The OxtendTM-01 study: Debio 4126, a new 12-week octreotide formulation, provides maintenance of disease control in patients with acromegaly switching from long-acting somatostatin analogues (SSAs)

19th International Pituitary Congress

Oxtend™ 01

Annick Menetrey[1], Dalit Rechavi-Robinson[1], Yanina Negievich[1], Justyna Nowakowska[1], Daniel Biasse[1], Moritz Marquardt[1], Bruno Gavillet[1], Anne Bellon[1], Pedro Caetano[1], Przemysław Witek[2], Mikkel Andreassen[3], Philippe Chanson[4], Thierry Brue[5]

[1] Debiopharm International SA, Lausanne, Switzerland; [2] Medical University of Warsaw, Department of Internal Medicine Endocrinology and Diabetes, Warsaw, Poland; [3] Rigshospitalet, Department of Nephrology and Endocrinology, Copenhagen, Denmark; [4] Assistance Publique-Hôpitaux de Paris, Université Paris-Saclay, Hôpital Bicêtre, Service d'Endocrinologie et des Maladies de la Reproduction et Centre de Référence des Maladies Rares de l'Hypophyse (HYPO), Le Kremlin-Bicêtre, France; [5] Aix Marseille Univ, INSERM, UMR1251, Marseille Medical Genetics, Institut MarMaRa and Department of Endocrinology, La Conception Hospital, APHM, Marseille, France

INTRODUCTION

Debiopharm International SA is developing Debio 4126, a 12-week octreotide formulation, for the treatment of patients with acromegaly. Final results from the acromegaly cohort in the OxtendTM-01 (NCT05364944, also known as Debio 4126-102) are presented here for the first time.

METHODS

The Debio 4126-102 trial (OXTEND-01) was a Phase 1b, open-label, multicenter trial in patients with acromegaly to characterize the pharmacokinetics (PK), pharmacodynamics (PD), safety, and tolerability of Debio 4126 administered intramuscularly (IM) every 12 weeks (Q12W).

The primary objective was to characterize the octreotide plasma PK profile in acromegaly and gastroenteropancreatic neuroendocrine tumor (GEP-NET) patients after single and repeated doses of Debio 4126.

The trial design (**Figure 1**) included 4 Debio 4126 administrations in 2 parallel cohorts of patients with acromegaly and GEP-NETs (15 patients in each cohort). This poster discusses only the acromegaly cohort.

The acromegaly cohort enrolled adult patients with controlled disease (insulin-like growth factor 1 [IGF-1] ≤1.3x upper limit of normal [ULN]) on a stable dose of octreotide or lanreotide as monotherapy (≥6 months).

At the beginning of the Run-in period, 4 weeks before Day (D)1, patients received a last dose of the somatostatin analog (SSA) they were receiving previously. On D1 of the treatment period, patients received Debio 4126 at a starting dose corresponding to the SSA dose (see **Table 2**).

Plasma octreotide (by LC-MS/MS) and serum IGF-1 (by chemiluminescence assay) were measured centrally. Abdominal ultrasounds were performed at least every 3 months to monitor for potential cholelithiasis.

Figure 1 : Debio 4126-102 trial design

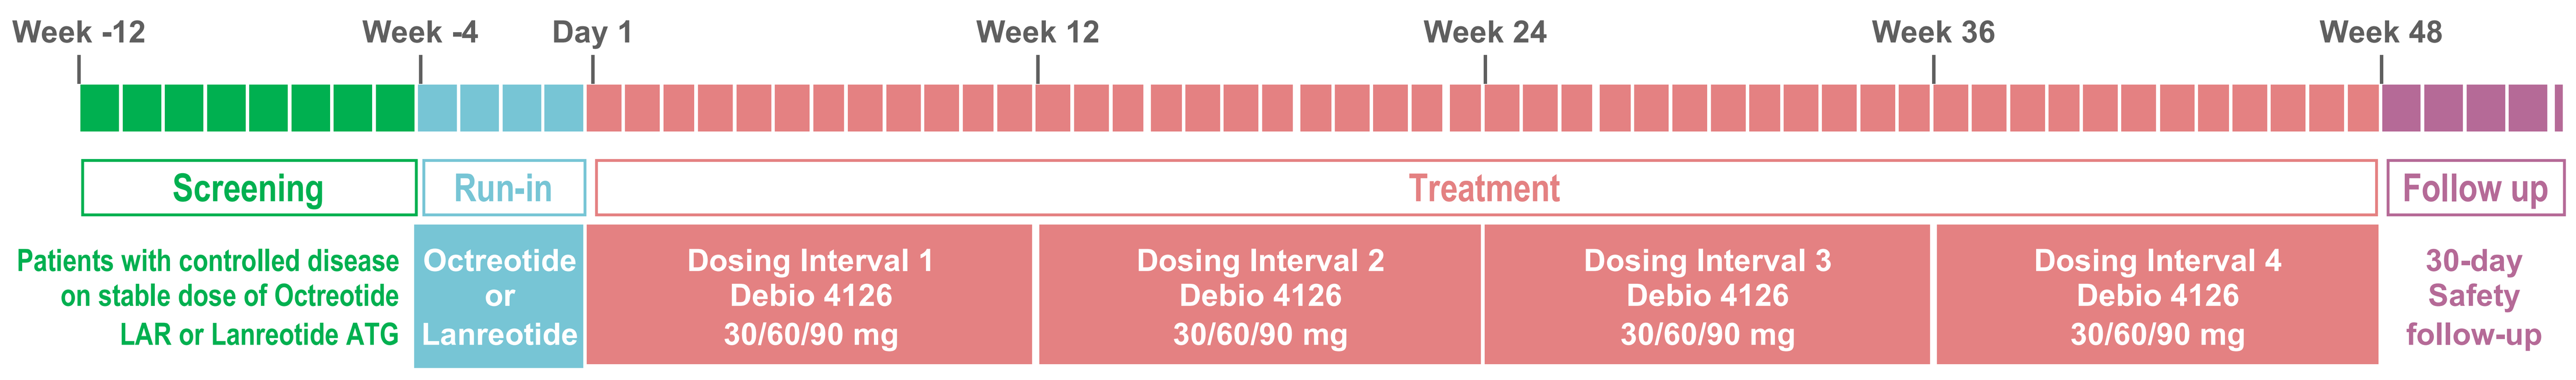
Week -12
Week -4
Day 1
Week 12
Week 24
Week 36
Week 48
Screening
Run-in
Treatment
Follow up
Patients with controlled disease on stable dose of Octreotide LAR or Lanreotide ATG
Octreotide or Lanreotide
Dosing Interval 1 Debio 4126 30/60/90 mg
Dosing Interval 2 Debio 4126 30/60/90 mg
Dosing Interval 3 Debio 4126 30/60/90 mg
Dosing Interval 4 Debio 4126 30/60/90 mg
30-day Safety follow-up

RESULTS

Recruitment began in October 2022, and the last patient last visit occurred in December 2024. The trial enrolled 16 patients with acromegaly, of which 15 completed the trial per protocol. One patient discontinued the trial after 2 dosing intervals, due to physician decision not related to safety concerns.

Patient baseline parameters are presented in **Table 1**. Starting doses vs. treatment during the Run-in period are presented in **Table 2**.

Treatment-emergent adverse events (TEAEs) related to Debio 4126 are presented in **Table 3**. No deaths or serious adverse events (SAEs) were observed. Cholelithiasis was reported in 4 patients (including 3 with biliary sludge without stones), all asymptomatic, and not requiring any treatment or medical interventions. In 1 patient, a Grade 3 alanine aminotransferase per Common Terminology Criteria for Adverse Events (CTCAE) was observed. The event resolved spontaneously without change in treatment. All other TEAEs were Grade 1-2.

Table 1: Patient baseline characteristics

Parameter	Patients with acromegaly N=16
Age <65 years: n (%)	14 (87.5)
Age (years): minimum, **median**, maximum	33, **57.0**, 74
Females: n (%)	7 (43.8)
Height: mean (SD)	173.8 (12.4)
Weight: mean (SD)	87.2 (22.5)
BMI <30 kg/m^2: n (%)	11 (68.8)
BMI (kg/m^2): mean (SD)	28.6 (5.7)
Years from acromegaly diagnosis to Run-in: minimum, **median**, maximum	2.9, **9.37**, 33.3
Prior cholecystectomy: n (%)	4 (25.0)
IGF-1 at screening	
≤1x ULN: n (%)	13 (81.3)
>1x ULN ≤1.3x ULN: n (%)	3 (18.8)
IGF-1 at baseline (last assessment prior to first dose)	
≤1x ULN: n (%)	12 (75.0)
>1x ULN ≤1.3x ULN: n (%)	3 (18.8)
>1.3x ULN: n (%)	1 (6.3)

IGF-1, insulin-like growth factor 1; N, overall number; n, number out of N; SD, standard deviation

Table 2: Starting dose vs. treatment during Run-in

		SSA during the Run-in Period	
Debio 4126 starting dose	N	Octreotide N (dose)	Lanreotide N (dose)
30 mg	3	1 (10 mg)	2 (60 mg)
60 mg	5	2 (20 mg)	3 (90 mg)
90 mg	8	6 (30 mg)	2 (120 mg)

N, number; SSA, somatostatin analog

PK AND PD RESULTS

PK results showed a sustained octreotide release over the intended 12-week treatment period (**Figure 2**). Octreotide exposure increased with Debio 4126 dose across 30 mg, 60 mg, and 90 mg, without a formal dose-proportionality.

Debio 4126 did not show a relevant octreotide accumulation after repeated administrations and reached the steady state after the second administration. The Debio 4126 PK profile was similar to that observed in healthy volunteers (see ref. 1).

During the trial, there was only 1 up-titration in a patient who had IGF-1 <1x ULN at screening, but whose IGF-1 increased up to 1.8x ULN during the Run-in period (**Figure 3**, Patient 2). Following the up-titration to Debio 4126 60 mg on C2D1, IGF-1 levels decreased slightly initially, but stayed above 1x ULN. The other 15 patients (**Figure 3**) had IGF-1 <1.3x ULN at baseline. **Of 14 patients with both baseline IGF-1 ≤1.3x ULN and an IGF-1 assessment at the end of the fourth dosing interval, 13 (92.8%) maintained an IGF-1 level ≤1.3x ULN.** Of 11 patients with both baseline IGF-1 ≤1.0x ULN and an IGF-1 assessment at the end of the fourth dosing interval, 9 (81.8%) maintained an IGF-1 level ≤1.0x ULN. The results suggest that the conversion used between commercial SSAs and the Debio 4126 starting doses allows to maintain IGF-1 levels.

Figure 2 : PK profiles after a single and repeated administration

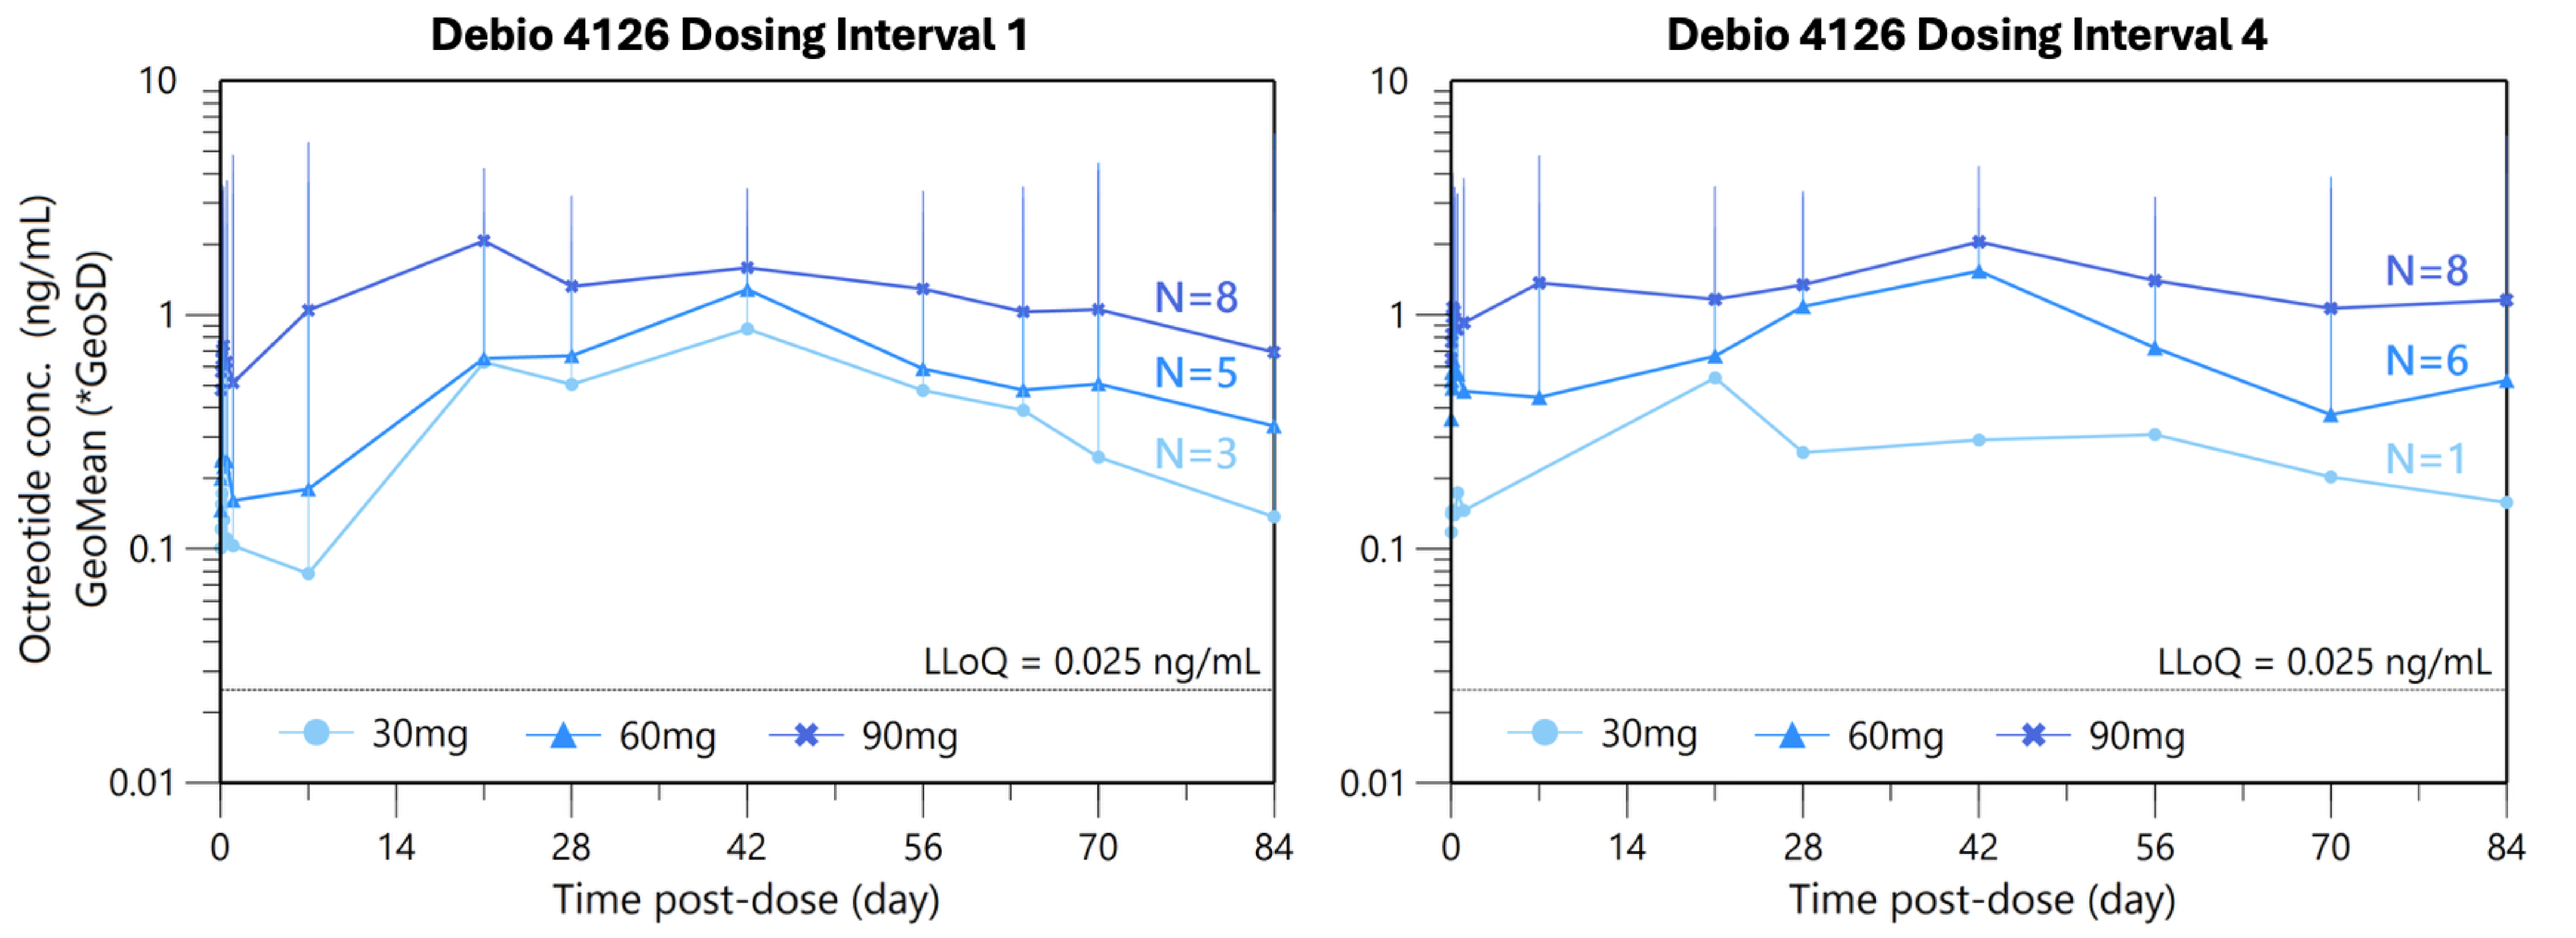
Debio 4126 Dosing Interval 1
Debio 4126 Dosing Interval 4
Octreotide conc. (ng/mL) GeoMean (*GeoSD)
10
1
0.1
0.01
N=8
N=5
N=3
N=8
N=6
N=1
LLoQ = 0.025 ng/mL
30mg
60mg
90mg
0
14
28
42
56
70
84
Time post-dose (day)

Abbreviations: Geo: Geometric; LLoQ: Lower limit of quantification; N: number of individual profiles; SD: Standard deviation

Table 3: TEAEs related to Debio 4126 occurring in at least 2 patients, by preferred term

System Organ Class Preferred Term	Debio 4126 30 mg N=3	Debio 4126 60 mg N=5	Debio 4126 90 mg N=8	Total N=16
Any TEAE related to Debio 4126	3 (100)	2 (40.0)	5 (62.5)	10 (62.5)
General disorders and administration site conditions	**1 (33.3)**	**1 (20.0)**	**2 (25.0)**	**4 (25.0)**
Injection site erythema	1 (33.3)	0	1 (12.5)	2 (12.5)
Injection site induration	1 (33.3)	1 (20.0)	0	2 (12.5)
Injection site inflammation	0	0	2 (25.0)	2 (12.5)
Hepatobiliary disorders	**1 (33.3)**	**0**	**2 (25.0)**	**4 (25.0)**
Cholelithiasis	1 (33.3)	0	2 (25.0)	4 (25.0)
Nervous system disorders	**1 (33.3)**	**0**	**1 (12.5)**	**2 (12.5)**
Headache	1 (33.3)	0	1 (12.5)	2 (12.5)

Abbreviations: N, number; TEAE, treatment-emergent adverse event
Note: AE reported terms were coded using the Medical Dictionary for Regulatory Activities (MedDRA) Version 27.0. Patients are counted once within each SOC and PT. The same patient may contribute to 2 or more preferred terms in the same SOC.

Figure 3 : Individual IGF-1 concentrations per visit, as multiples of ULN

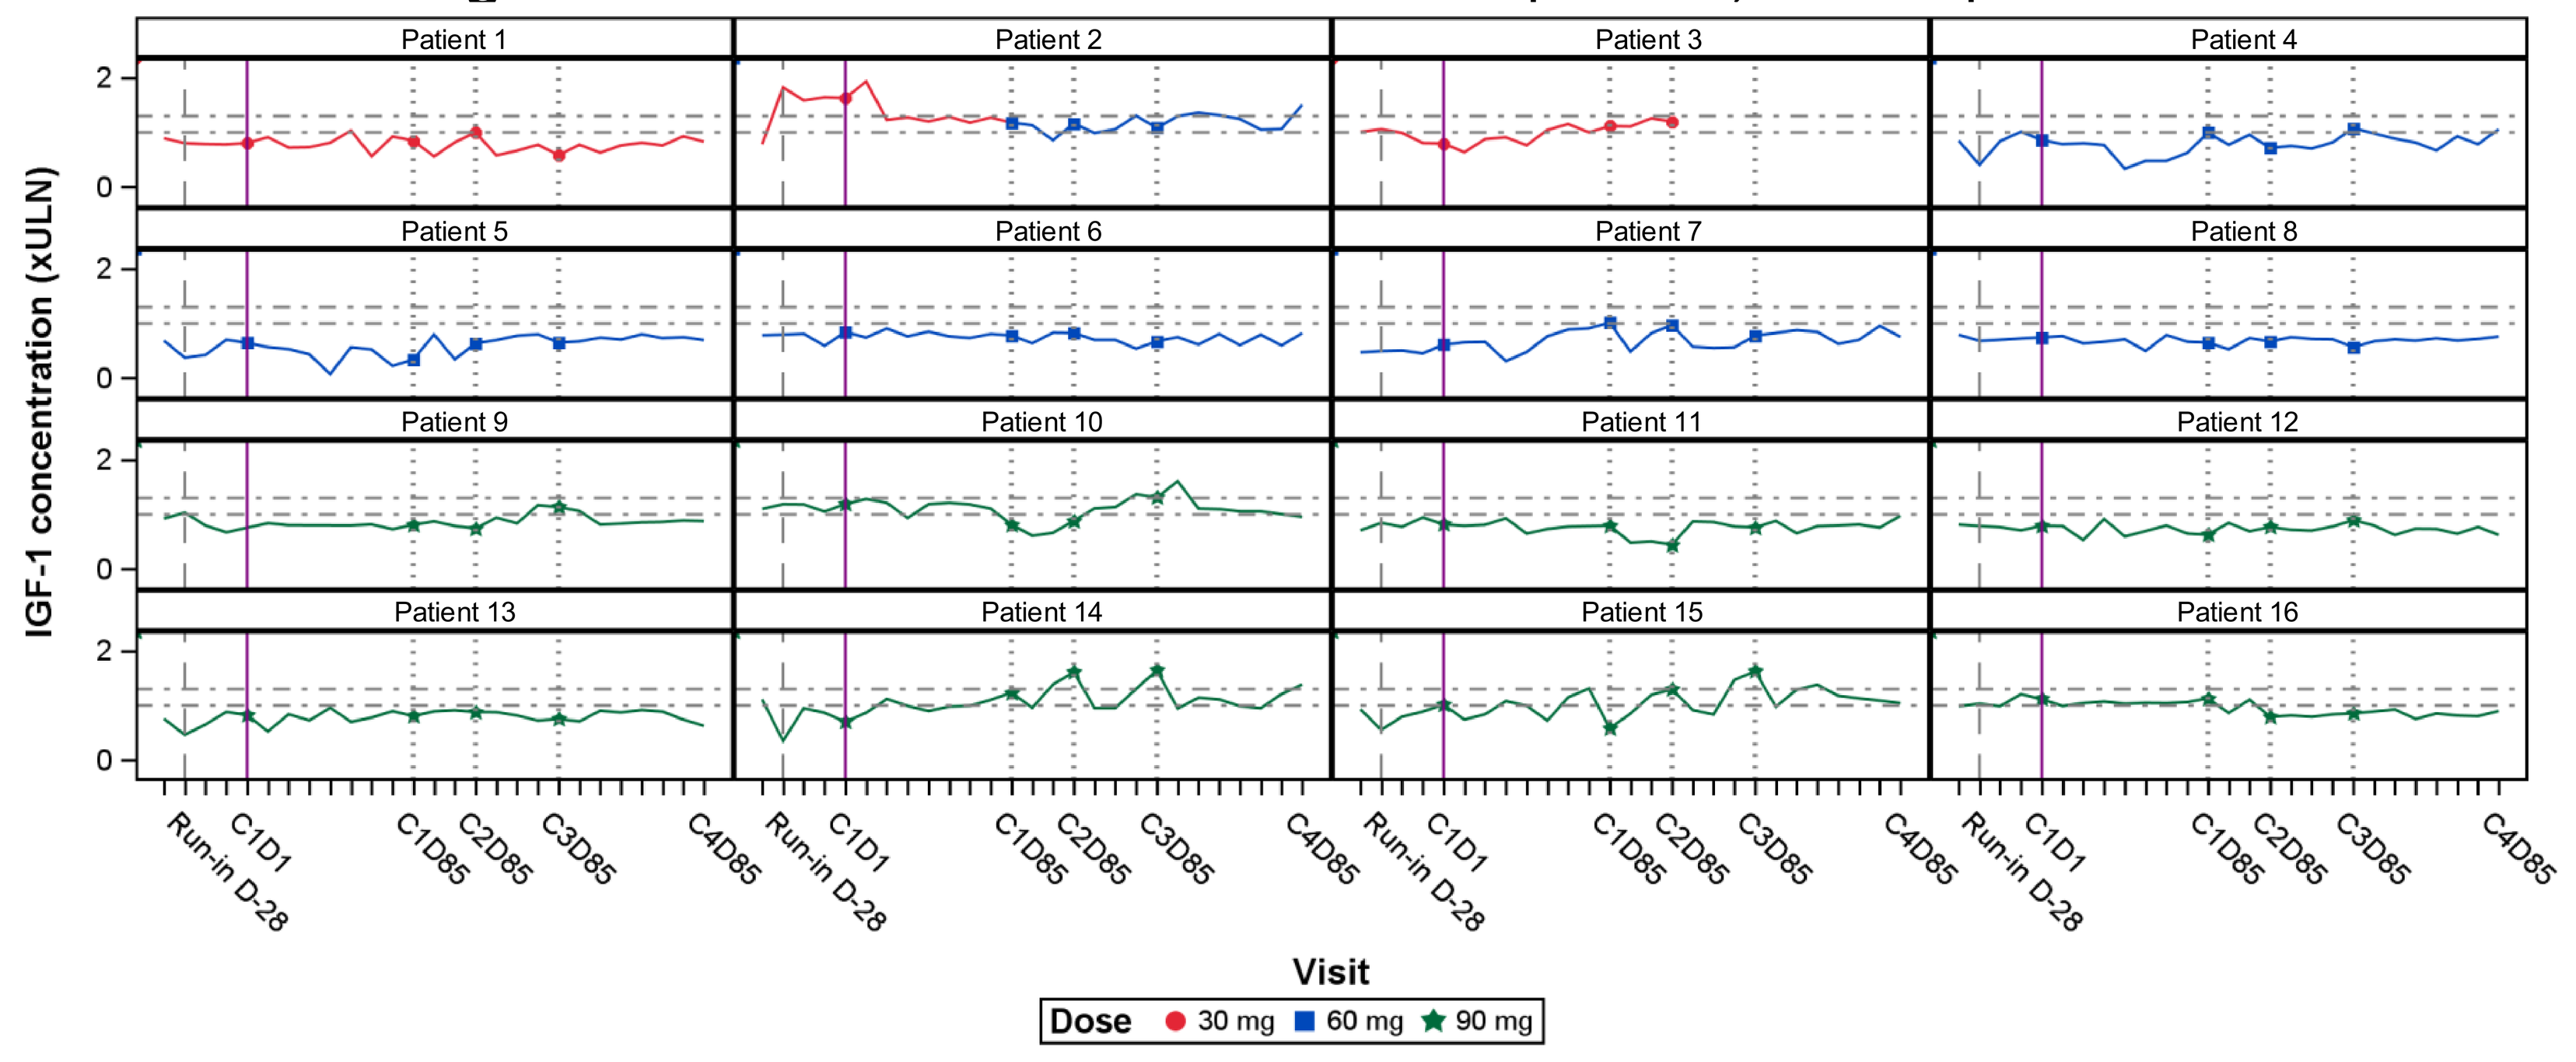
Patient 1
Patient 2
Patient 3
Patient 4
Patient 5
Patient 6
Patient 7
Patient 8
Patient 9
Patient 10
Patient 11
Patient 12
Patient 13
Patient 14
Patient 15
Patient 16
IGF-1 concentration (xULN)
Run-in D-28
C1D1
C1D85
C2D85
C3D85
C4D85
Visit
Dose 30 mg 60 mg 90 mg

Abbreviations: CxDy, dosing interval x, Day y; IGF-1, insulin-like growth factor 1; ULN, Upper limit of normal.
Notes: The grey dashed line represents the start of the run-in period. The purple solid line represents the start of the first dosing interval. The grey dotted lines represent the start of the other dosing intervals. The horizontal dotted lines represent IGF-1 =1x ULN and IGF-1 =1.3x ULN.

CONCLUSIONS

- Debio 4126 administered IM Q12W provides sustained release of octreotide over the intended 12-week dosing interval, without a relevant octreotide accumulation after repeated administrations.
- In the majority of patients with acromegaly, baseline IGF-1 levels were maintained until the end of the 48-week treatment at the Debio 4126 dose matching the preceding SSA dose.
- The safety and tolerability profile is favorable and consistent with that of other SSAs.
- These data support the potential of Debio 4126 administered Q12W to be an effective maintenance therapy for patients with acromegaly.
- The less frequent injection schedule would decrease patient burden.
- Debio 4126 efficacy and safety will be further assessed in OxtendTM-03 (NCT06930625), a pivotal Phase 3 trial currently recruiting.

Acknowledgements: The authors would like to thank all patients who accepted to participate in the trial as well as the medical and clinical staff who conducted the trial. Furthermore, the following persons significantly contributed to this work: Anna Perkio, Florilene Bouisset, Simona Ispas-Jouron, Claire Laprun, Claudia Krolzig-Anyanwu, Catherine Dutreix, as well as Medpace, Q2S, and ICON staff.

Conflicts of interest: AM, DRR, YN, JN, DB, MM, BG, AB, and PCa are employees of Debiopharm; PW, MA, PCh, and TB were investigators in the Debio 4126 trial and received compensation from Debiopharm for the work performed.

References: (1) Bellon A. *et al.*, ENEA 2022

CONTACT

**Debiopharm International S.A.,
Lausanne, Switzerland.
www.debiopharm.com
Yanina.Negievich@debiopharm.com**

DOWNLOAD

**This poster is available via:
www.debiopharm.com/medias/publications**

Debiopharm
WE DEVELOP FOR PATIENTS