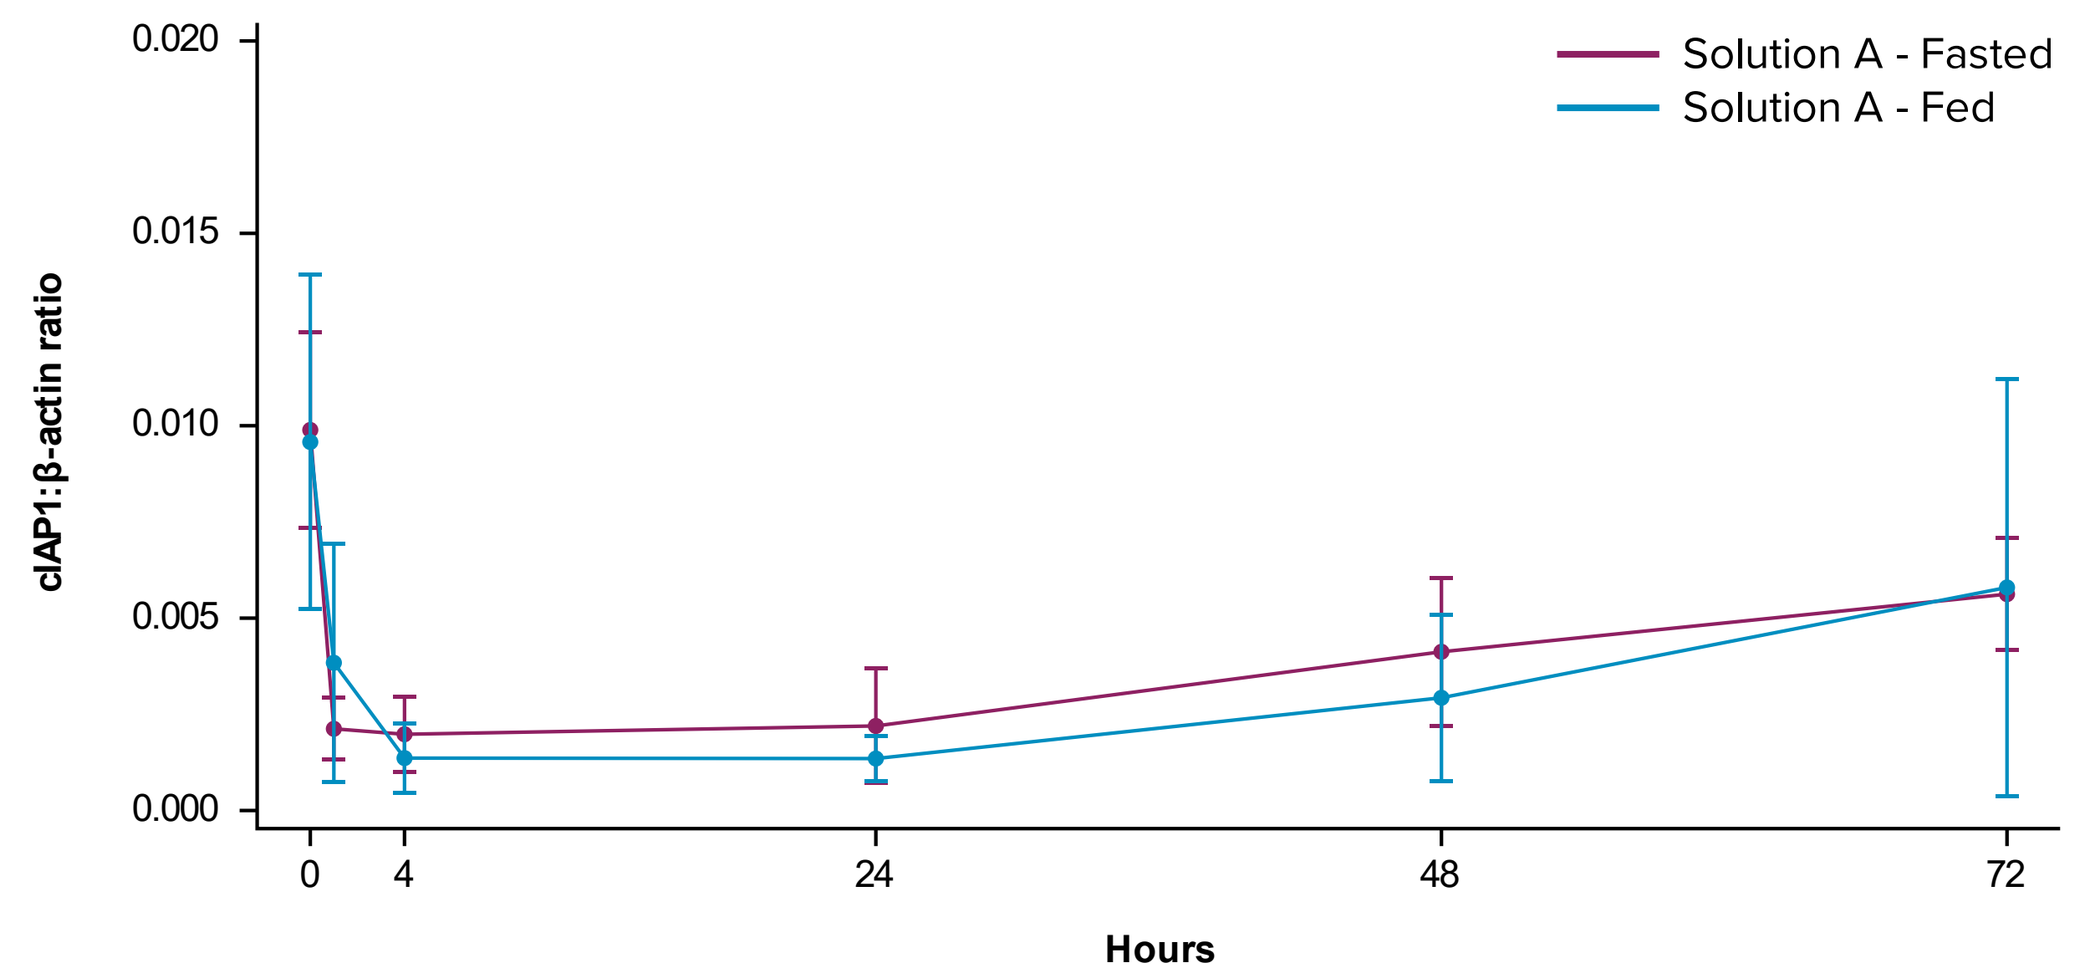
cIAP1:β-actin ratio
0.020
0.015
0.010
0.005
0.000
0 4 24 48 72
Hours
Solution A - Fasted
Solution A - Fed

Each data point represents mean (SD).
cIAP1, cellular inhibitor of apoptosis protein 1.

Figure 4. Xevinapant plasma concentrations over time with or without PPI in stage 2.

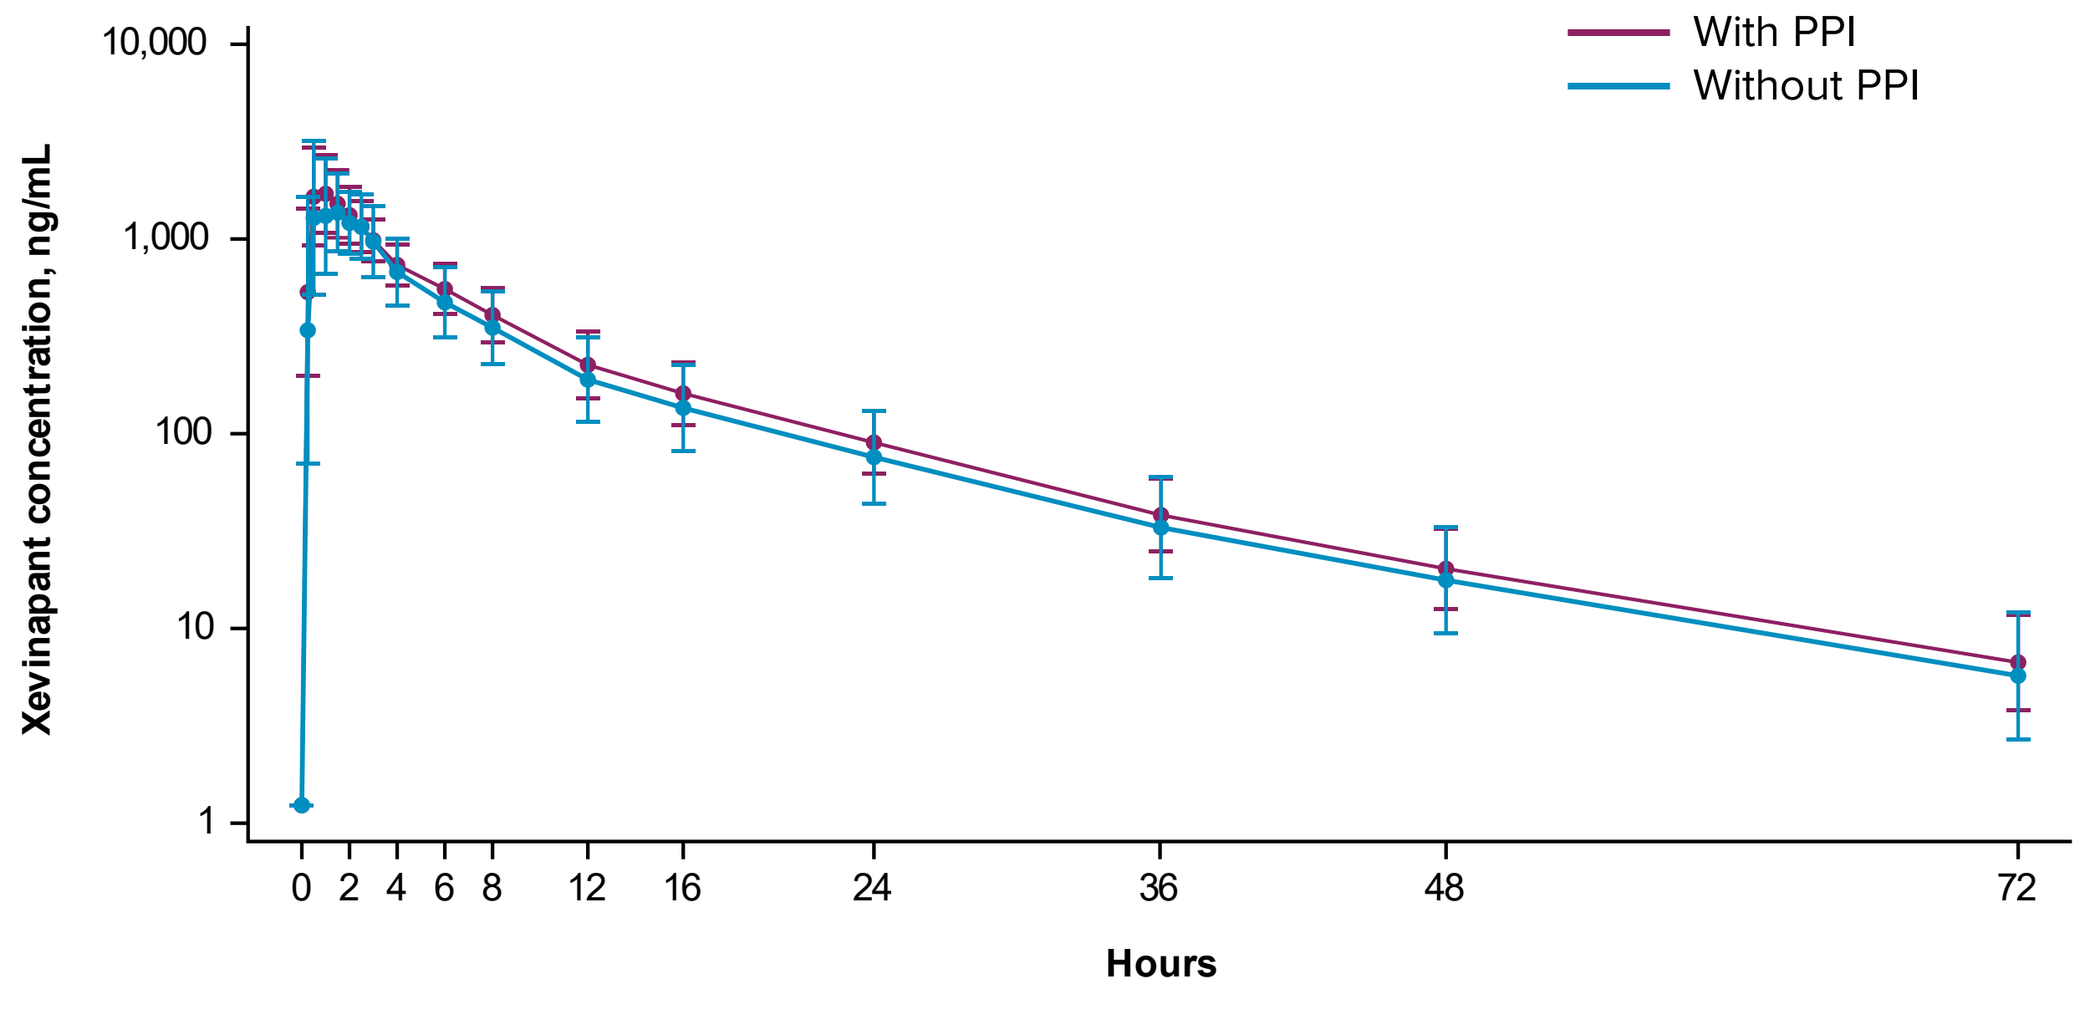
Xevinapant concentration, ng/mL
10,000
1,000
100
10
1
0 2 4 6 8 12 16 24 36 48 72
Hours
With PPI
Without PPI

Each data point represents geometric mean (geometric SD).
PPI, proton pump inhibitor.

REFERENCES: 1. Saba NF, *et al. Int J Radiat Oncol Biol Phys.* 2024;118:e21. **2.** Ferris RL, *et al. Future Oncol.* Published online January 10, 2024. **3.** Matzinger O, *et al. Radiother Oncol.* 2015;116:495–503. **4.** Cai Q, *et al. J Med Chem.* 2011;54:2714–26. **5.** Thibault B, *et al. Sci Rep.* 2018;8:17862. **6.** Dougan SK, *et al. Immunotherapy.* 2018;10:787–96. **7.** Gomez-Roca C, *et al. Clin Transl Sci.* 2022;15:55–62. **8.** Yu H, *et al. Signal Transduct Target Ther.* 2020;5:209. **9.** Sun XS, *et al. Lancet Oncol.* 2020;21:1173–87. **10.** TAO Y, *et al. Eur J Cancer.* 2023;183:24–37. **11.** Vugmeyster Y, *et al. Clin Pharmacol Ther.* 2024;115:52–61.
DISCLOSURES: **V. Nicolas-Métral, D. Spaggiari, N. Wiedemann, R. Crabbé, H. Nauwelaerts,** and **C. Haefliger** report employment with Debiopharm International SA. **E. Rouits, C. Riff,** and **P. Colin** report employment by Debiopharm International SA at the time the study was conducted. **E. Harrison-Moench** and **A. Schroeder** report employment with the healthcare business of Merck KGaA, Darmstadt, Germany. **Y. Vugmeyster** reports employment with EMD Serono, Billerica, MA, USA. **ACKNOWLEDGMENTS:** The authors thank the volunteers, study investigators, and study personnel across all sites for participating in this study. The study was conceptualized and sponsored by Debiopharm International SA; since December 2021, the study has been sponsored by the healthcare business of Merck KGaA, Darmstadt, Germany (CrossRef Funder ID: 10.13039/100009945). Medical writing support was provided by Jamie Ratcliffe of Nucleus Global and was funded by the healthcare business of Merck KGaA, Darmstadt, Germany.

Abstract No. CT140. Presented at the AACR Annual Meeting 2024, April 5–10, 2024; San Diego, CA, USA.