AACR Annual Meeting 2024, April 5–10, 2024; San Diego, CA, USA

Impact of food and high gastric pH on the bioavailability of the WEE1 inhibitor Debio 0123 assessed in a Phase 1 dose escalation study

ABSTRACT CT064

Anne Bellon[1], Omar Saavedra[2], Ingrid M. E. Desar[3], Mathilde Jalving[4], Jourik A. Gietema[4], Carla Van Herpen[3], Stefan Van Ravensteijn[3], Esteban Rodrigo Imedio[1], Sylvia Van Haren[1], Melanie Wirth[1], Sandrine Micallef[1], Vito Dozio[1], Rikke Frederiksen Franzen[1], Marie-Claude Roubaudi-Fraschini[1], Valerie Nicolas-Metral[1], Hans Gelderblom[5].

[1]Debiopharm International S.A., Lausanne, Switzerland, [2]Vall d'Hebron University Hospital, Barcelona, Spain, [3]Radboud University Medical Center, Nijmegen, Netherlands, [4]University Medical Center Groningen (UMCG), Groningen, Netherlands, [5]Leiden University Medical Center, Leiden, Netherlands

BACKGROUND

Debio 0123, an oral brain-penetrant, highly selective WEE1 inhibitor, is currently in clinical development, both as monotherapy and in combination, for the treatment of patients with solid tumors. Food and gastric acid-reducing agents e.g. proton pump inhibitors (PPIs) may modify the solubility, bioavailability, safety and efficacy of oral cancer drugs. Polymedication in cancer patients is common and food restrictions can impact patient compliance and wellbeing. In order to adequately inform drug dosing in clinical trials, effects of food and high gastric pH on Debio 0123 bioavailability were studied over three cycles in patients participating in arm B of a dose escalation Phase 1 study in combination with carboplatin (CTID NCT03968653).

The Phase 1 study was composed of 2 arms with different schedules. While Arm A was the first-in-patient study of Debio 0123, in arm B, Debio 0123 was given once daily on Days 1-3 and 8-10 of each 21-day cycle in combination with carboplatin on Day 1. Standard administration was in fasted conditions and PPIs were prohibited within 72h before and 4h after dosing. In arm B, dose-limiting toxicities were evaluated in cycle 1. The effect of food and high-gastric pH were assessed in cycles 2 and 3, respectively.

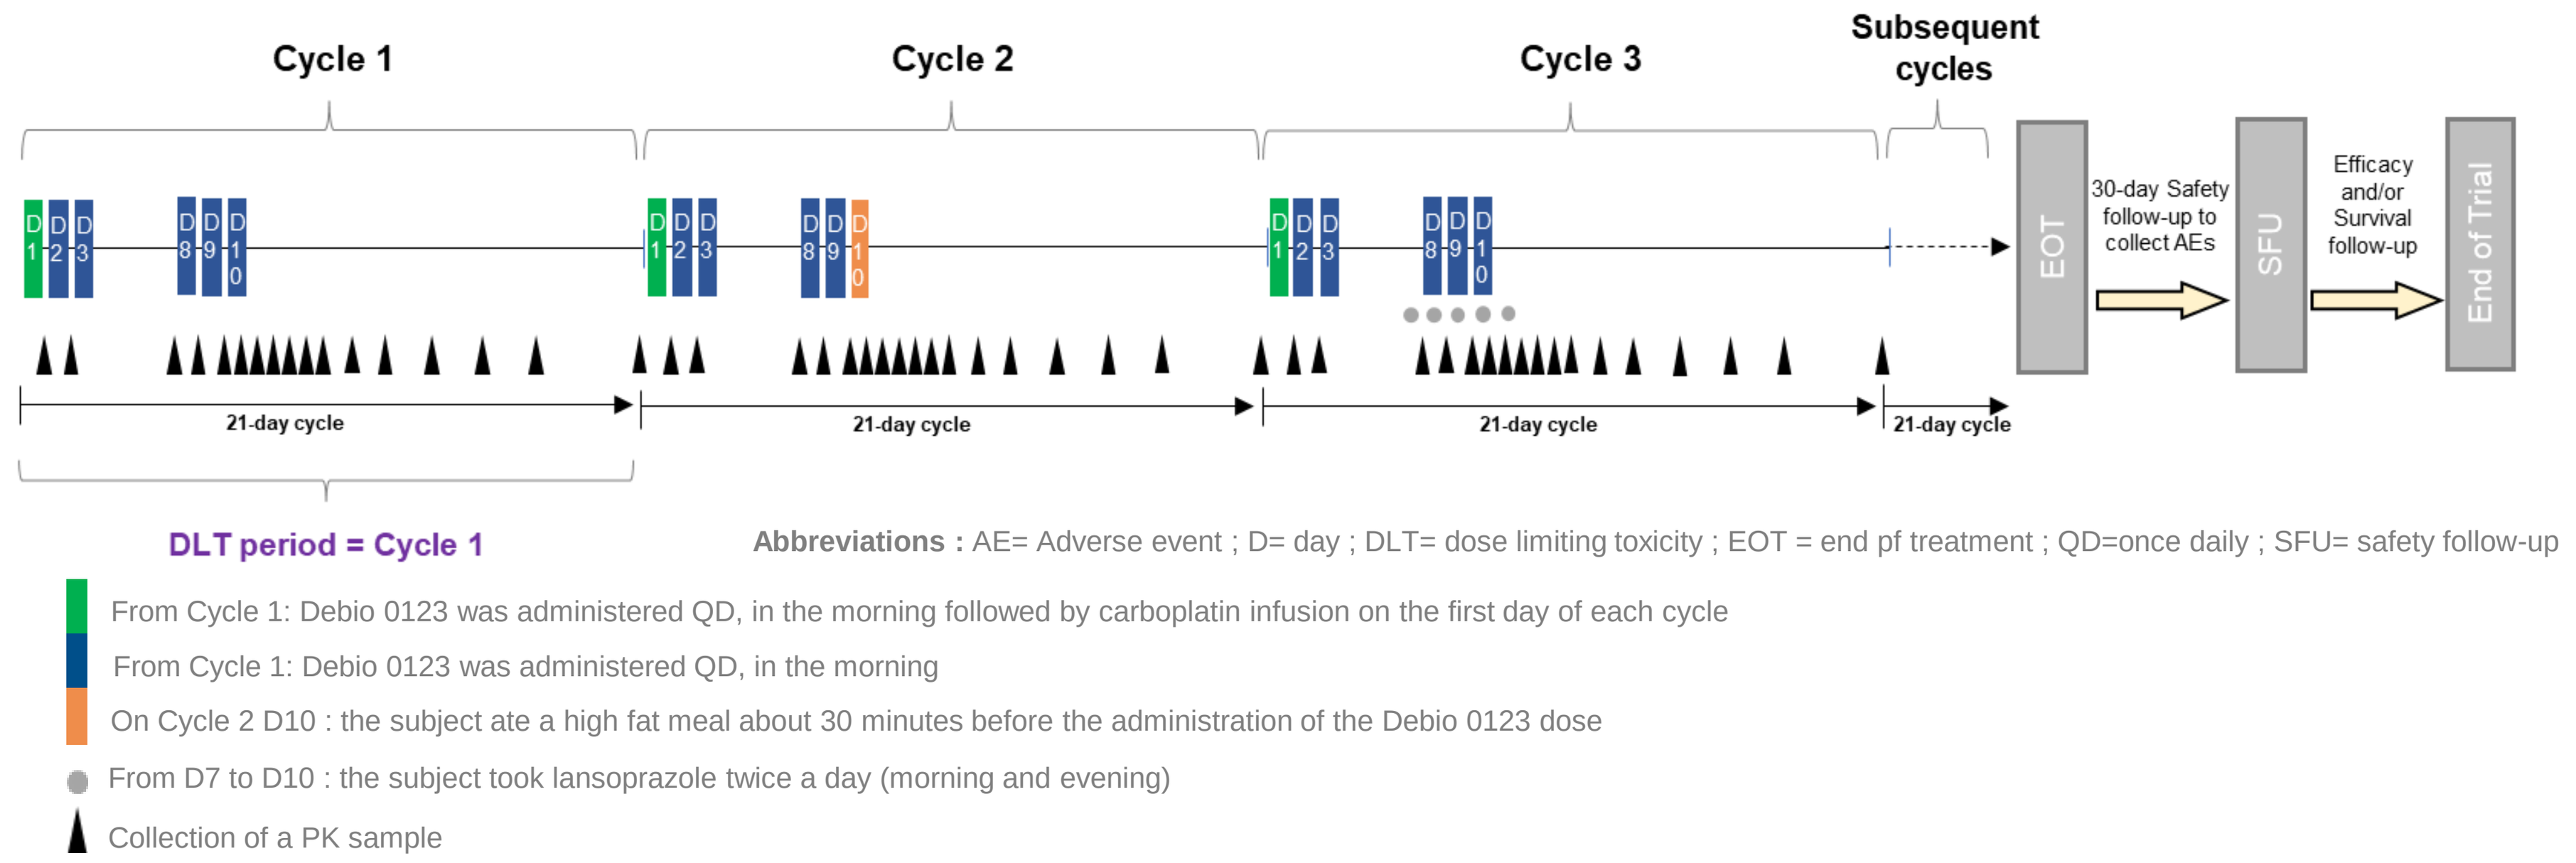
Cycle 1
Cycle 2
Cycle 3
Subsequent cycles
EOT
30-day Safety follow-up to collect AEs
SFU
Efficacy and/or Survival follow-up
End of Trial
21-day cycle
DLT period = Cycle 1
Abbreviations : AE= Adverse event ; D= day ; DLT= dose limiting toxicity ; EOT = end pf treatment ; QD=once daily ; SFU= safety follow-up
From Cycle 1: Debio 0123 was administered QD, in the morning followed by carboplatin infusion on the first day of each cycle
From Cycle 1: Debio 0123 was administered QD, in the morning
On Cycle 2 D10 : the subject ate a high fat meal about 30 minutes before the administration of the Debio 0123 dose
From D7 to D10 : the subject took lansoprazole twice a day (morning and evening)
Collection of a PK sample

Figure 1. *Study design of the Phase 1 study showing the dosing schedule of the arm B over the first three cycles and PK samples collection*

METHODS

Each patient in arm B of the dose escalation study received Debio 0123 and carboplatin AUC 5 mg/mL•min on Day 1 of each cycle of 21 days, followed by several doses of Debio 0123 as illustrated in Figure 1. Dose levels studied in the 17 patients included in the arm were 300, 400, 520 and 720 mg.

On Day 10 of cycle 2 exclusively, the patient was offered a high fat meal 30 min before Debio 0123 administration. High fat meal contained a total of 800-1000 Kcal including 55 to 65 g of fat. Several options including vegetarian meal were proposed. Most of the patients accepted to eat at least 80% of the proposed meal.

On cycle 3, lansoprazole, a proton pump inhibitor, was administered at a dose of 30 mg in the mornings and evenings of D7, 8, 9, and 10. High gastric pH was reached before Day 10 and environment was kept by readministering lansoprazole on D11 as well.

The same PK samples were collected post-D10 dose in cycle 1, 2 and 3: 6 samples on Day 10, followed by 1 sample on Day 11, 12, 14, 16, 18 and predose on Day 1 in the next cycle. Trough levels before each dose of the three cycles were collected to assess potential accumulation between cycles. Figure 2 illustrates dose-normalized mean trough levels on Day 3 and 8 of all patients included in at least one comparison. Box plots clearly show an absence of Debio 0123 accumulation between cycles, allowing for inter-cycle comparisons.

PK parameters (area under the curve from D10 until predose in the next cycle (AUC_{last}), maximum concentration (C_{max})) were determined post-Day 10 dose and compared. Each patient was his/her own control.

Individual PK profiles and PK parameters were determined using Phoenix WinNonlin version 8.3.

The intra-individual difference in exposure (AUC_{last} and C_{max}) between cycle 2 and cycle 1 for food effect or cycle 3 and cycle 1 for high gastric pH effect was calculated after having assessed the absence of any intercurrent event(s).

The following intercurrent events precluding use of at least one cycle were: vomiting events within 2h of dose administration on D10 of any cycle, reduced dose on D8, 9 or 10, absence of several main PK samples collection (e.g. due to COVID isolation), use of a prohibited medication with impact on absorption, distribution, metabolism or elimination, patient out of study for any reason before cycle 2 or 3 or refusal of food / PPI intake from the patient.

Data from 5 cohorts, 4 dose levels (n=17) resulted in 7 and 5 pairwise comparisons for food and for high gastric pH effect respectively.

No accumulation of Debio 0123 in plasma between cycles

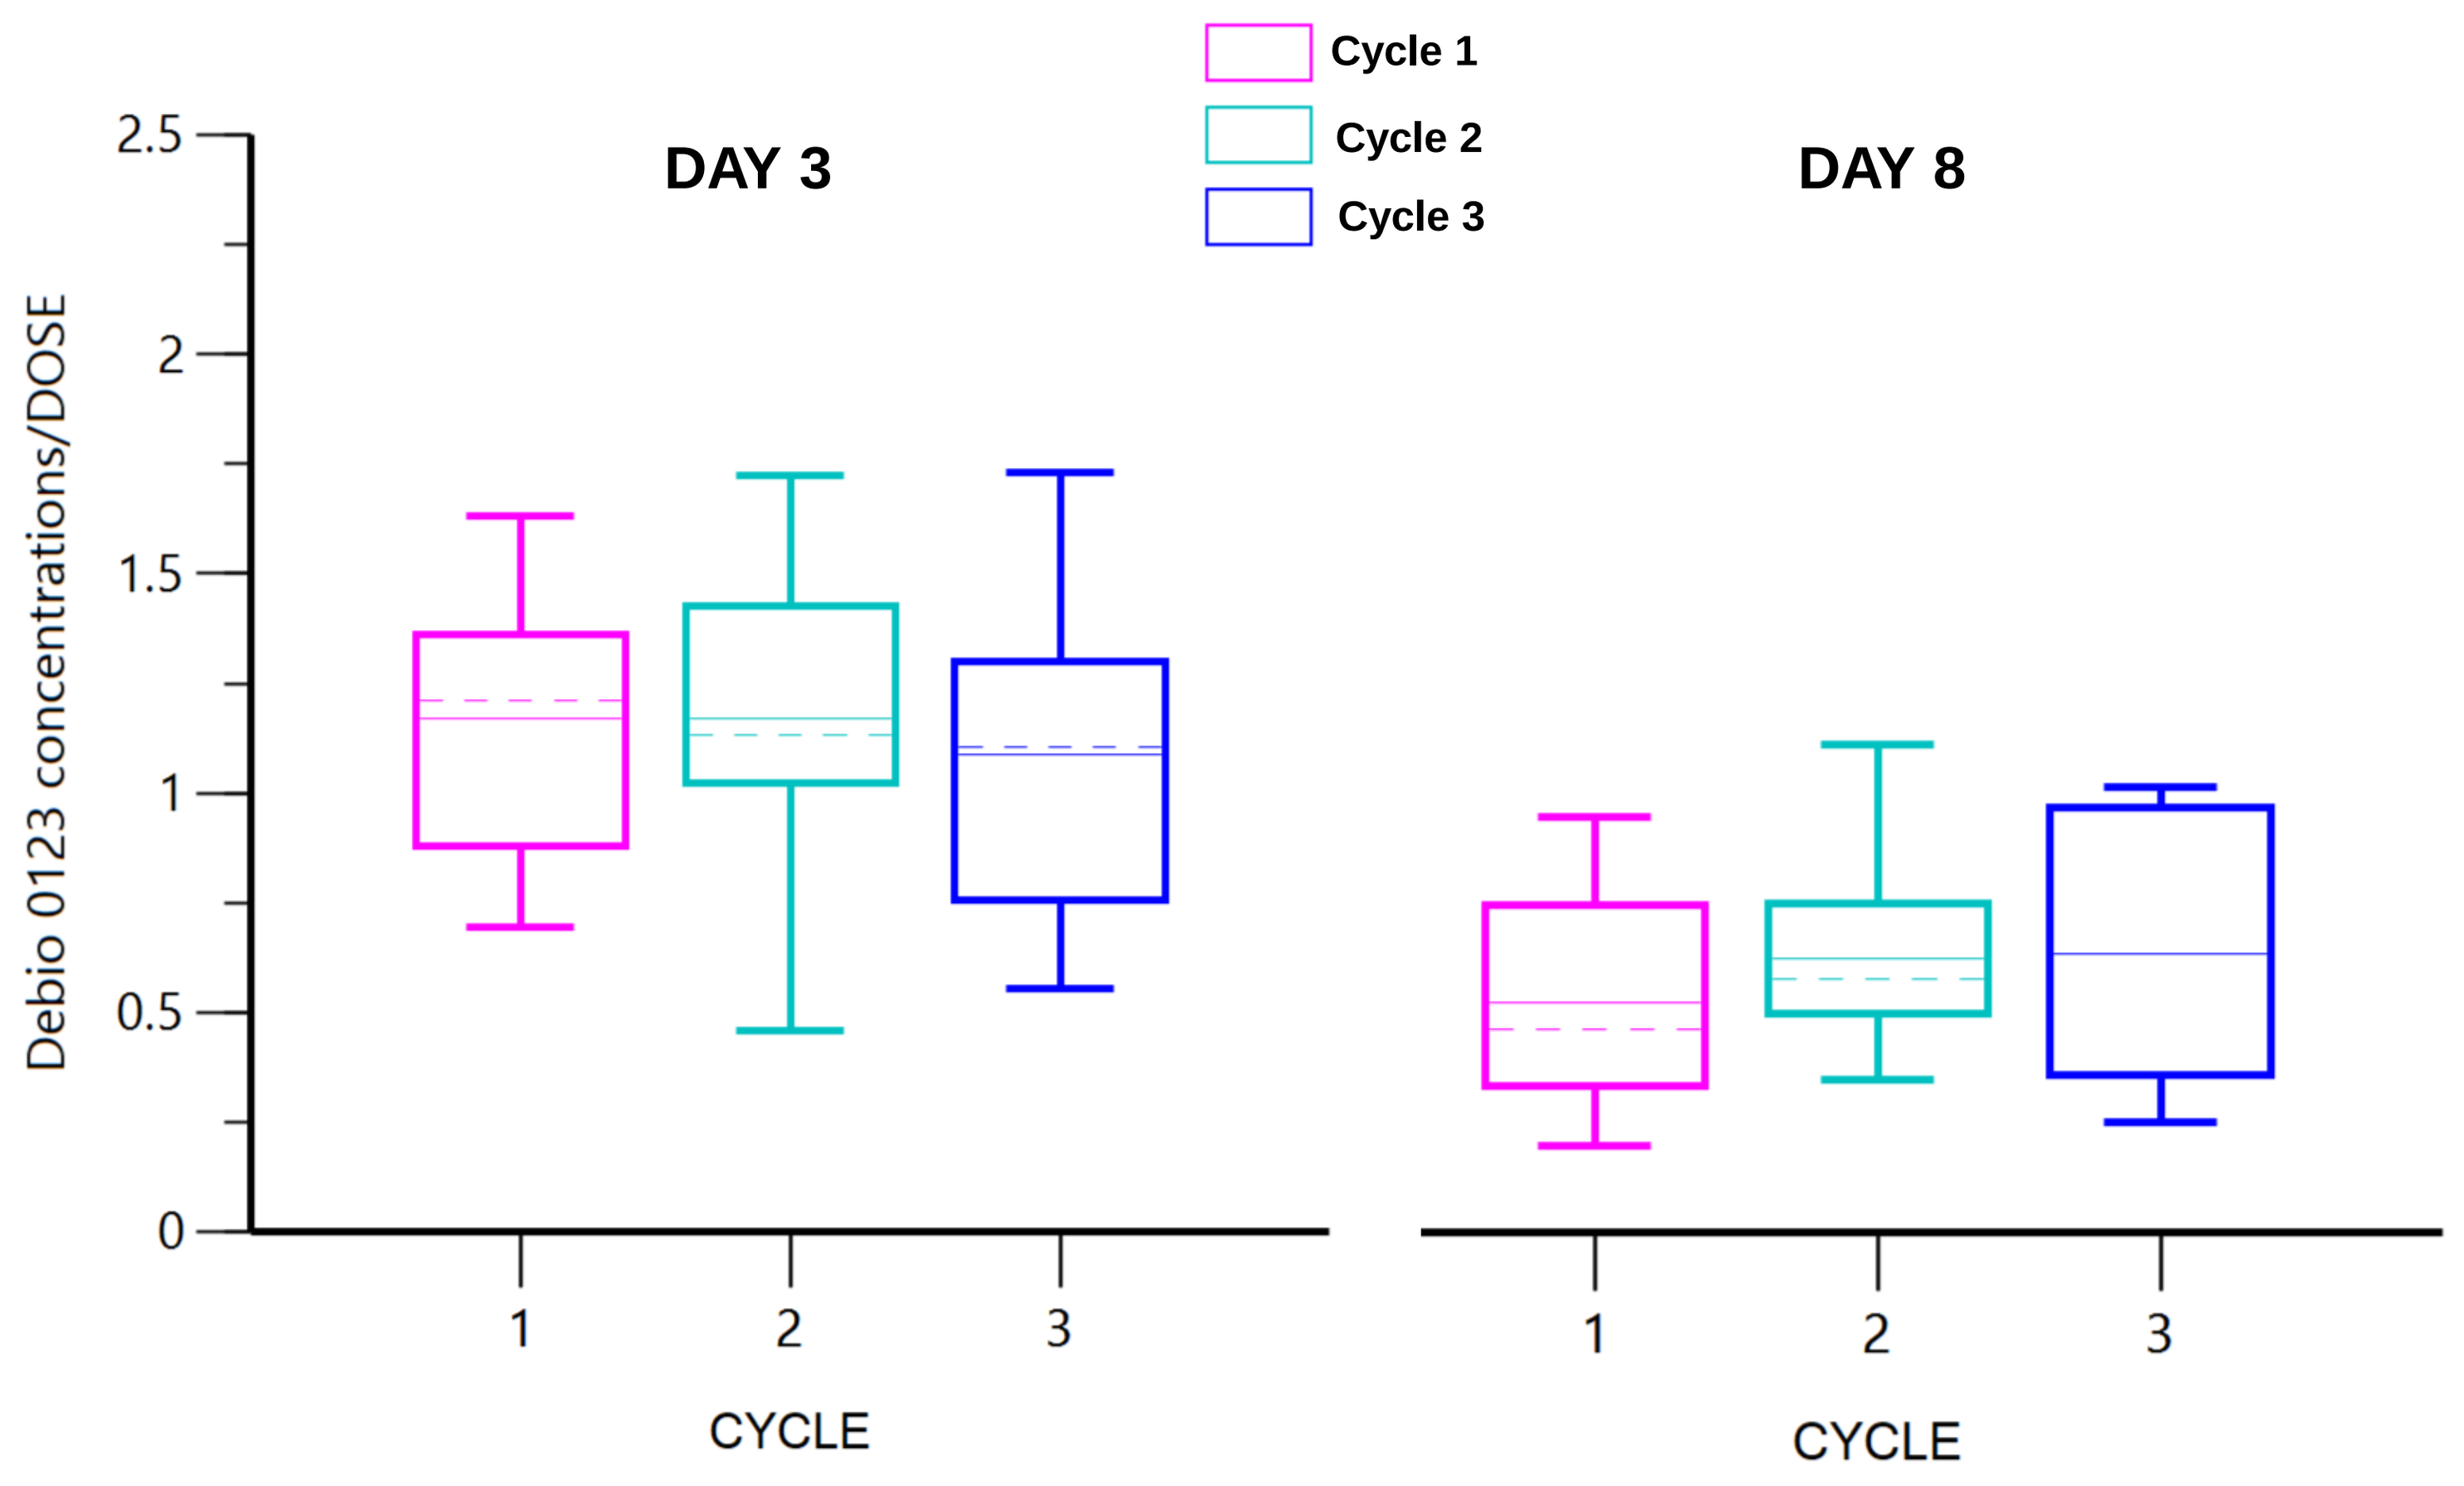
Cycle 1
Cycle 2
Cycle 3
DAY 3
DAY 8
Debio 0123 concentrations/DOSE
2.5
2
1.5
1
0.5
0
1
2
3
CYCLE

Figure 2. *Box plots representing trough levels normalized by the dose of all patients included in at least one comparison on Day 3 (left panel) and Day 8 (right panel)*

RESULTS

Out of the 17 patients included in the arm B, 8 coming from 3 dose level cohorts had at least one pairwise comparison.

Results are presented in Figure 3 and Table 1.

- Food effect was assessed in 7 study participants.

 Change in Debio 0123 plasma exposure between cycle 2 (fed) and 1 (fasted) was low, within expected bioanalytical assay variability.

- High gastric pH influence was assessed in 5 study participants.

 Debio 0123 plasma exposure was consistently reduced by about 40% on cycle 3 (with PPI) versus cycle 1 (without PPI).

Intra-individual comparison of PK profiles

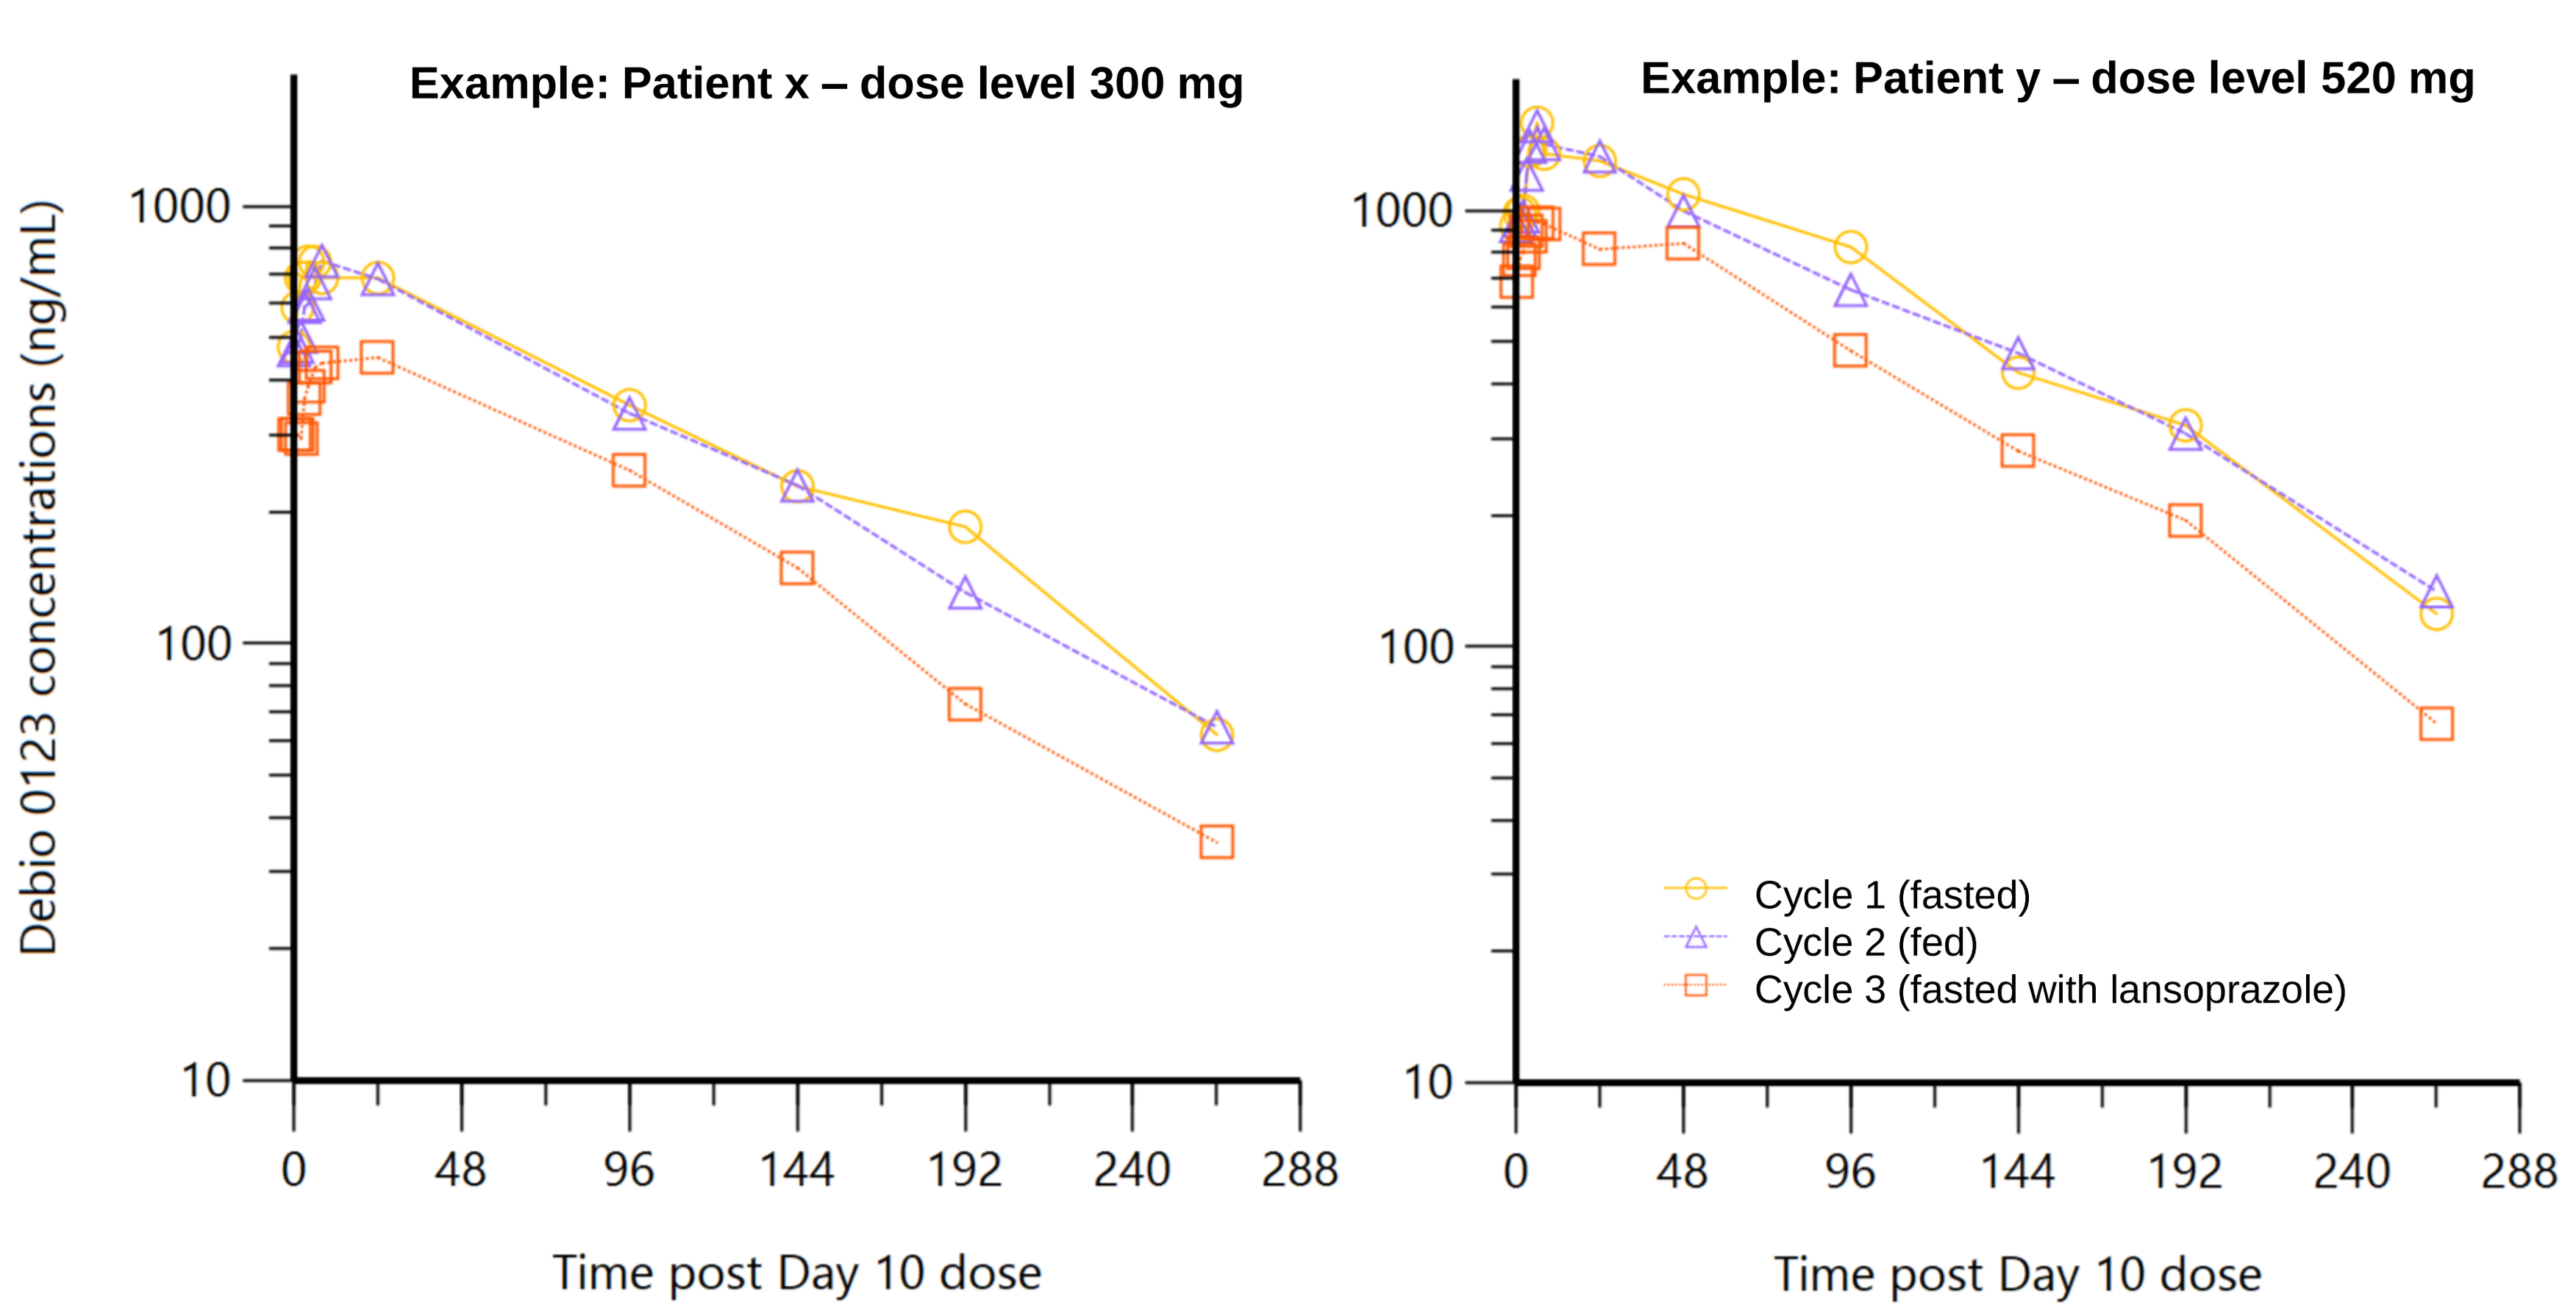
Example: Patient x – dose level 300 mg
Example: Patient y – dose level 520 mg
Debio 0123 concentrations (ng/mL)
1000
100
10
0
48
96
144
192
240
288
Time post Day 10 dose
Cycle 1 (fasted)
Cycle 2 (fed)
Cycle 3 (fasted with lansoprazole)

Figure 3. *Debio 0123 PK profiles after the last dose of cycle 1, 2 and 3*

Food does not impact overall exposure

	AUC_{last}	C_{max}
High fat meal (n=7)	2% (-16% +24%)	-9% (-26% 0%)

High gastric pH significantly reduces exposure

	AUC_{last}	C_{max}
Lansoprazole for 4 days (n=5)	-34% (-45% -23%)	-37% (-53% -20%)

Table 1. *Mean reduction (min-max) in Debio 0123 exposure - AUC_{last} and C_{max} - after high fat meal intake (upper panel) or under high gastric pH conditions (lower panel) achieved after PPI administration*

CONCLUSIONS AND PERSPECTIVES

- Food did not affect pharmacokinetics of Debio 0123 in patients with solid tumors. These results allow to administer Debio 0123 regardless of food intake. For patient convenience and well-being, administration with a light meal is being implemented in ongoing and new Debio 0123 clinical trials.
- Debio 0123 oral bioavailability is pH-dependent. Gastric acid-reducing agents such as proton pump inhibitors remain prohibited concomitantly with Debio 0123.

CONTACT

Debiopharm International S.A.,
Lausanne, Switzerland.
www.debiopharm.com
anne.bellon@debiopharm.com

DOWNLOAD

This poster is available via:
www.debiopharm.com/medias/publications

Acknowledgments : The authors would like to thank all study participants and their families, medical and paramedical staff of the investigational sites as well as the Debio 0123-101 Clinical Trial Team